An algorithm-aided flow cytometry data analysis pipeline to analyze non-human primate immunomonitoring data in large molecule PK/PD studies

Fabian Junker, Rubén Alvarez, Adrian Walmsley, Magali Muller, Benoît Hornsperger,
Roy Meoded, Jean-Philippe Carralot, Vijaya Pattabiraman and Bertolt Kreft

Bright Peak Therapeutics, Basel. Email: fjunker@brightpeaktx.com

ABSTRACT

Rationally designed, fully synthetic peptides have the potential to offer unmet therapeutic benefit in diseases such as autoimmunity and immune oncology. These molecules can be custom-designed and fine-tuned with regards to pharmacokinetics, cell surface receptor specificity and other properties. As part of their preclinical characterization, thorough immunomonitoring in non-human primate (NHP) PK/PD studies is systematically performed. To support this effort, we developed a highly scalable algorithm-aided flow cytometry data analysis pipeline. Flow cytometry raw data are first transformed and normalized prior to cell population clustering through self-organizing maps (FlowSOM). Subsequently, principal component analysis (PCA) as well as population statistics are performed comparing pre-treatment and on-treatment animals. Results are further visualized through Uniform Manifold Approximation and Projection for Dimension Reduction (UMAP). Finally, integrating hematology data, cell population abundance data are then transformed into absolute counts to monitor the expansion of specific cellular subsets in peripheral blood. We here introduce our flow cytometry data analysis pipeline showcasing regulatory T cell (Treg) expansion in NHP dosed with BPT264, a fully synthetic, site-specific 30 kDa PEGylated IL-2 variant engineered to completely ablate binding to IL-2Rβ whilst uniquely augmenting binding to IL-2Rα.

RATIONAL DESIGN OF BPT264

BPT264: Improved, fully synthetic interleukin-2 (IL-2) variant site-specifically conjugated to a 30 kDa PEG to improve pharmacokinetic (PK) properties[1].

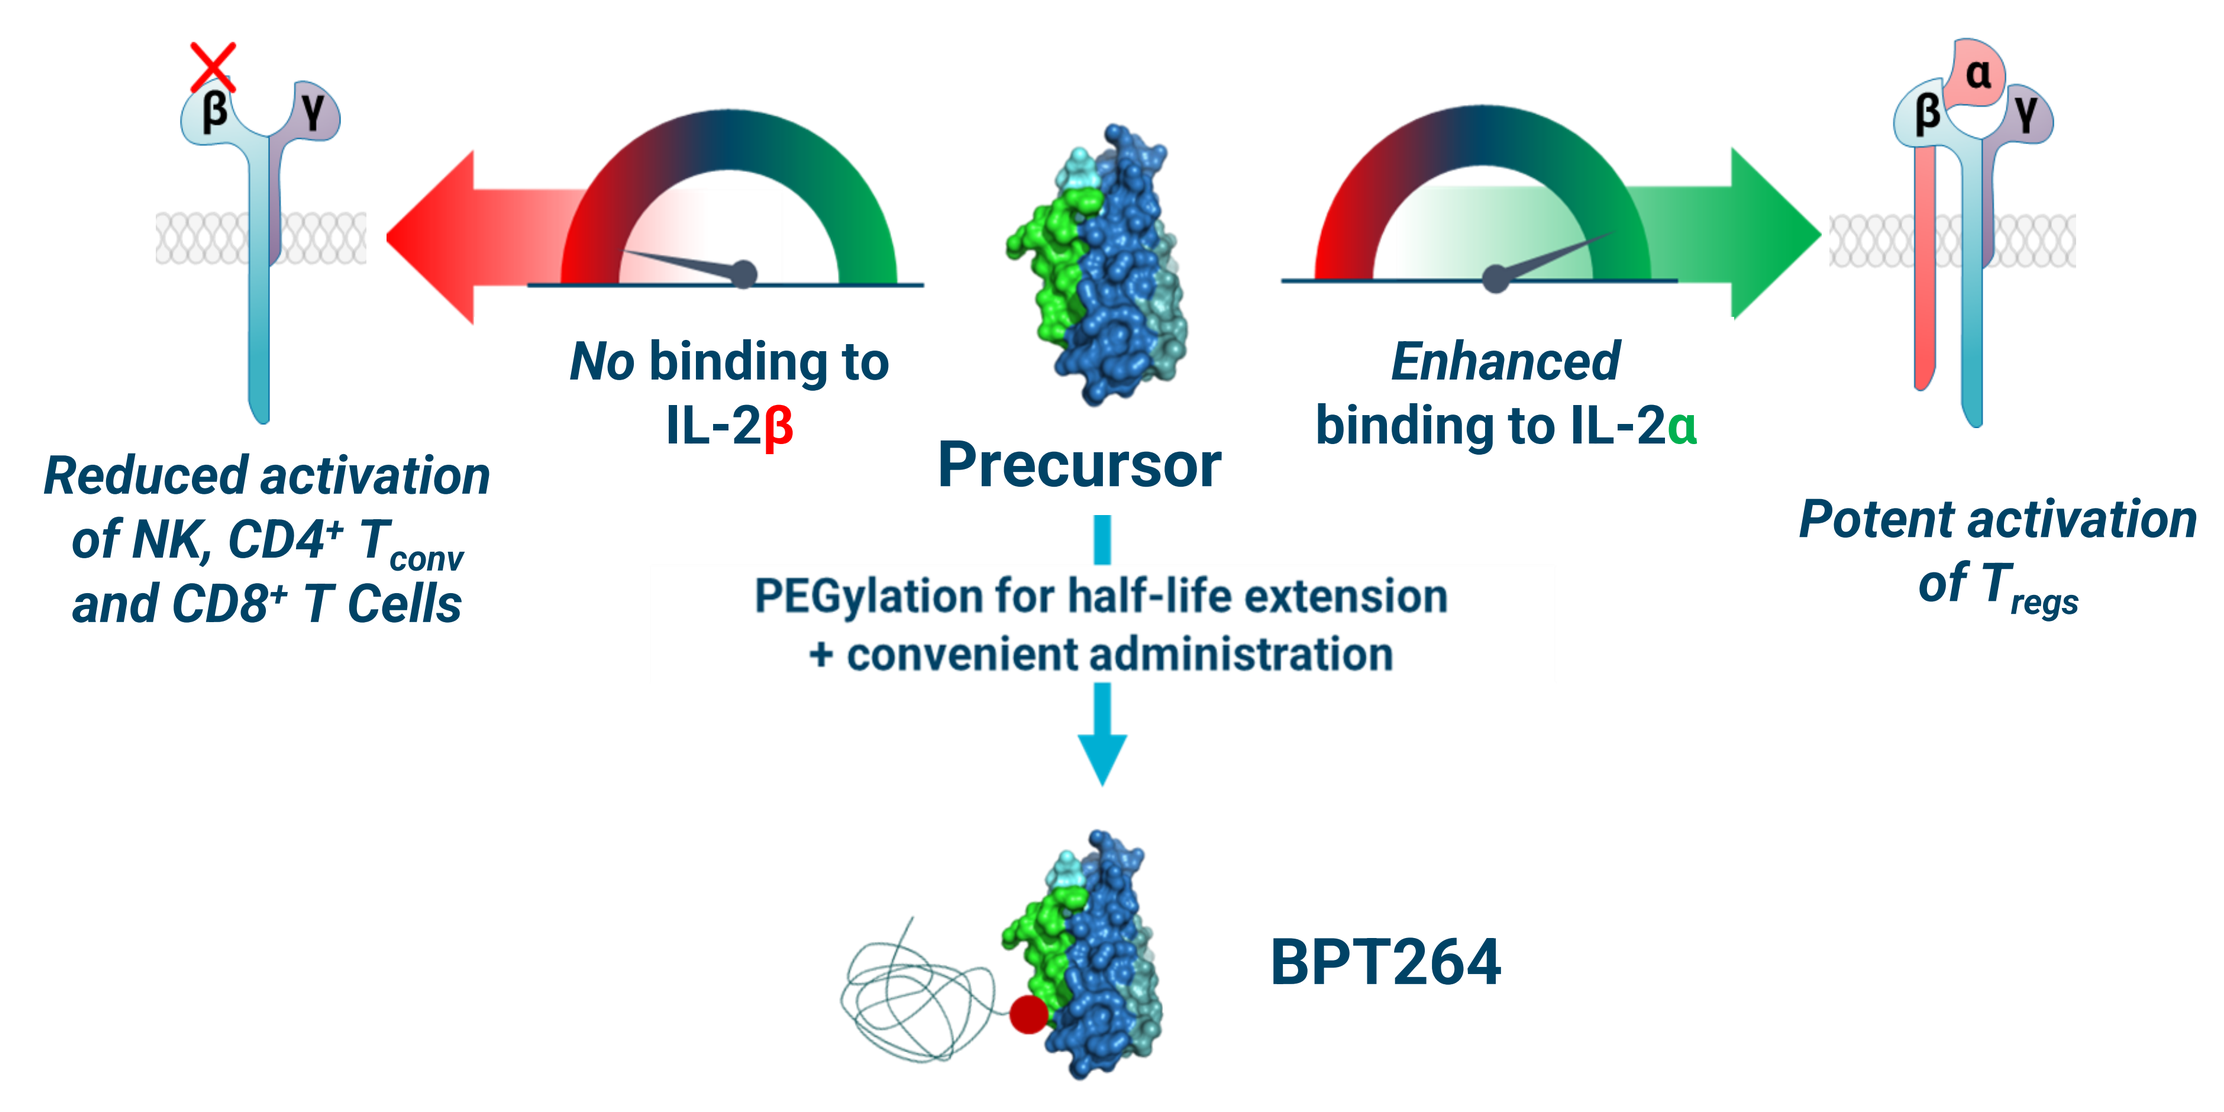
β
γ
Reduced activation of NK, CD4+ Tconv and CD8+ T Cells
No binding to IL-2β
Precursor
Enhanced binding to IL-2α
α
Potent activation of Tregs
PEGylation for half-life extension + convenient administration
BPT264

BPT264 IN VITRO CHARACTERISTICS

BPT264 Selectively Activates T_{regs} with Minimal Activity on CD8⁺ T cells, CD4⁺ T_{conv} and NK cells in Human, NHP and Mouse

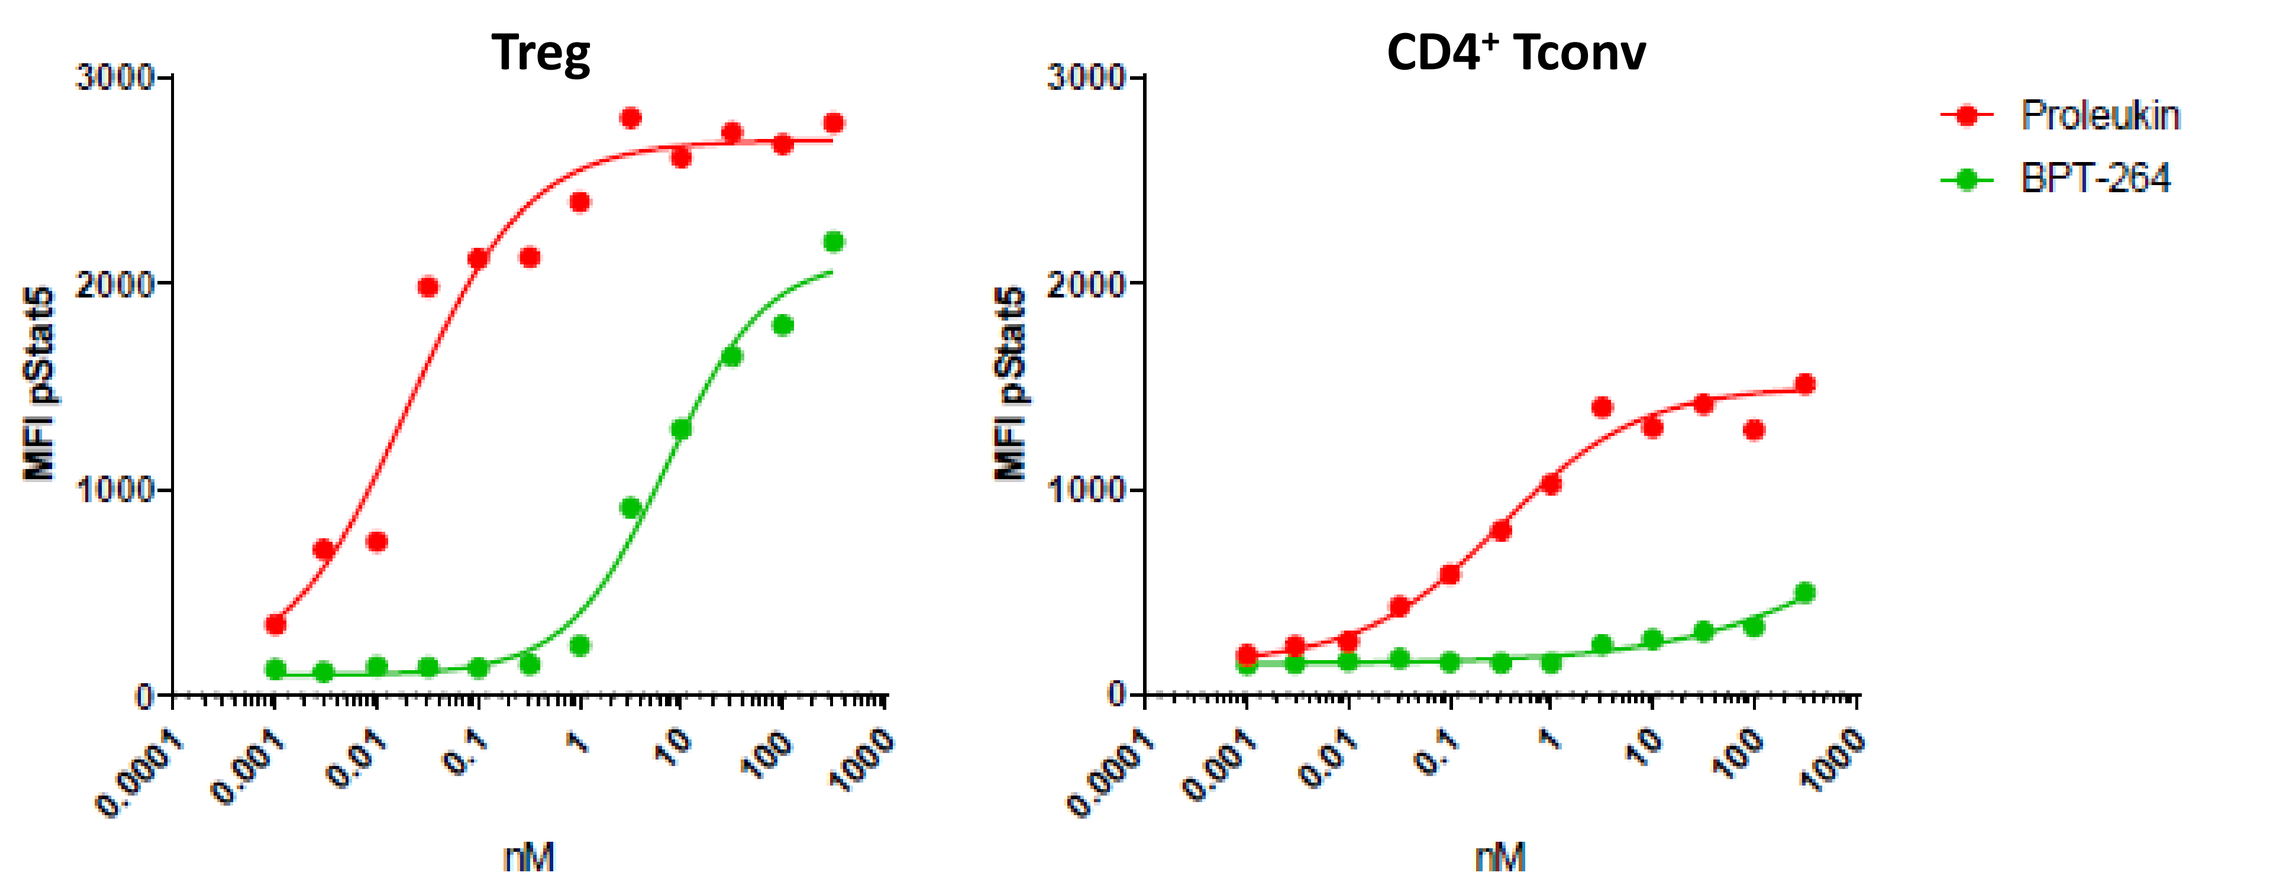
Treg
CD4+ Tconv
Proleukin
BPT-264
MFI pStat5
nM
0.0001
0.001
0.01
0.1
1
10
100
1000

Figure 1. STAT5 phosphorylation in primary human T_{regs} or CD4⁺ T_{conv} cells after stimulation with Proleukin (unmodified IL-2) and BPT264.

Species	BPT264 EC_{50} (nM)		
	Treg	CD4⁺ Tconv	CD8⁺ T cell
Human	15.8	>10000	>10,000
NHP	9.17	>10000	>10,000
Mouse	45.45	>10000	>10,000

Table 1: pSTAT5 phosphorylation assay EC_{50} data summary.

NHP IN VIVO PK/PD STUDY DESIGN

Study design: 3 dose groups, n=2/group

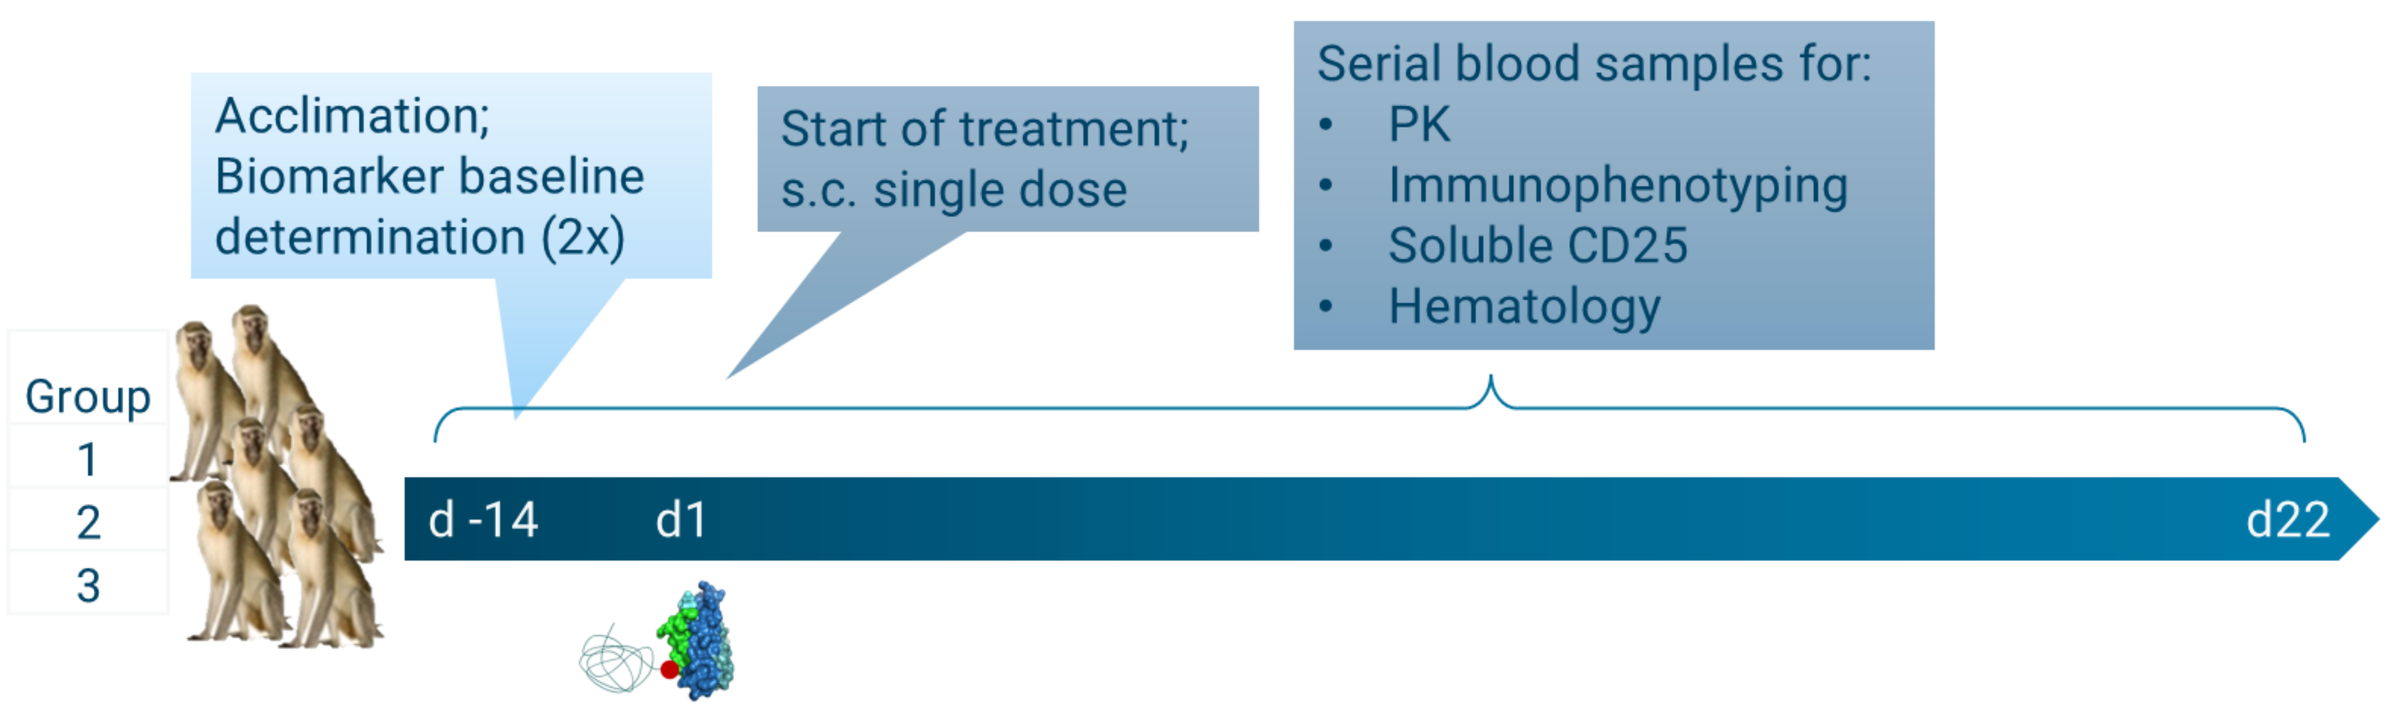
Acclimation;
Biomarker baseline determination (2x)
Start of treatment;
s.c. single dose
Serial blood samples for:
PK
Immunophenotyping
Soluble CD25
Hematology
Group
1
2
3
d -14
d1
d22

Flow cytometry for PD immunomonitoring to assess:

1. **Proof-of-concept (PoC):** Selective expansion of Tregs?
2. **Immuno-PD:** Kinetics of Treg expansion on-treatment?

FLOW CYTOMETRY IMMUNO-PD PANEL

Marker	Lineage
Live/Dead	
CD45	Leukocytes
CD25	Treg cells ($CD25^{++}$)[2]
CD4	Treg and conventional CD4⁺ T cells
CD45RA	Memory vs. naïve T cell subsets
CD8	Cytotoxic T cells
CD127	Effector T cells vs Treg cells ($CD127^{-/lo}$)[2]
NKG2A	NK cell marker
CD3	Total T cell marker

Table 2. Flow cytometry immuno-PD panel used in BPT264 PK-PD study. Raw data (FCS) files were subsequently analyzed in-house. Hematology data were available for the same timepoints.

ALGORITHM AIDED IMMUNO-PD ANALYSIS

We applied the following FCS raw data analysis workflow:

1. Data cleanup and extraction of live CD45⁺ events
 - *FlowJo* v10.8.1 (BD)
2. ArcSinh transformation and signal alignment (lineage markers):
 - *R* (v4.1.3) and *R Studio* (v22.12.2)
 - *flowCore*[3]
3. Quality control: compare population abundance of original data with transformed FCS files
4. Event clustering using CD3, CD4, CD12, CD127 and NKG2A to identify main lineages across samples
 - *FlowSOM*[4]
 - Heatmap generation for cluster assignment
5. PCA to identify major differences between BL and on-treatment specimens
 - *factoextra*[5]
6. *Base R* and *dplyr*[6] were used to calculate lineage abundance (% total mononuclear events), absolute counts and fold-change (FC) from baseline:
 - Lineage expansion was illustrated using *UMAP*[7] for FlowSOM clusters and marker abundance;
 - Other plots were generated using *ggplot2*[8];
 - Summary statistics (mean, SEM) were calculated using *Rmisc summarySE*[9].
7. For Tregs, relative abundance was compared to manual hierarchical gating to ensure robustness of the results.

IDENTIFYING CELL CLUSTER CHANGES

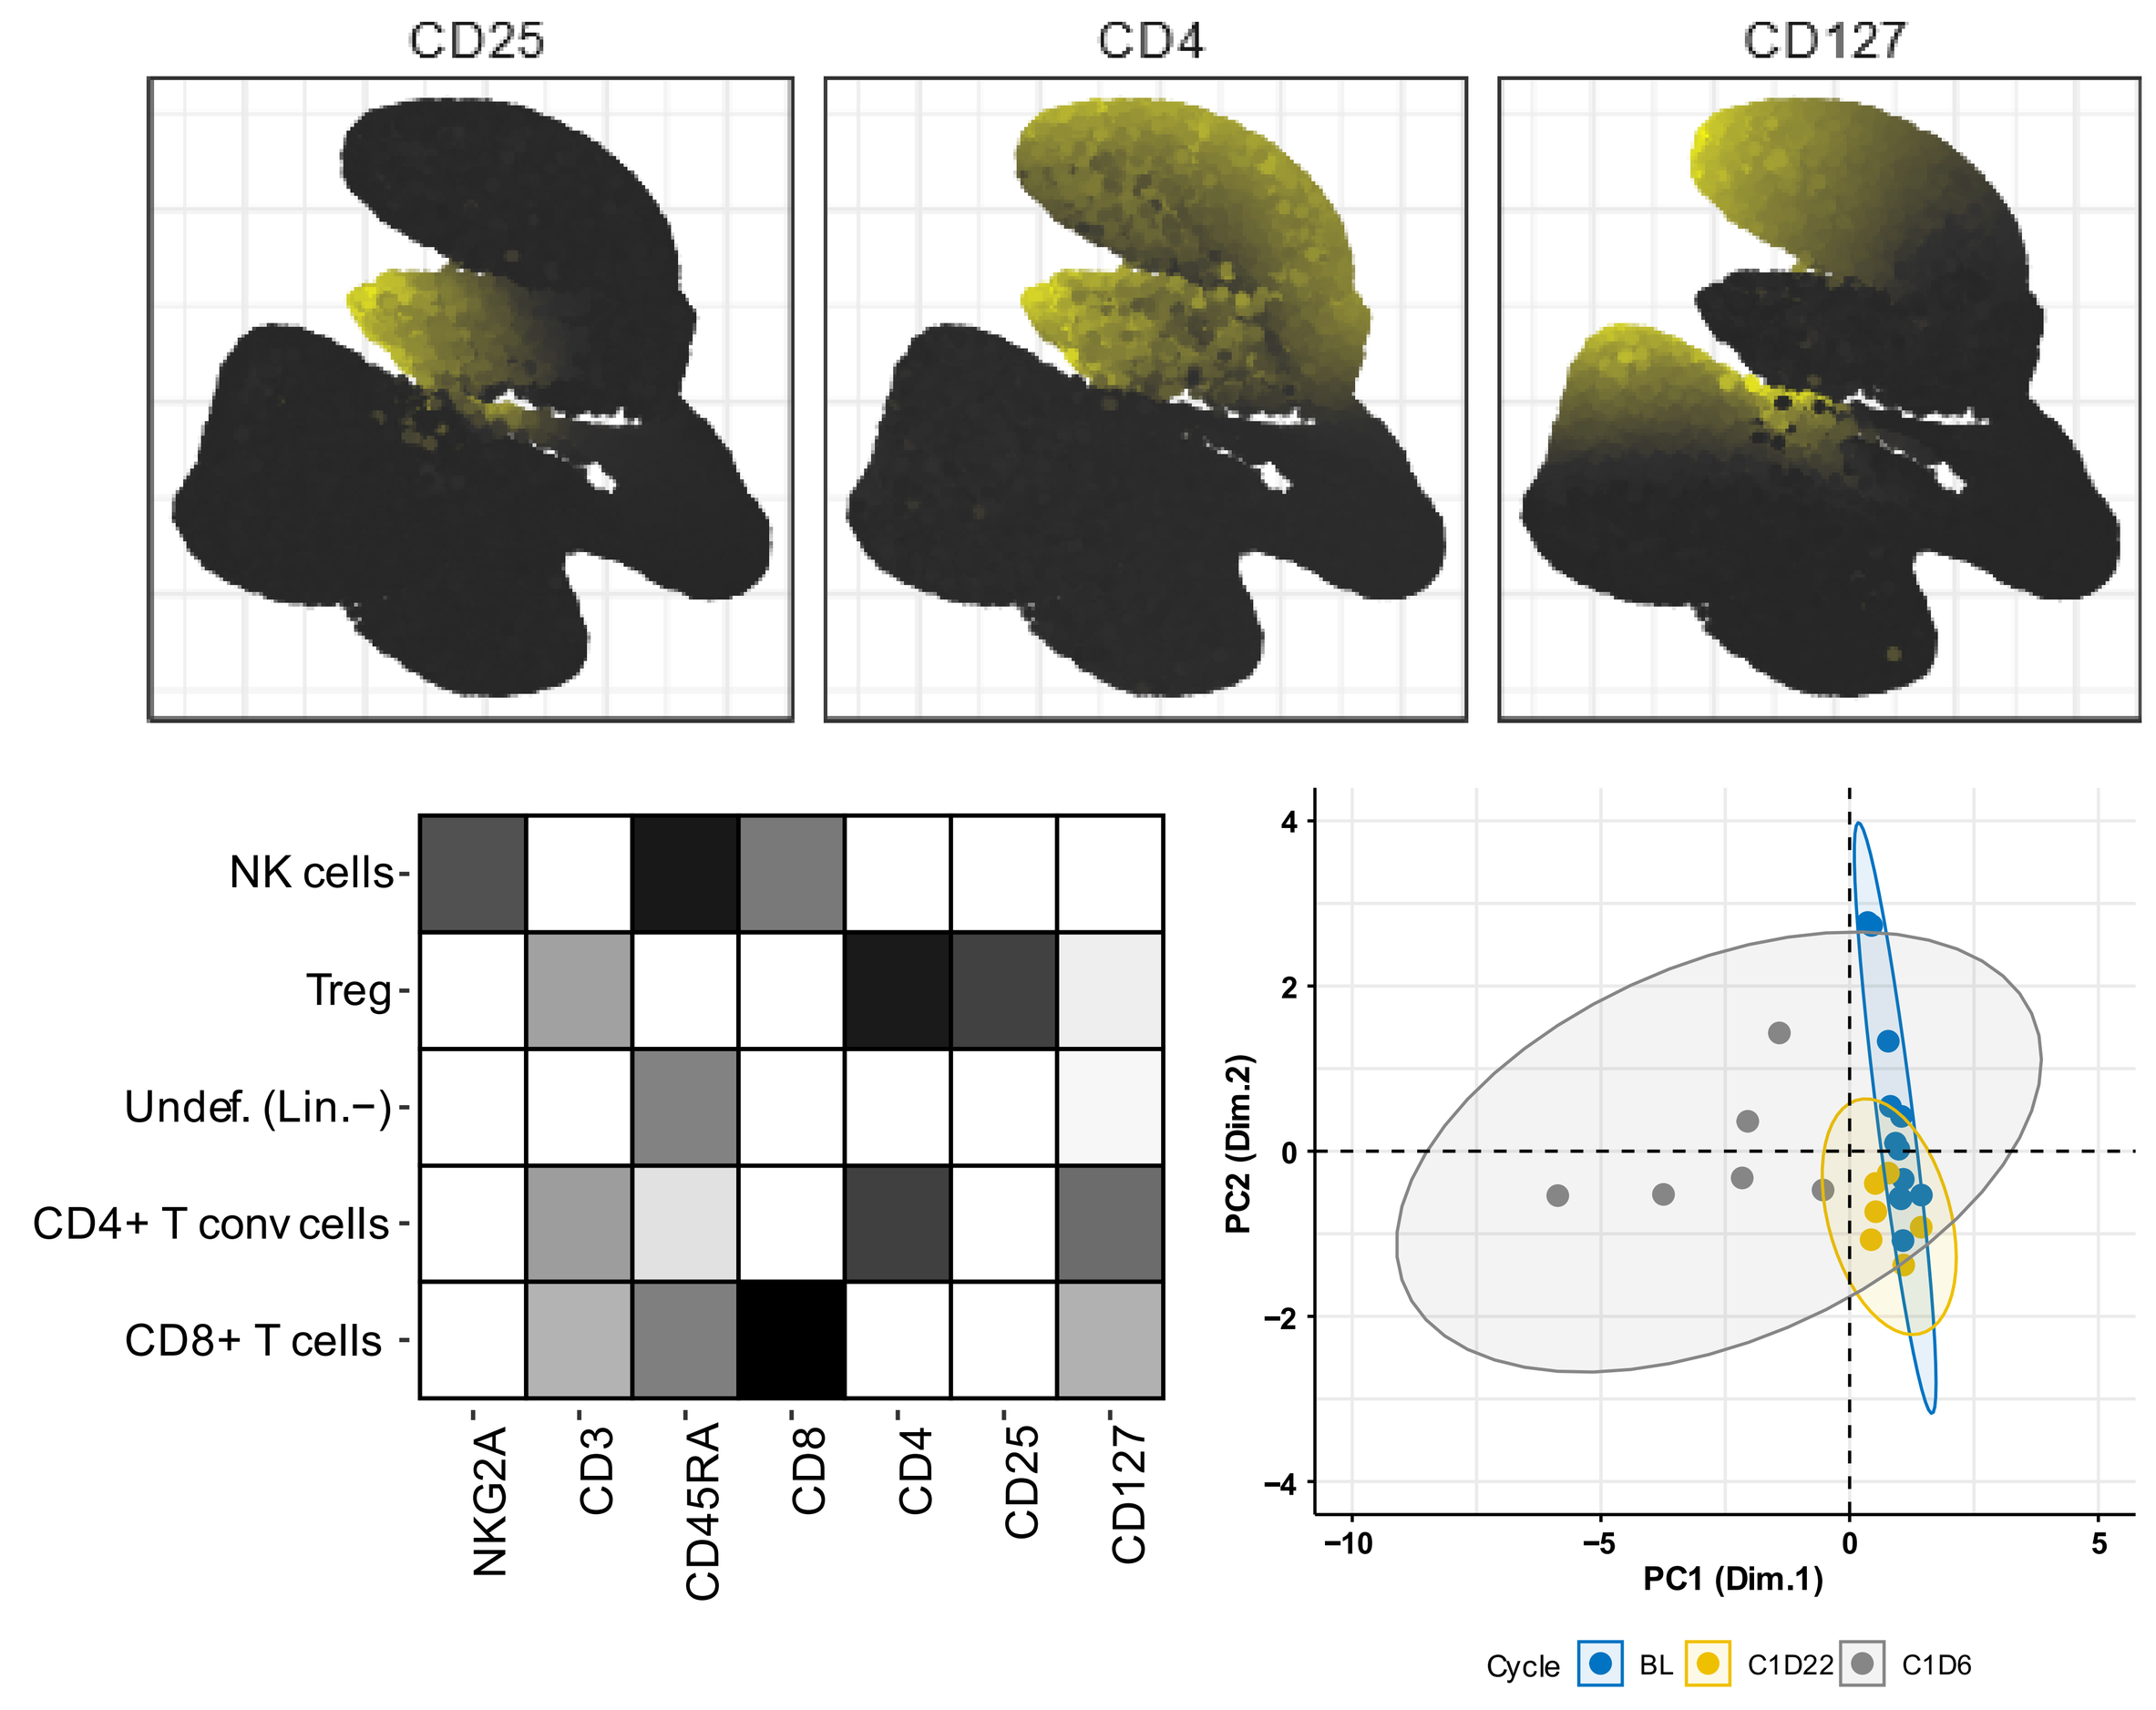
CD25
CD4
CD127
NK cells
Treg
Undef. (Lin.–)
CD4+ T conv cells
CD8+ T cells
NKG2A
CD3
CD45RA
CD8
CD4
CD25
CD127
PC2 (Dim.2)
PC1 (Dim.1)
Cycle
BL
C1D22
C1D6

Figure 2: Top – UMAP example illustration of CD25, CD127 and CD4 expression on mononuclear cells from all animals. Bottom left: FlowSOM clustering identifies canonical cell clusters such as NK cells, CD8⁺, CD4⁺ effector T cells (T conv) and Tregs (shown is a fraction of all identified subsets). Bottom right - PCA suggests treatment-related changes in cell composition in BPT264 dosed animals.

DOSE-DEPENDENT RELATIVE TREG EXPANSION

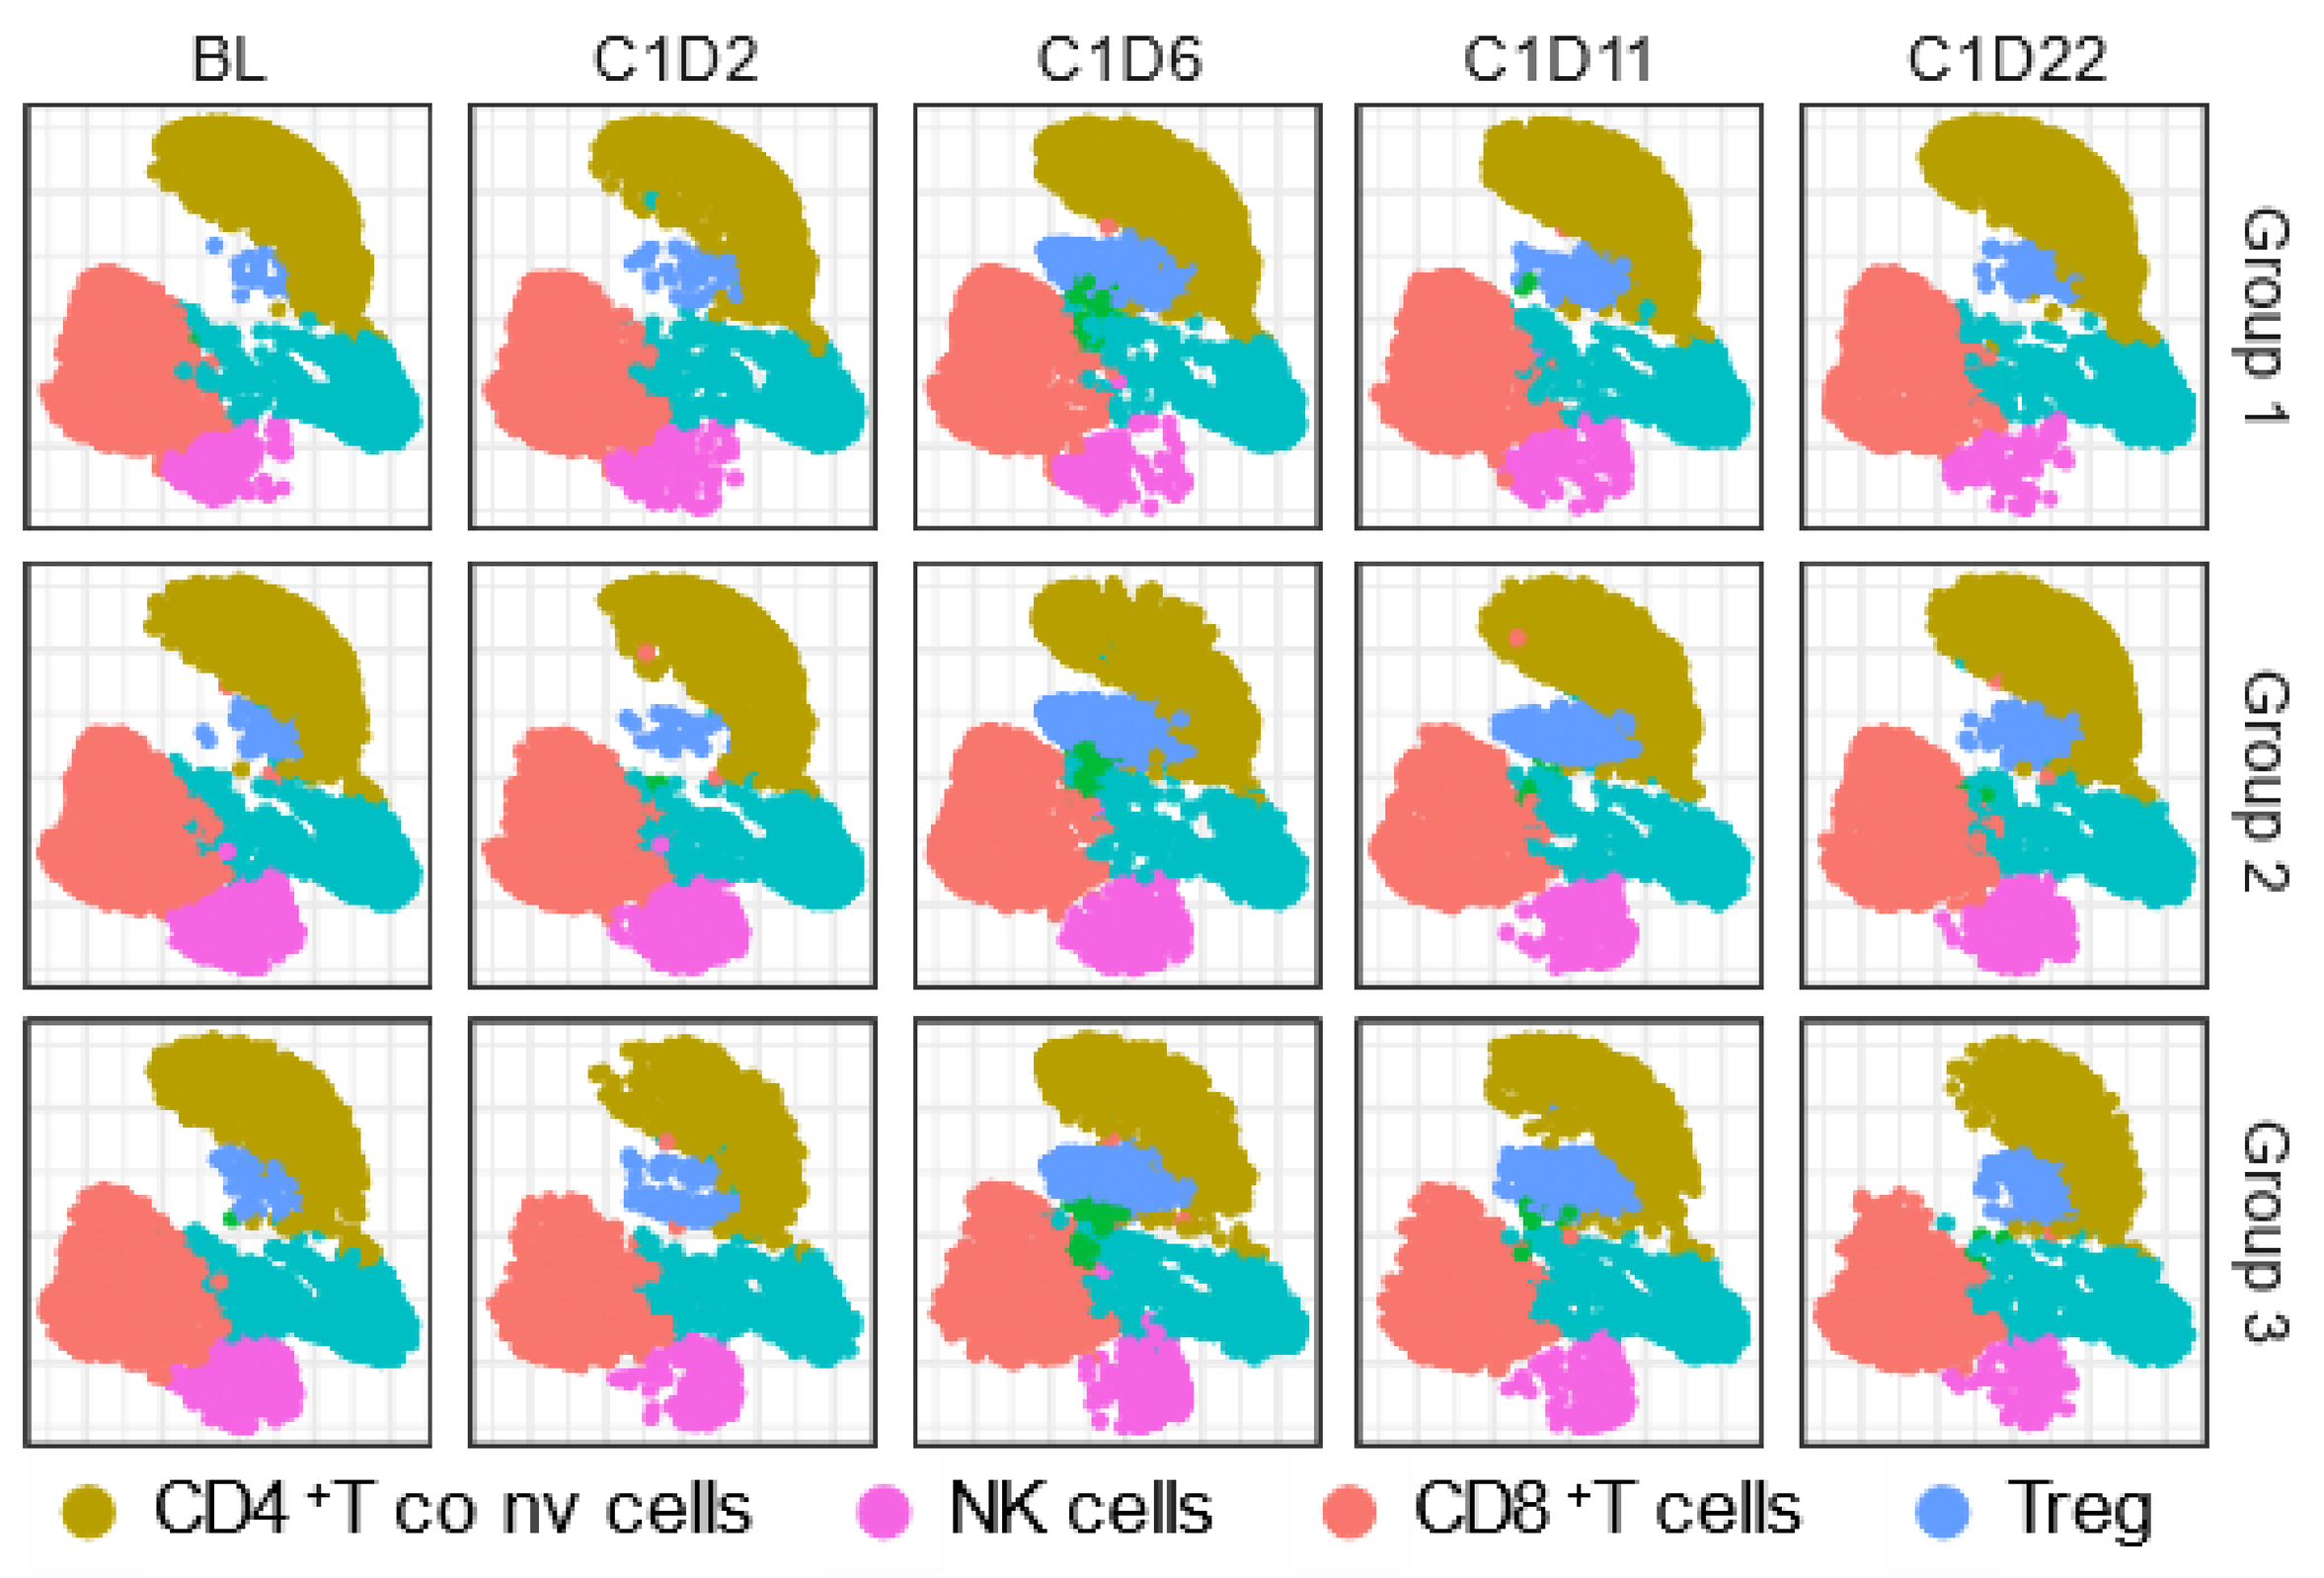
BL
C1D2
C1D6
C1D11
C1D22
Group 1
Group 2
Group 3
CD4 ⁺T co nv cells
NK cells
CD8 ⁺T cells
Treg

Figure 3: Tregs (blue) selectively expand in a BPT264 dose-dependent manner (3 dosing groups, 10'000 events/group). Shown is a subset of timepoints.

BPT264 SELECTIVELY EXPANDS NHP TREGS

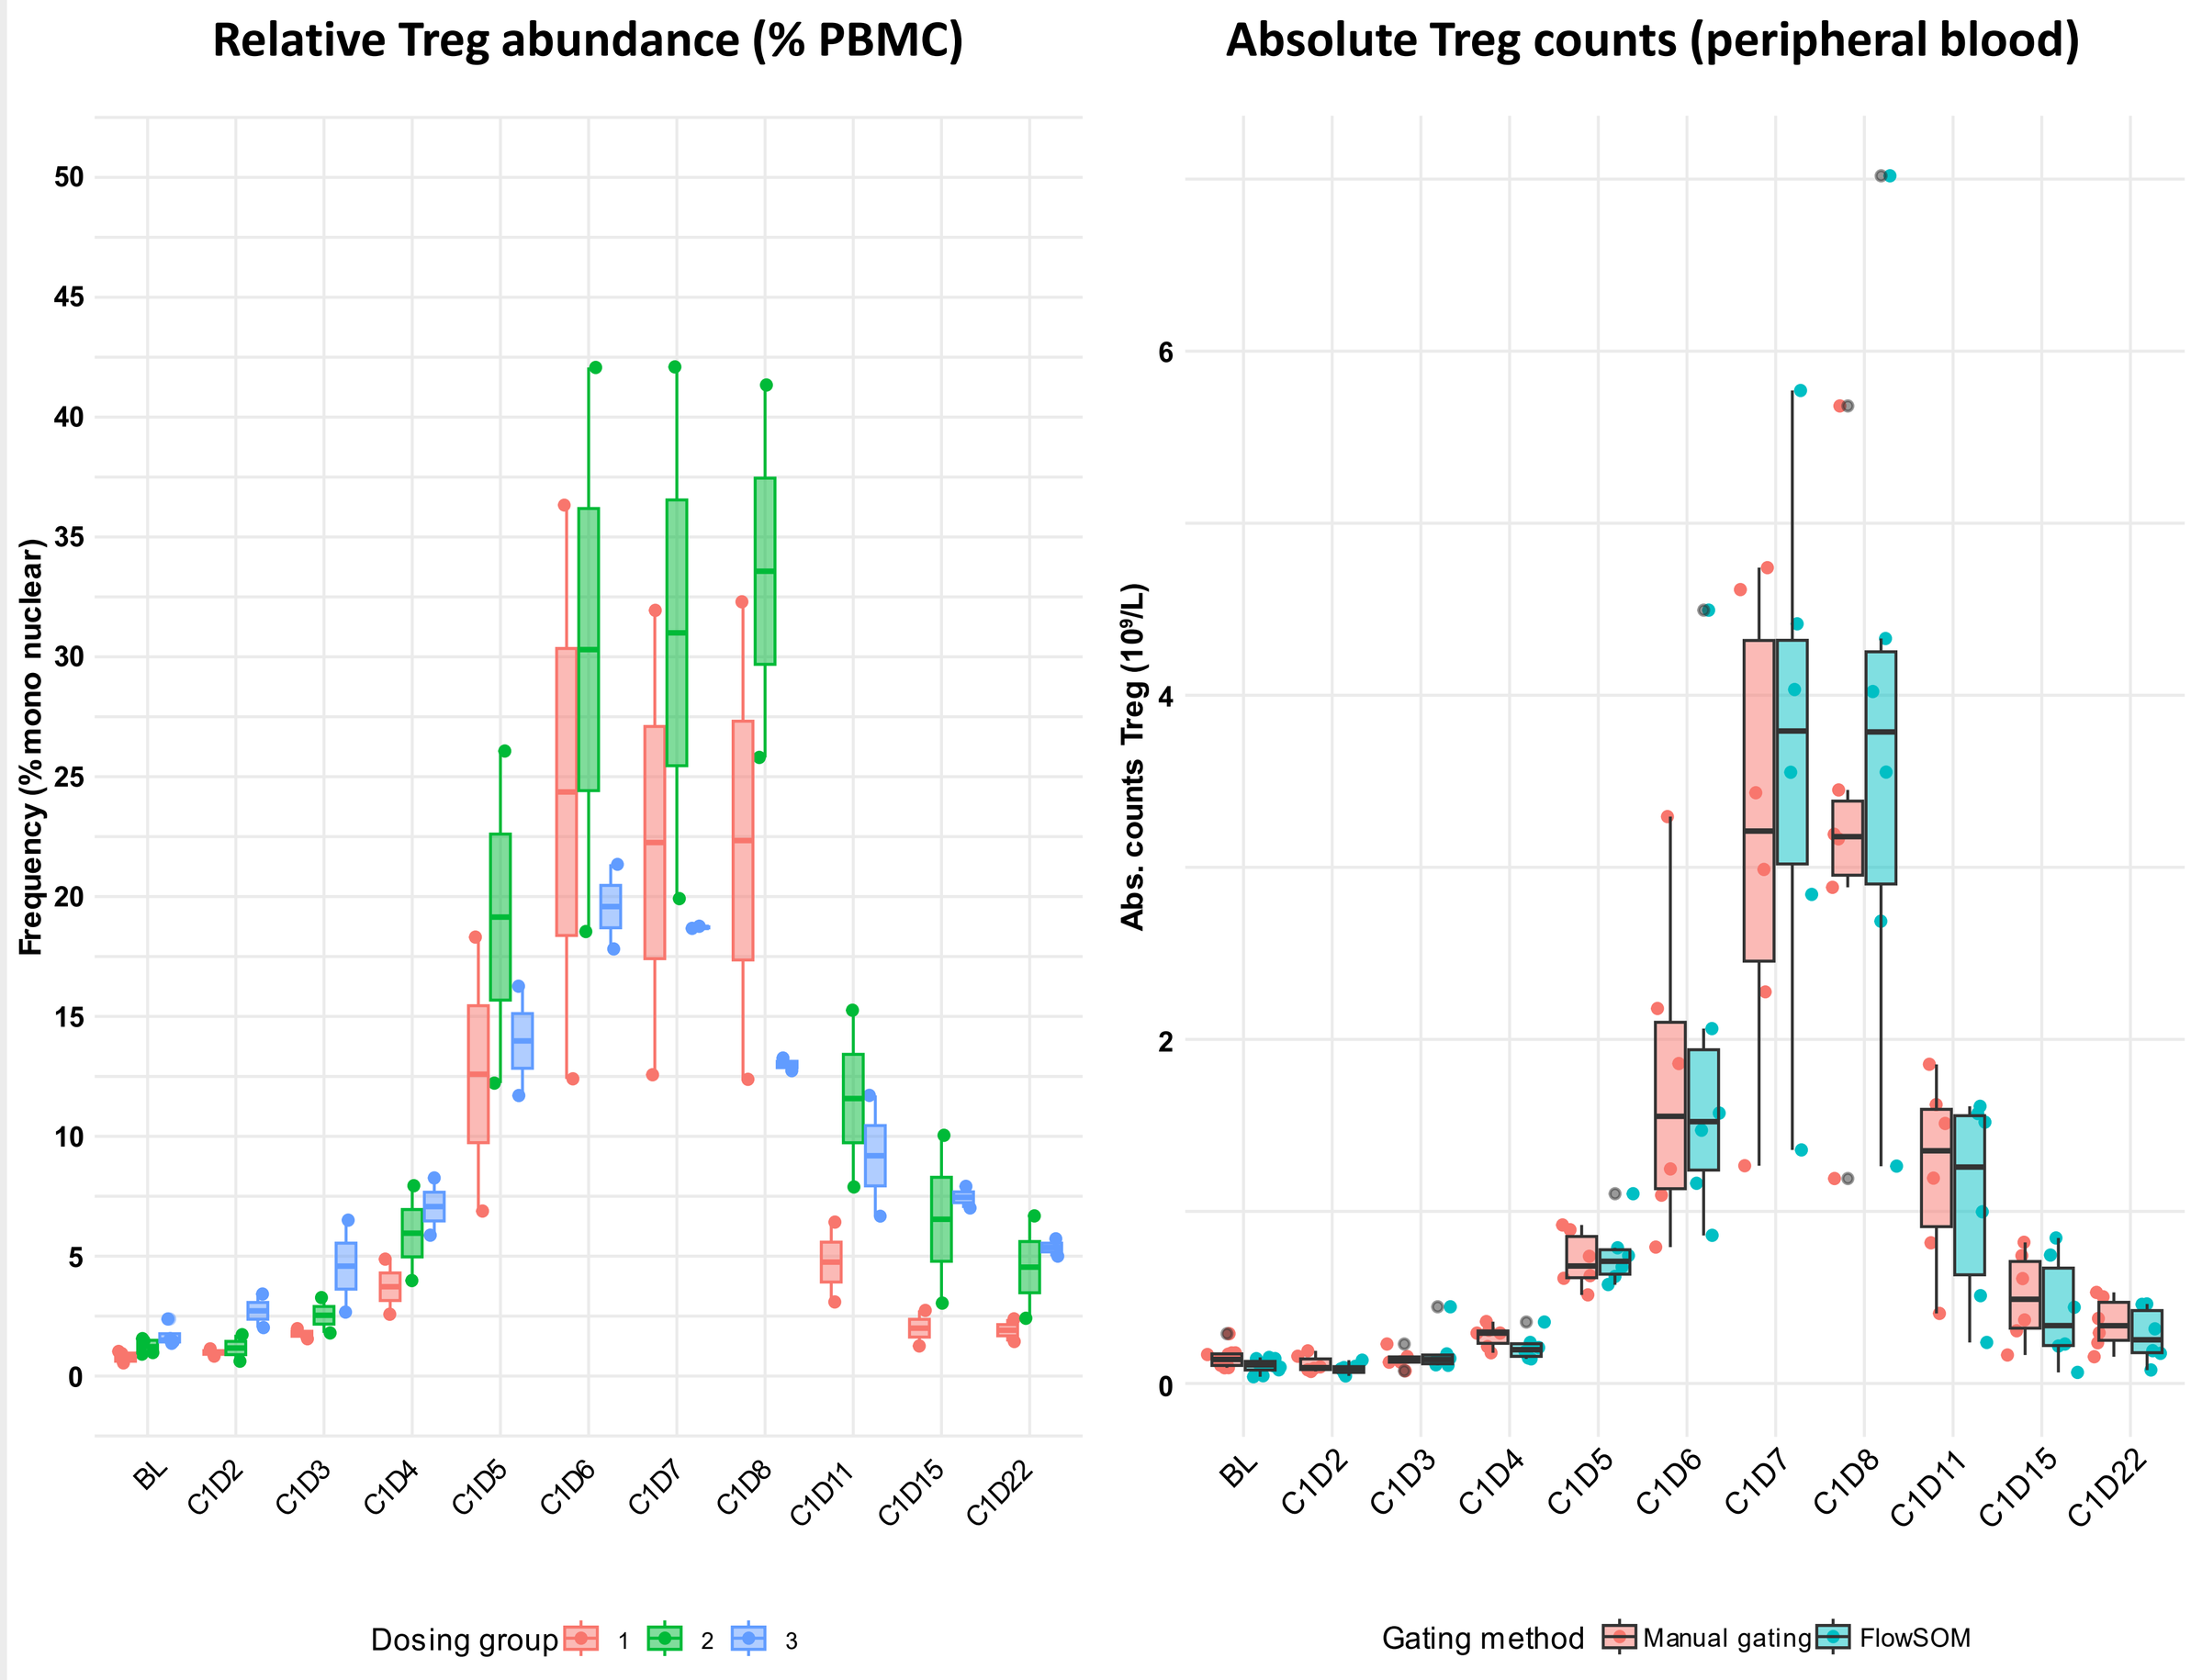
Relative Treg abundance (% PBMC)
Absolute Treg counts (peripheral blood)
Frequency (% mono nuclear)
Abs. counts Treg (10⁹/L)
BL
C1D2
C1D3
C1D4
C1D5
C1D6
C1D7
C1D8
C1D11
C1D15
C1D22
Dosing group
1
2
3
Gating method
Manual gating
FlowSOM

Figure 4: Calculation of Treg cluster abundance (left) and absolute cell numbers (right) in BPT264 dosed animals. Treg absolute counts were calculated based on hierarchical gating (blue) and algorithm-aided clustering (red) for QC. There were no significant differences between the 2 methods.

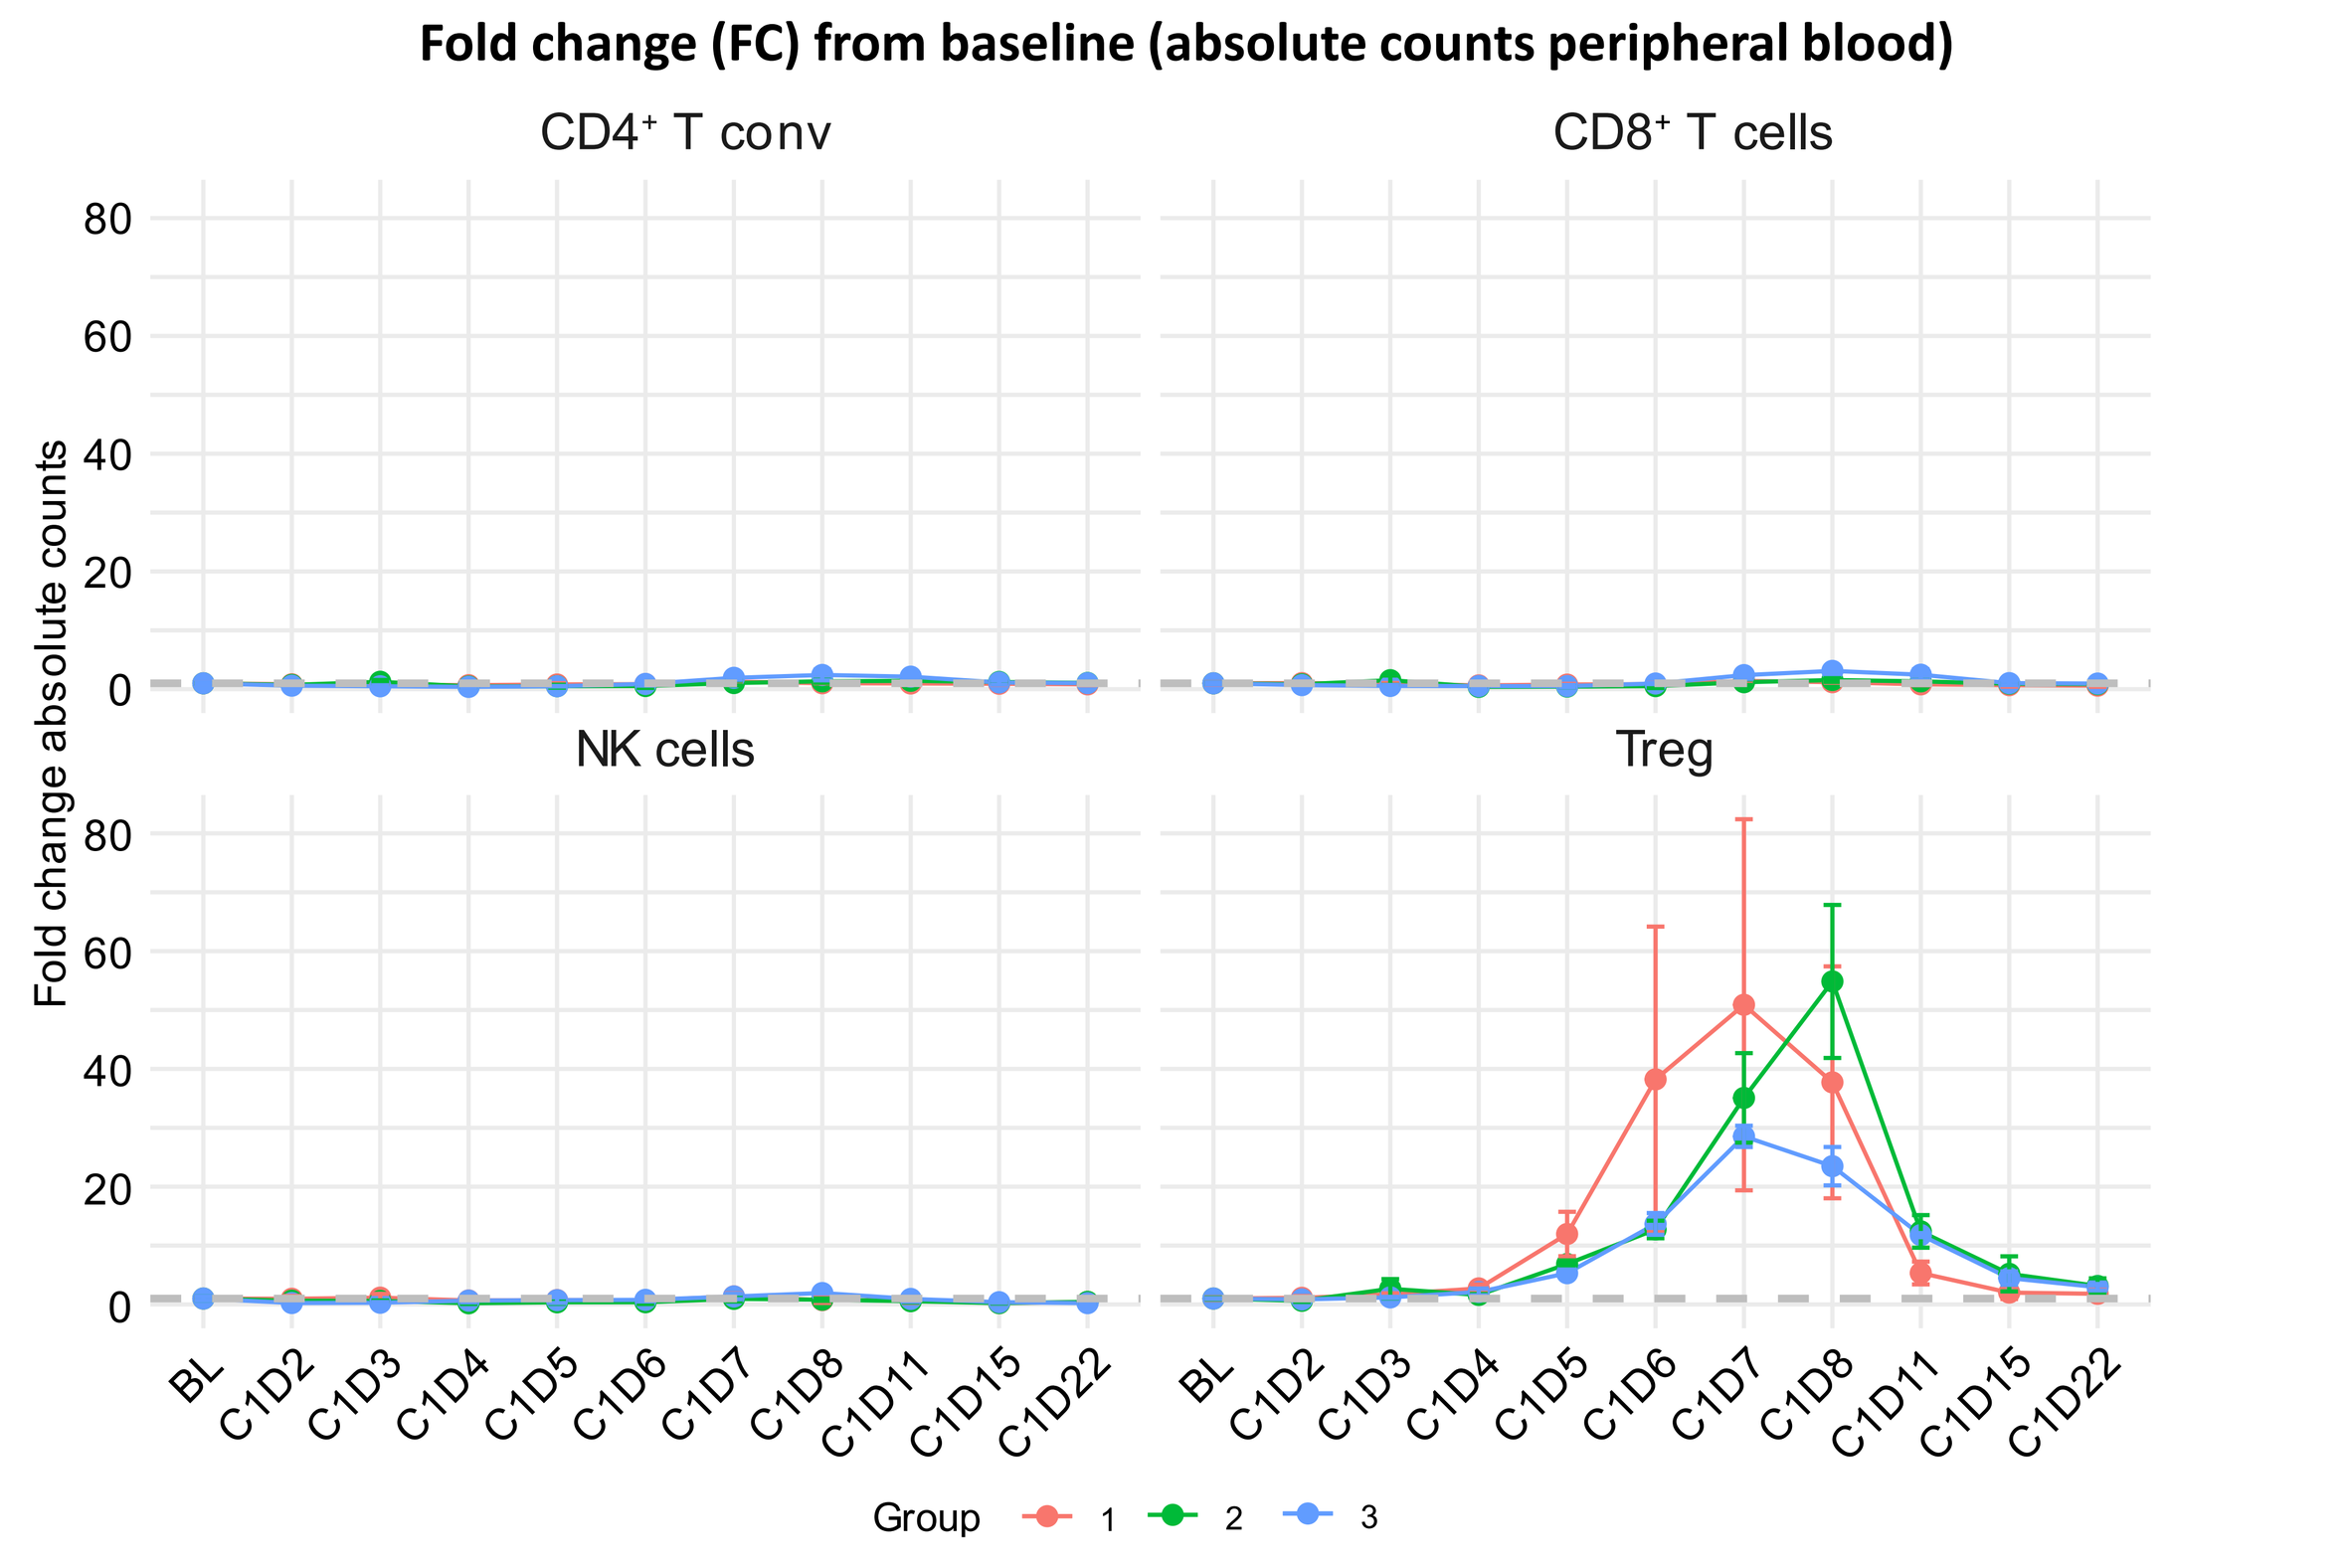
Fold change (FC) from baseline (absolute counts peripheral blood)
CD4⁺ T conv
CD8⁺ T cells
NK cells
Treg
Fold change absolute counts
BL
C1D2
C1D3
C1D4
C1D5
C1D6
C1D7
C1D8
C1D11
C1D15
C1D22
Group
1
2
3

Figure 5: Normalizing lymphocyte counts to baseline reveals selective ~60-fold expansion of Tregs (but not other lineages) in BPT264 dosed animals.

CONCLUSIONS

- BPT264 is a uniquely α-enhanced/β-dead half-life extended IL-2, generated using our novel chemical protein synthesis technology with best-in-class properties.
- We developed a flow cytometry data analysis pipeline allowing to support BPT264 *in vivo* PK/PD studies.
- The R based workflow enables reproducible and scalable analysis of immuno-PD data confirming PoC in NHP.
- Algorithm-aide immuno-PD analysis suggests a selective ~60-fold Treg expansion in NHP with no effect on CD8⁺ T, NK cells and CD4⁺ T_{conv} .
- This flow cytometry data analysis pipeline is not restricted to NHP PK/PD experiments and is now widely used at Bright Peak to support large molecule development.

REFERENCES

1. https://brightpeaktx.com/posters/
2. Clark SM et al, Tox Path, 2012
3. V1.38.2, *via* BioConductor; Ellis B et al, 2022
4. V2.6.0, *via* BioConductor; Van Gassen S et al, Cytometry A, 2015
5. V1.0.7, Kassambara A et al, 2020
6. V1.0.1, Wickham H et al, 2022
7. V0.2.9, McInnes L et al, arxiv, 2018
8. V3.4.0, Wickham H, Springer, 2016
9. V1.5.1; Hope RM 2022

ABOUT BRIGHT PEAK

Bright Peak is a privately held biotechnology company based in Basel, Switzerland and San Diego, CA. We are rapidly advancing a robust portfolio of next generation, multifunctional, immunotherapies for the treatment of patients with cancer and autoimmune disease. We accomplish this by leveraging our world class protein engineering capabilities and our unique cell-free technology platform to chemically synthesize and conjugate novel protein therapeutics that reflect state-of-the-art insights into cytokine and immune checkpoint biology. Our pipeline stretches from discovery to IND-enabling and encompasses enhanced cytokines, antibody-cytokine conjugates and other novel formats. Bright Peak is funded by a syndicate of leading healthcare investors.