Generation of a Highly Potent, *Cis*-signaling PD1-IL2 Immunocytokine Using a Novel Chemical Synthesis and Conjugation Platform

Jean-Philippe Carralot, M. Arévalo Ruiz, R. Alvarez Sanchez, R. Tam, L. Gremlich, A. Chi, V. R. Pattabiraman, B. Kreft
Bright Peak Therapeutics Inc., Basel, Switzerland & San Diego, CA. Email: science@brightpeaktx.com

Poster #1195
10) Immuno-Conjugates and Chimeric Molecules
c. Bispecific Molecules

INTRODUCTION

- Immunocytokines (IC) simultaneously engage distinct and complementary mechanisms of action using antibodies to deliver cytokine payloads to the surface of the same immune cell ("*cis*-signaling") resulting in enhanced anti-tumor activity.[1]
- To generate ICs, we developed an entirely novel approach based on site-specific, chemical conjugation of engineered cytokines to existing, unmodified antibodies (Ab).[1]
- We utilized LZM-009, an anti-PD-1 Ab in late-stage clinical development, for generation of proprietary multi-functional PD-1 targeted ICs in order to inhibit the dominant immune checkpoint pathway and target PD-1+ antigen-experienced T cells.
- Using our unique cell-free, chemical engineering platform, we generated a PD1-IL2 IC (BPT331) to target an α-dead IL-2 to tumor resident PD-1high effector T cells while simultaneously releasing these cells from PD-1-mediated immune suppression.

CONCLUSIONS

- Our ability to rapidly generate ICs from existing Abs enables highly efficient screening of cytokine payloads and antibody-cytokine combinations.
- We identified an optimized α-dead/β-enhanced IL-2 payload to generate BPT331, a potent PD1-IL2 with high selectivity for PD-1high cells.
- *In vivo*, BPT331 triggers the selective expansion of PD-1high effector memory CD8+ T (TEM) cells within tumors.
- Specific expansion of PD-1high CD8+ TEMs in the tumor vs blood and tumor-specific retention of PD1-IL2 but not of non-targeted Her2-IL2 provide a rationale for tumor selectivity of BPT331.
- BPT331 exhibits potent *in vivo* efficacy in multiple tumor models including those resistant to anti-PD-1 treatment.
- BPT331 *in vivo* efficacy is likely achieved by simultaneous blockade of PD-1 and enhanced IL-2 signaling *in cis*.

REFERENCE

1. Carralot *et al.*, proceedings of the annual meeting AACR 2022 ; New Orleans. Abstract #4223.

UNIQUE TECHNOLOGY PLATFORM

Rapid Generation of Immunocytokines via Chemical Conjugation

Anti-human PD-1 Ab LZM-009

- **High affinity** human IgG4

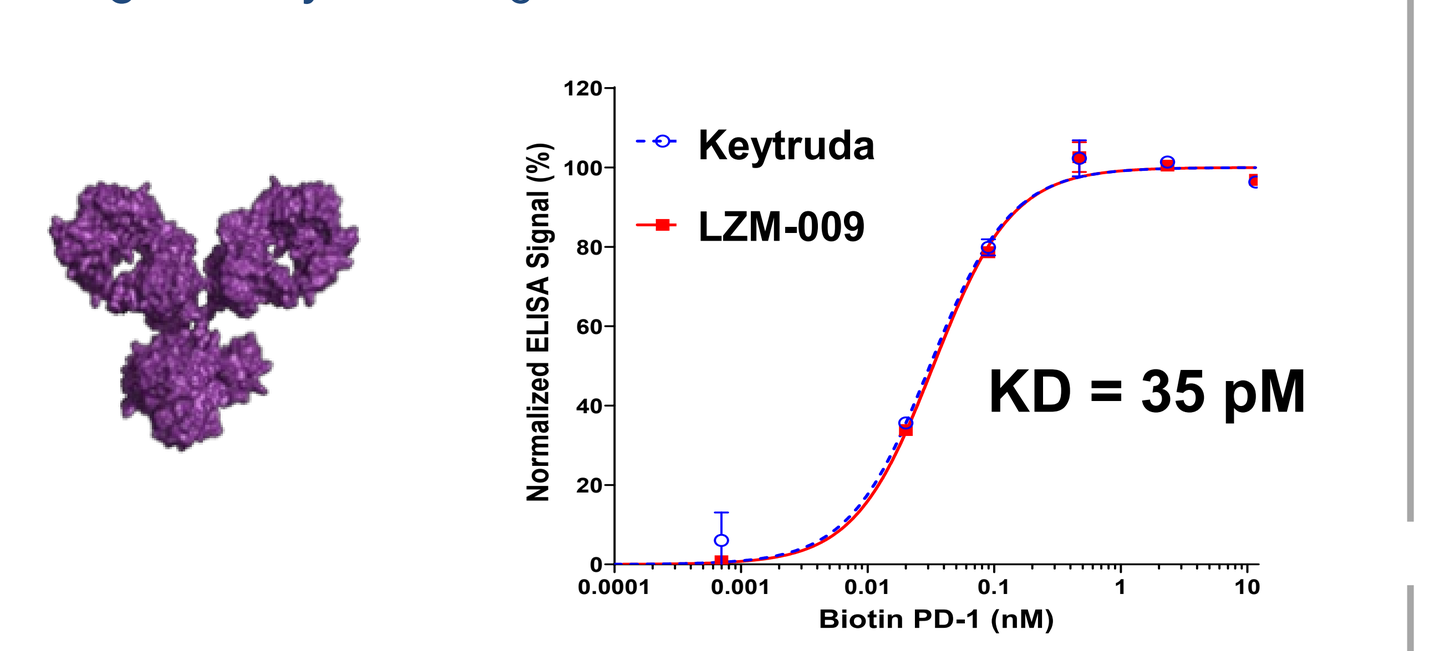
Keytruda
LZM-009
KD = 35 pM
Normalized ELISA Signal (%)
Biotin PD-1 (nM)

Figure 1. ELISA assay measuring binding to human PD-1.

Synthetic α-Dead/β-Enhanced IL-2

- Potency reduced in T_{regs} and enhanced in CD8+ T-cells
- Conjugatable via **"click" chemistry**

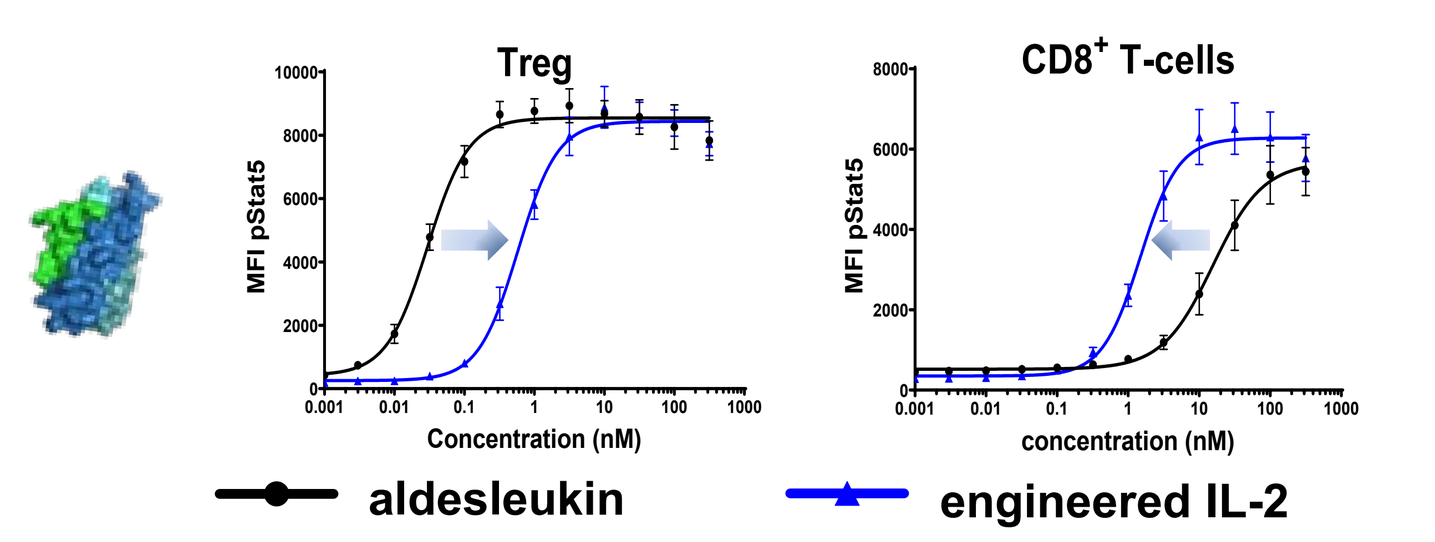
Treg
CD8+ T-cells
MFI pStat5
Concentration (nM)
aldesleukin
engineered IL-2

Figure 2. STAT5 phosphorylation in primary human CD8+ Teff and Tregs.

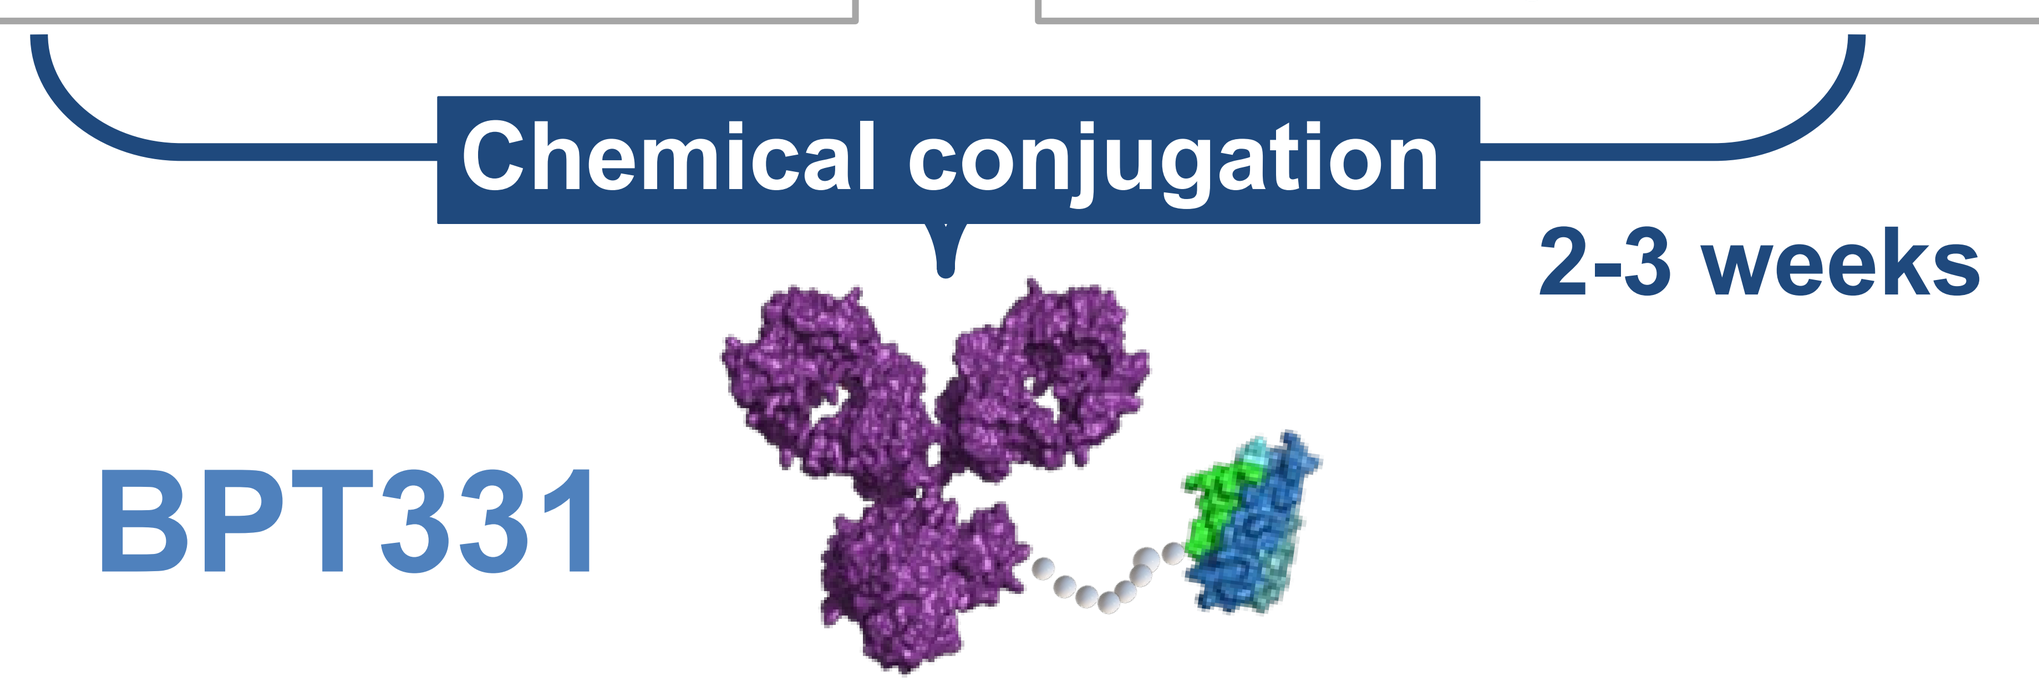
Chemical conjugation
2-3 weeks
BPT331

- ➔ "Off-the-shelf" human IgG antibodies, **no antibody engineering** required.
- ➔ **Site-specific conjugation** to defined lysine residue within Fc domain.
- ➔ Controlled drug-antibody ratio (DAR).
- ➔ No significant impact on antibody or cytokine (payload) functional properties.

PHARMACODYNAMIC EFFECTS

BPT331 Specifically Expands PD-1high Intratumoral Effector Memory CD8+ T Cells (CD8+ TEM)

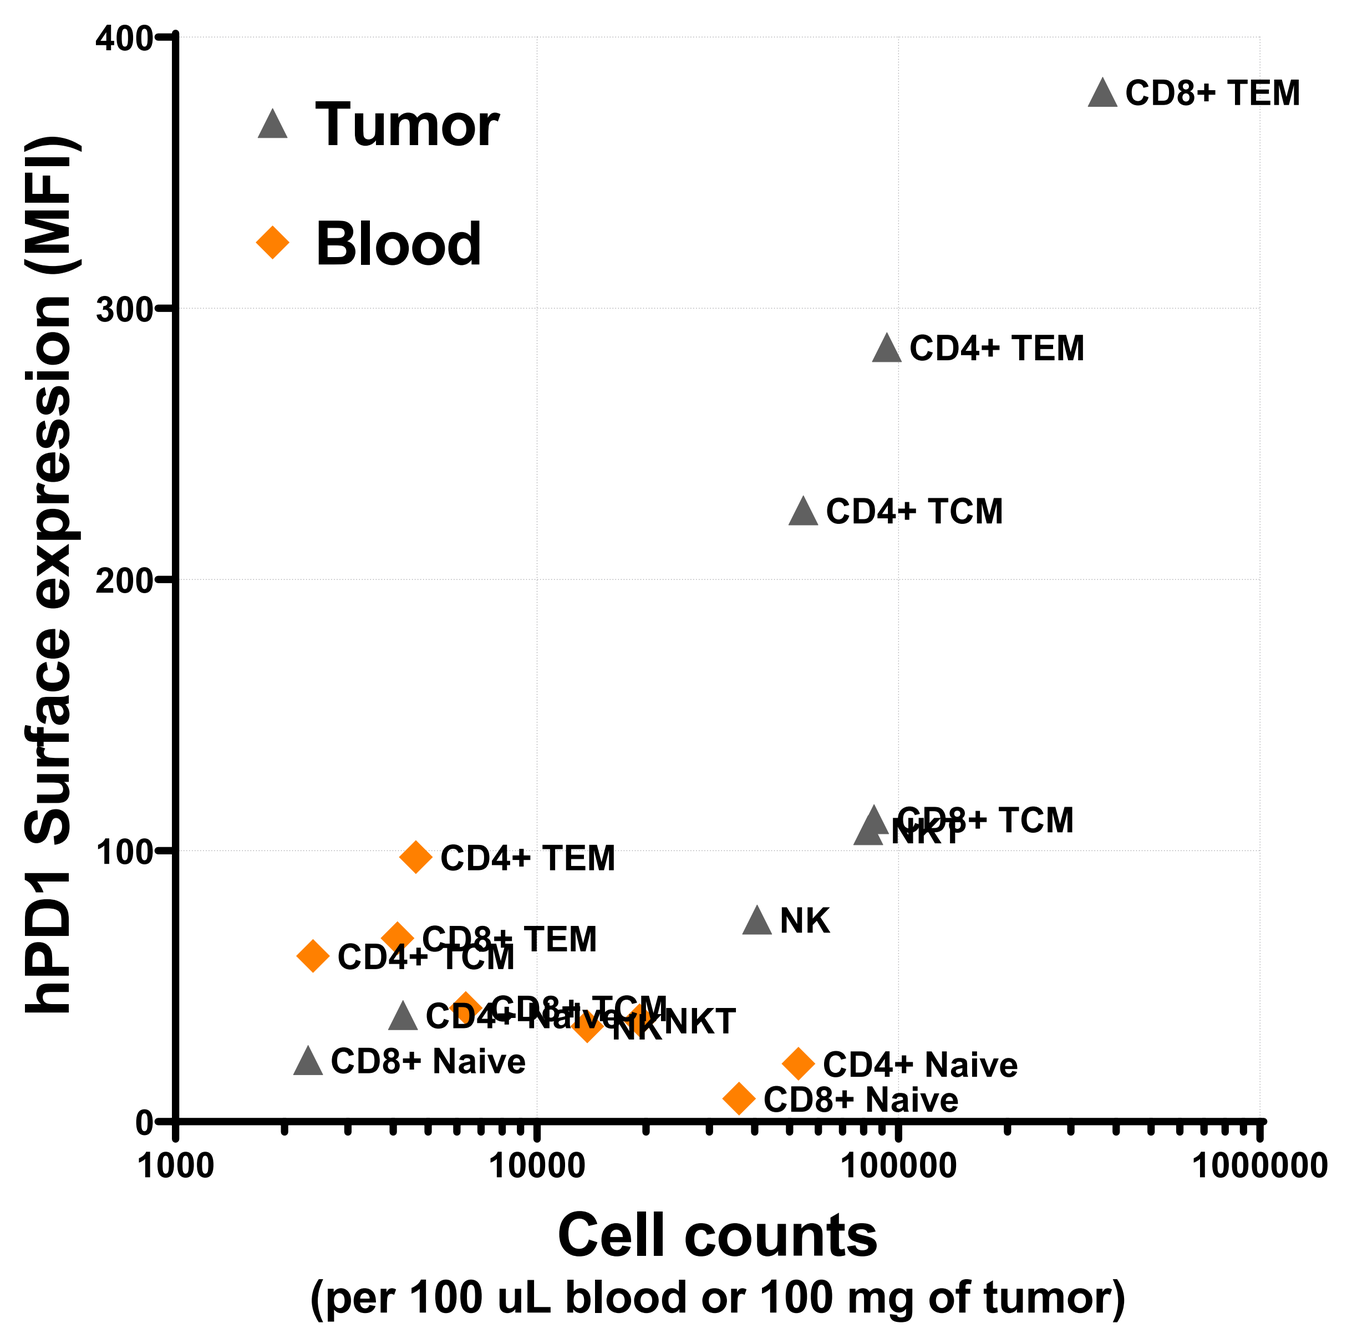
Tumor
Blood
hPD1 Surface expression (MFI)
Cell counts
(per 100 uL blood or 100 mg of tumor)
CD8+ TEM
CD4+ TEM
CD4+ TCM
CD8+ TCM
NKT
NK
CD8+ Naive
CD4+ Naive

Figure 4. Counts of immune cells and their respective PD-1 surface expression levels in blood vs. MC38 tumors in huPD-1 transgenic C57BL/6 mice. *(average of 8 control animals)*

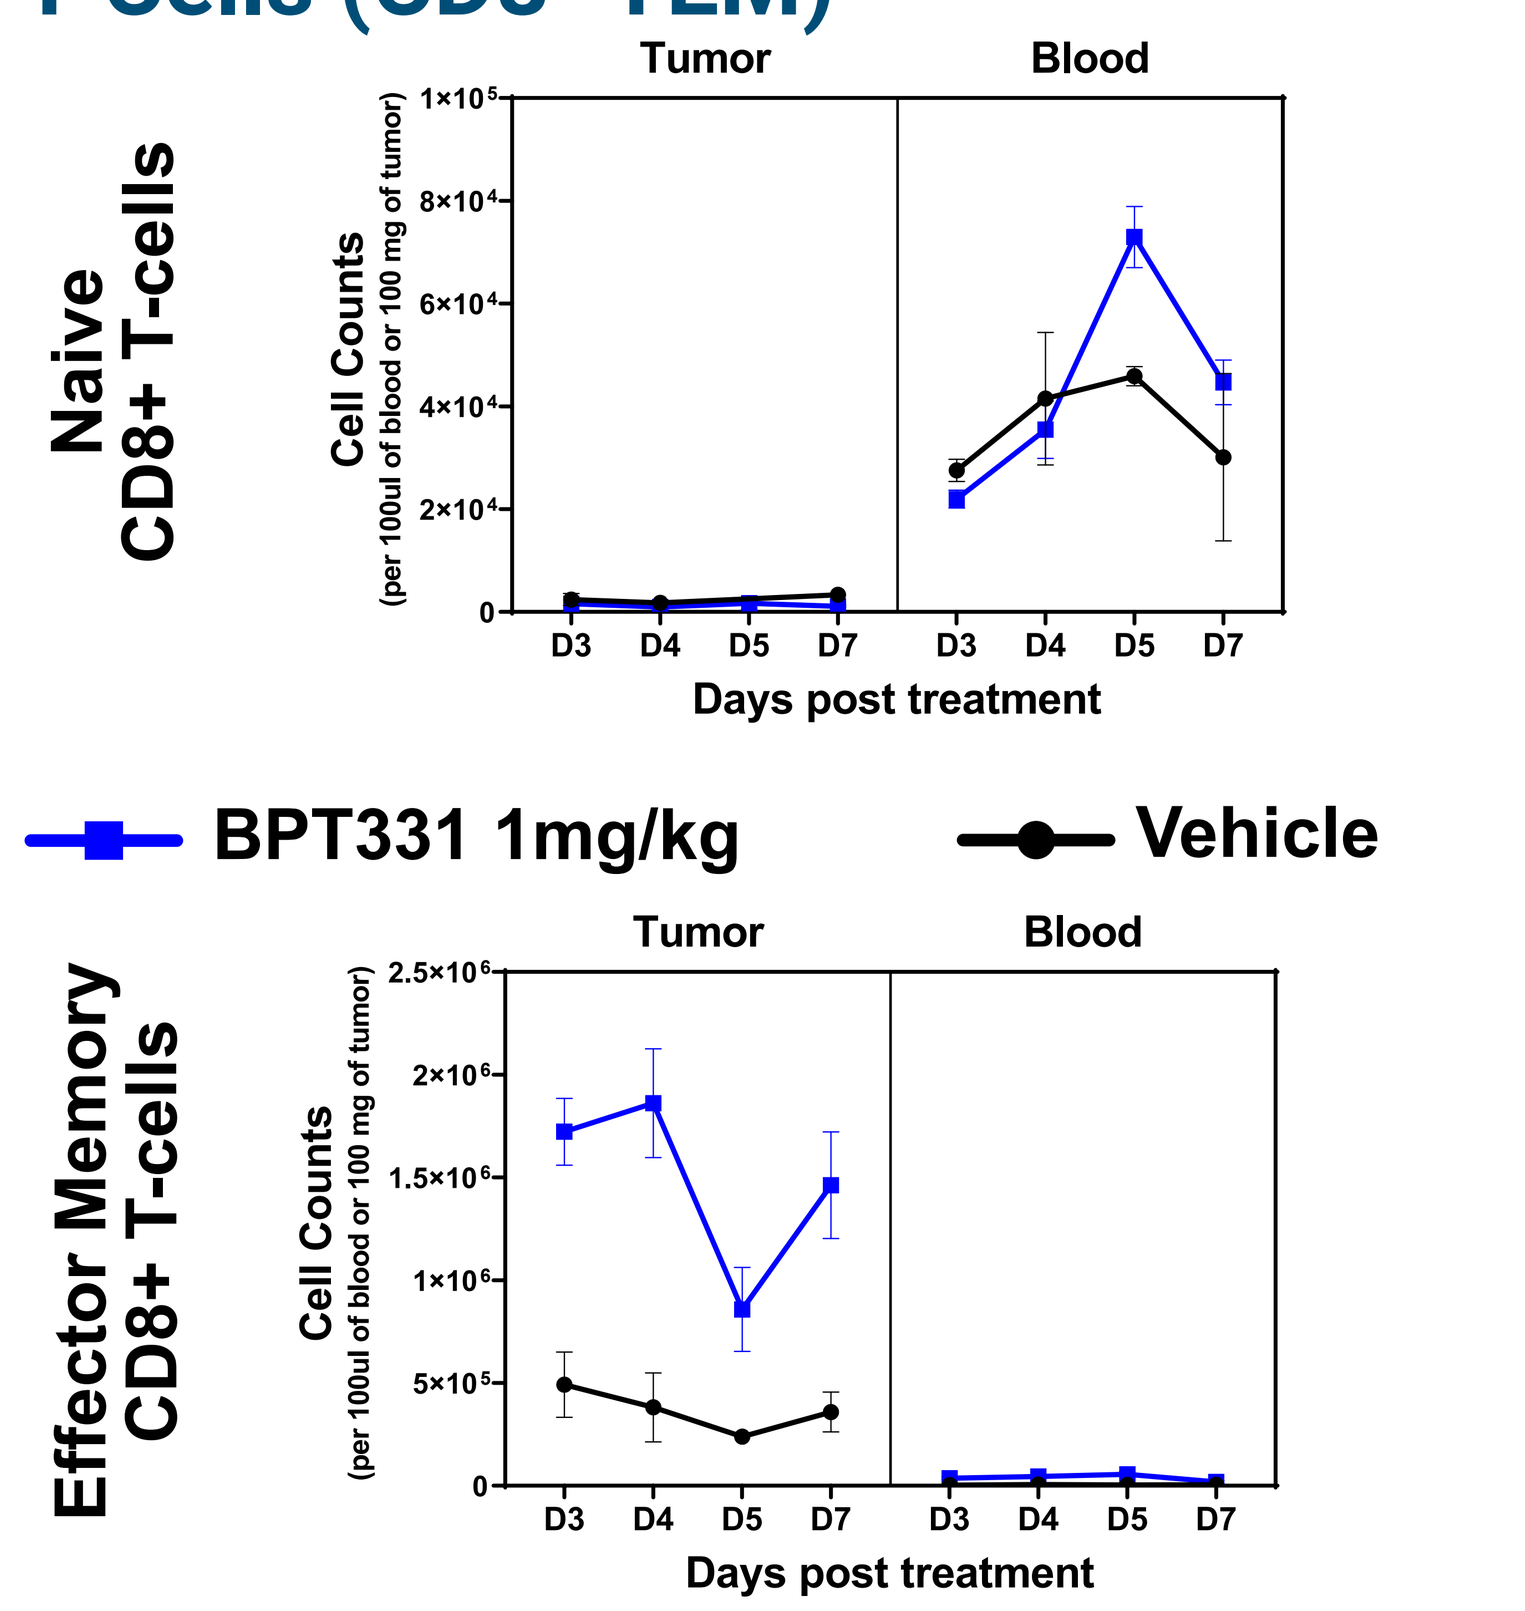
Naive CD8+ T-cells
Effector Memory CD8+ T-cells
Tumor
Blood
Cell Counts
Days post treatment
BPT331 1mg/kg
Vehicle

Figure 5. Absolute counts of naïve (CD3+CD8+CD44lowCD62Lhigh) and effector memory (CD3+CD8+CD44highCD62Llow) CD8+ T cells in MC38 tumor-bearing C57BL/6 mice expressing huPD-1 (single dose of BPT331 @ 0.3 mg/kg i.v., average ± SEM of 2-3 mice)

- ➔ MC38 tumors are **highly infiltrated with PD-1high** CD8+ TEM.
- ➔ BPT331 **specifically expands PD-1high CD8+ TEM within the tumor** with minimal effect on blood CD8+.

IN VIVO EFFICACY

BPT331 Exhibits Potent Anti-Tumor Efficacy That is Superior to the Combination of anti-PD-1 Ab Plus Non-Targeted Her2-IL2

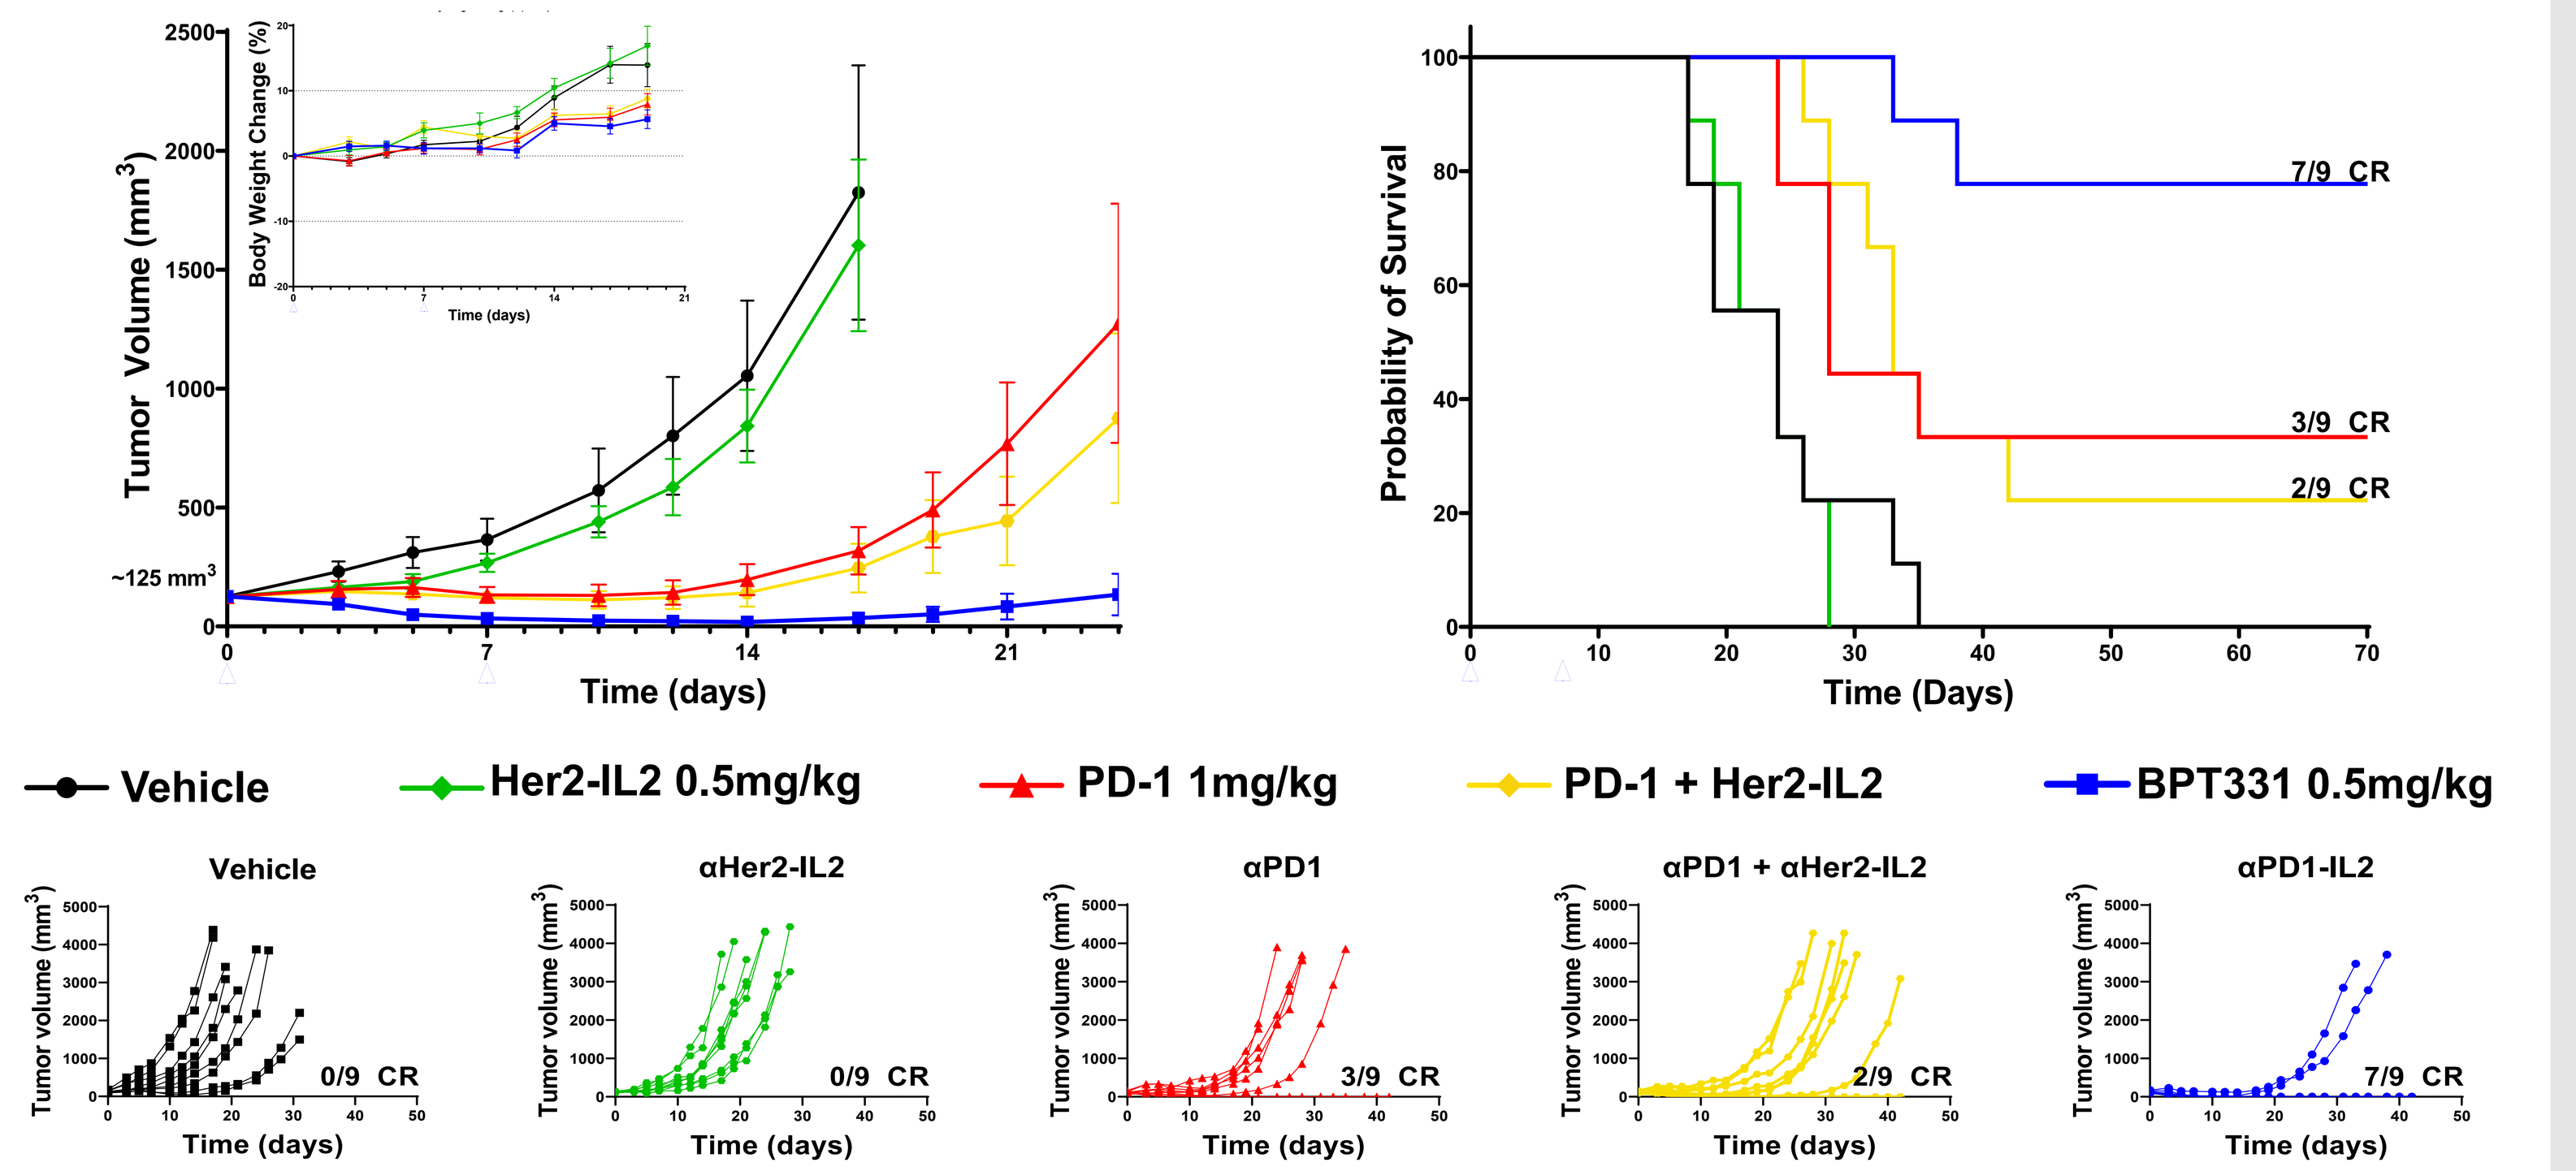
Tumor Volume (mm3)
Body Weight Change (%)
Time (days)
Probability of Survival
Time (Days)
7/9 CR
3/9 CR
2/9 CR
Vehicle
Her2-IL2 0.5mg/kg
PD-1 1mg/kg
PD-1 + Her2-IL2
BPT331 0.5mg/kg
αHer2-IL2
αPD1
αPD1 + αHer2-IL2
αPD1-IL2
0/9 CR

Figure 7. MC38 tumor-bearing C57BL/6 mice expressing huPD-1 were treated with the indicated molecules (QWx2, i.v., average ± SEM of 9 mice per group; CR: Complete Response)

- ➔ **BPT331 is highly potent** (78% CR) and **well tolerated** in mice.
- ➔ Efficacy of **BPT331 is superior to anti-PD-1 Ab**, to non-targeted Her2-IL2, and to the combination of these two agents.
- ➔ Enhanced efficacy of BPT331 appears to be **achieved by simultaneous blockade of PD-1 and enhanced IL-2 signaling *in cis*.**

RATIONAL DESIGN

IL-2 Payload Screening and Selection to Maximize PD-1-Dependent *Cis*-Signaling

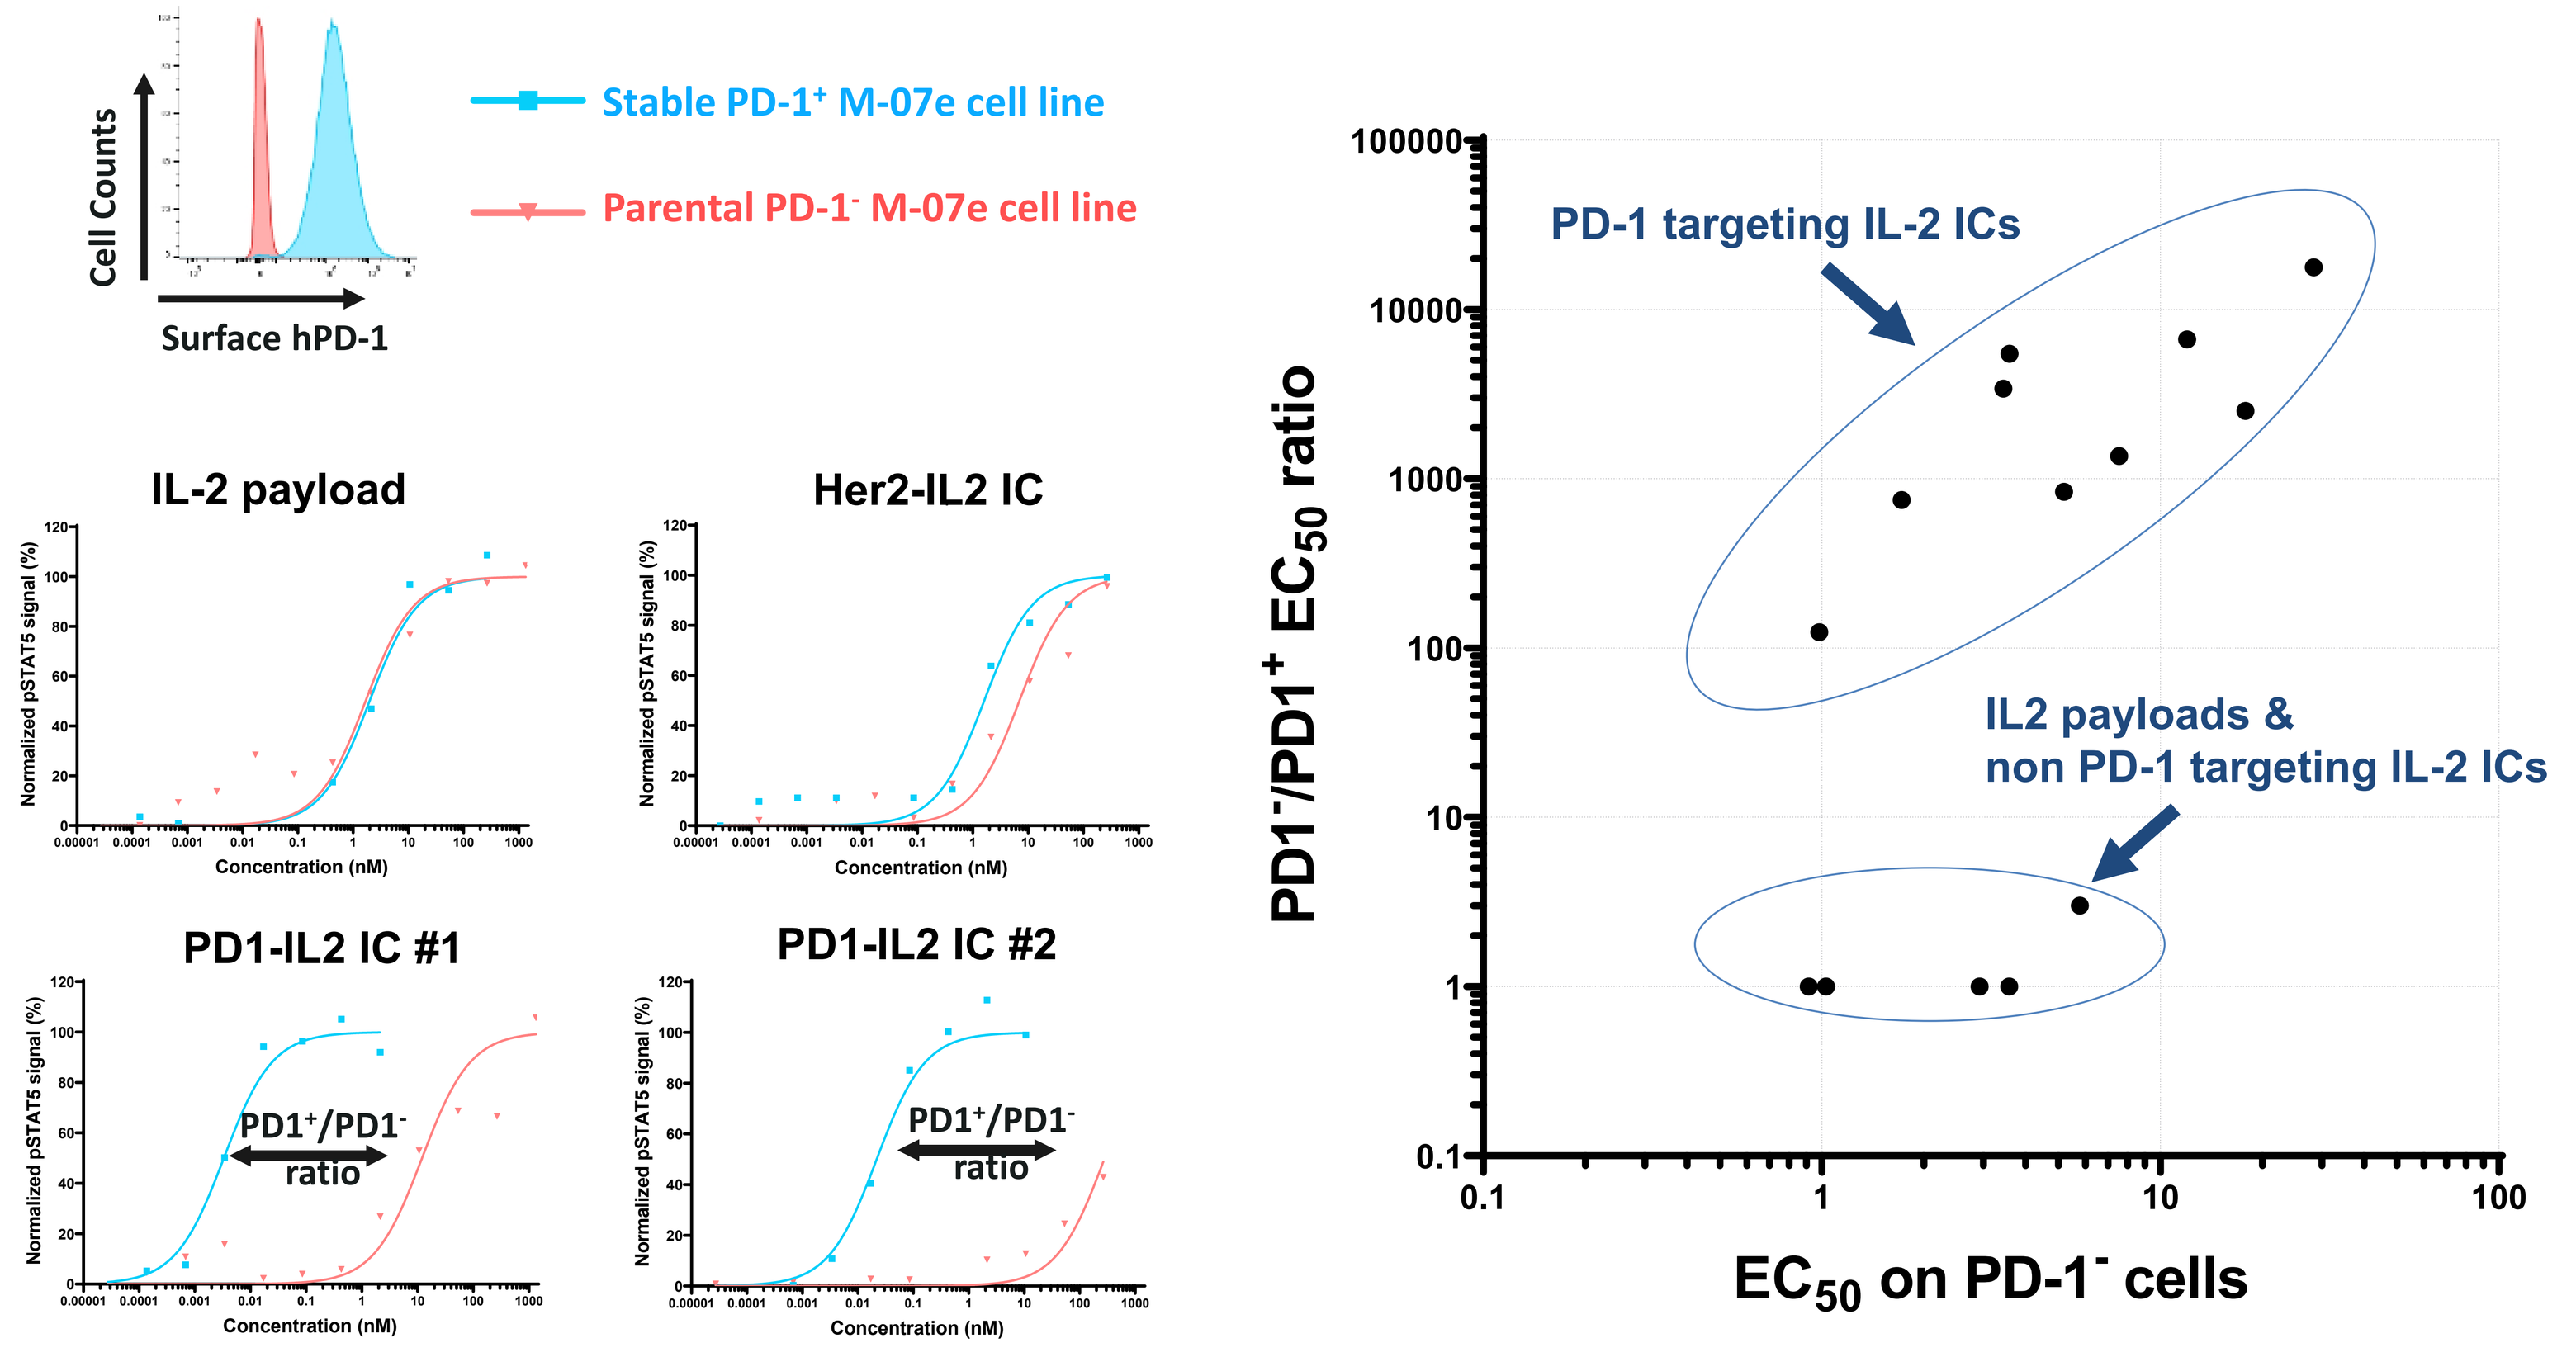
Cell Counts
Surface hPD-1
Stable PD-1+ M-07e cell line
Parental PD-1- M-07e cell line
IL-2 payload
Her2-IL2 IC
PD1-IL2 IC #1
PD1-IL2 IC #2
Normalized pSTAT5 signal (%)
Concentration (nM)
PD1+/PD1- ratio
PD-1 targeting IL-2 ICs
IL2 payloads &
non PD-1 targeting IL-2 ICs
PD1-/PD1+ EC50 ratio
EC50 on PD-1- cells

Figure 3. STAT5 phosphorylation in PD-1- vs. PD-1+ M-07e cells.

- ➔ Platform enables **rapid screening of IL-2 payloads** in the context of a PD1-IL2.
- ➔ Results in **optimal Ab-payload combination** for enhanced selectivity & potency.
- ➔ PD1-IL2 exhibits **>500-fold selectivity** for PD-1+ cells.

PHARMACOKINETICS

PD1-IL2 is Efficiently and Selectively Retained in Tumors vs Plasma and vs a Non-Targeted Her2-IL2

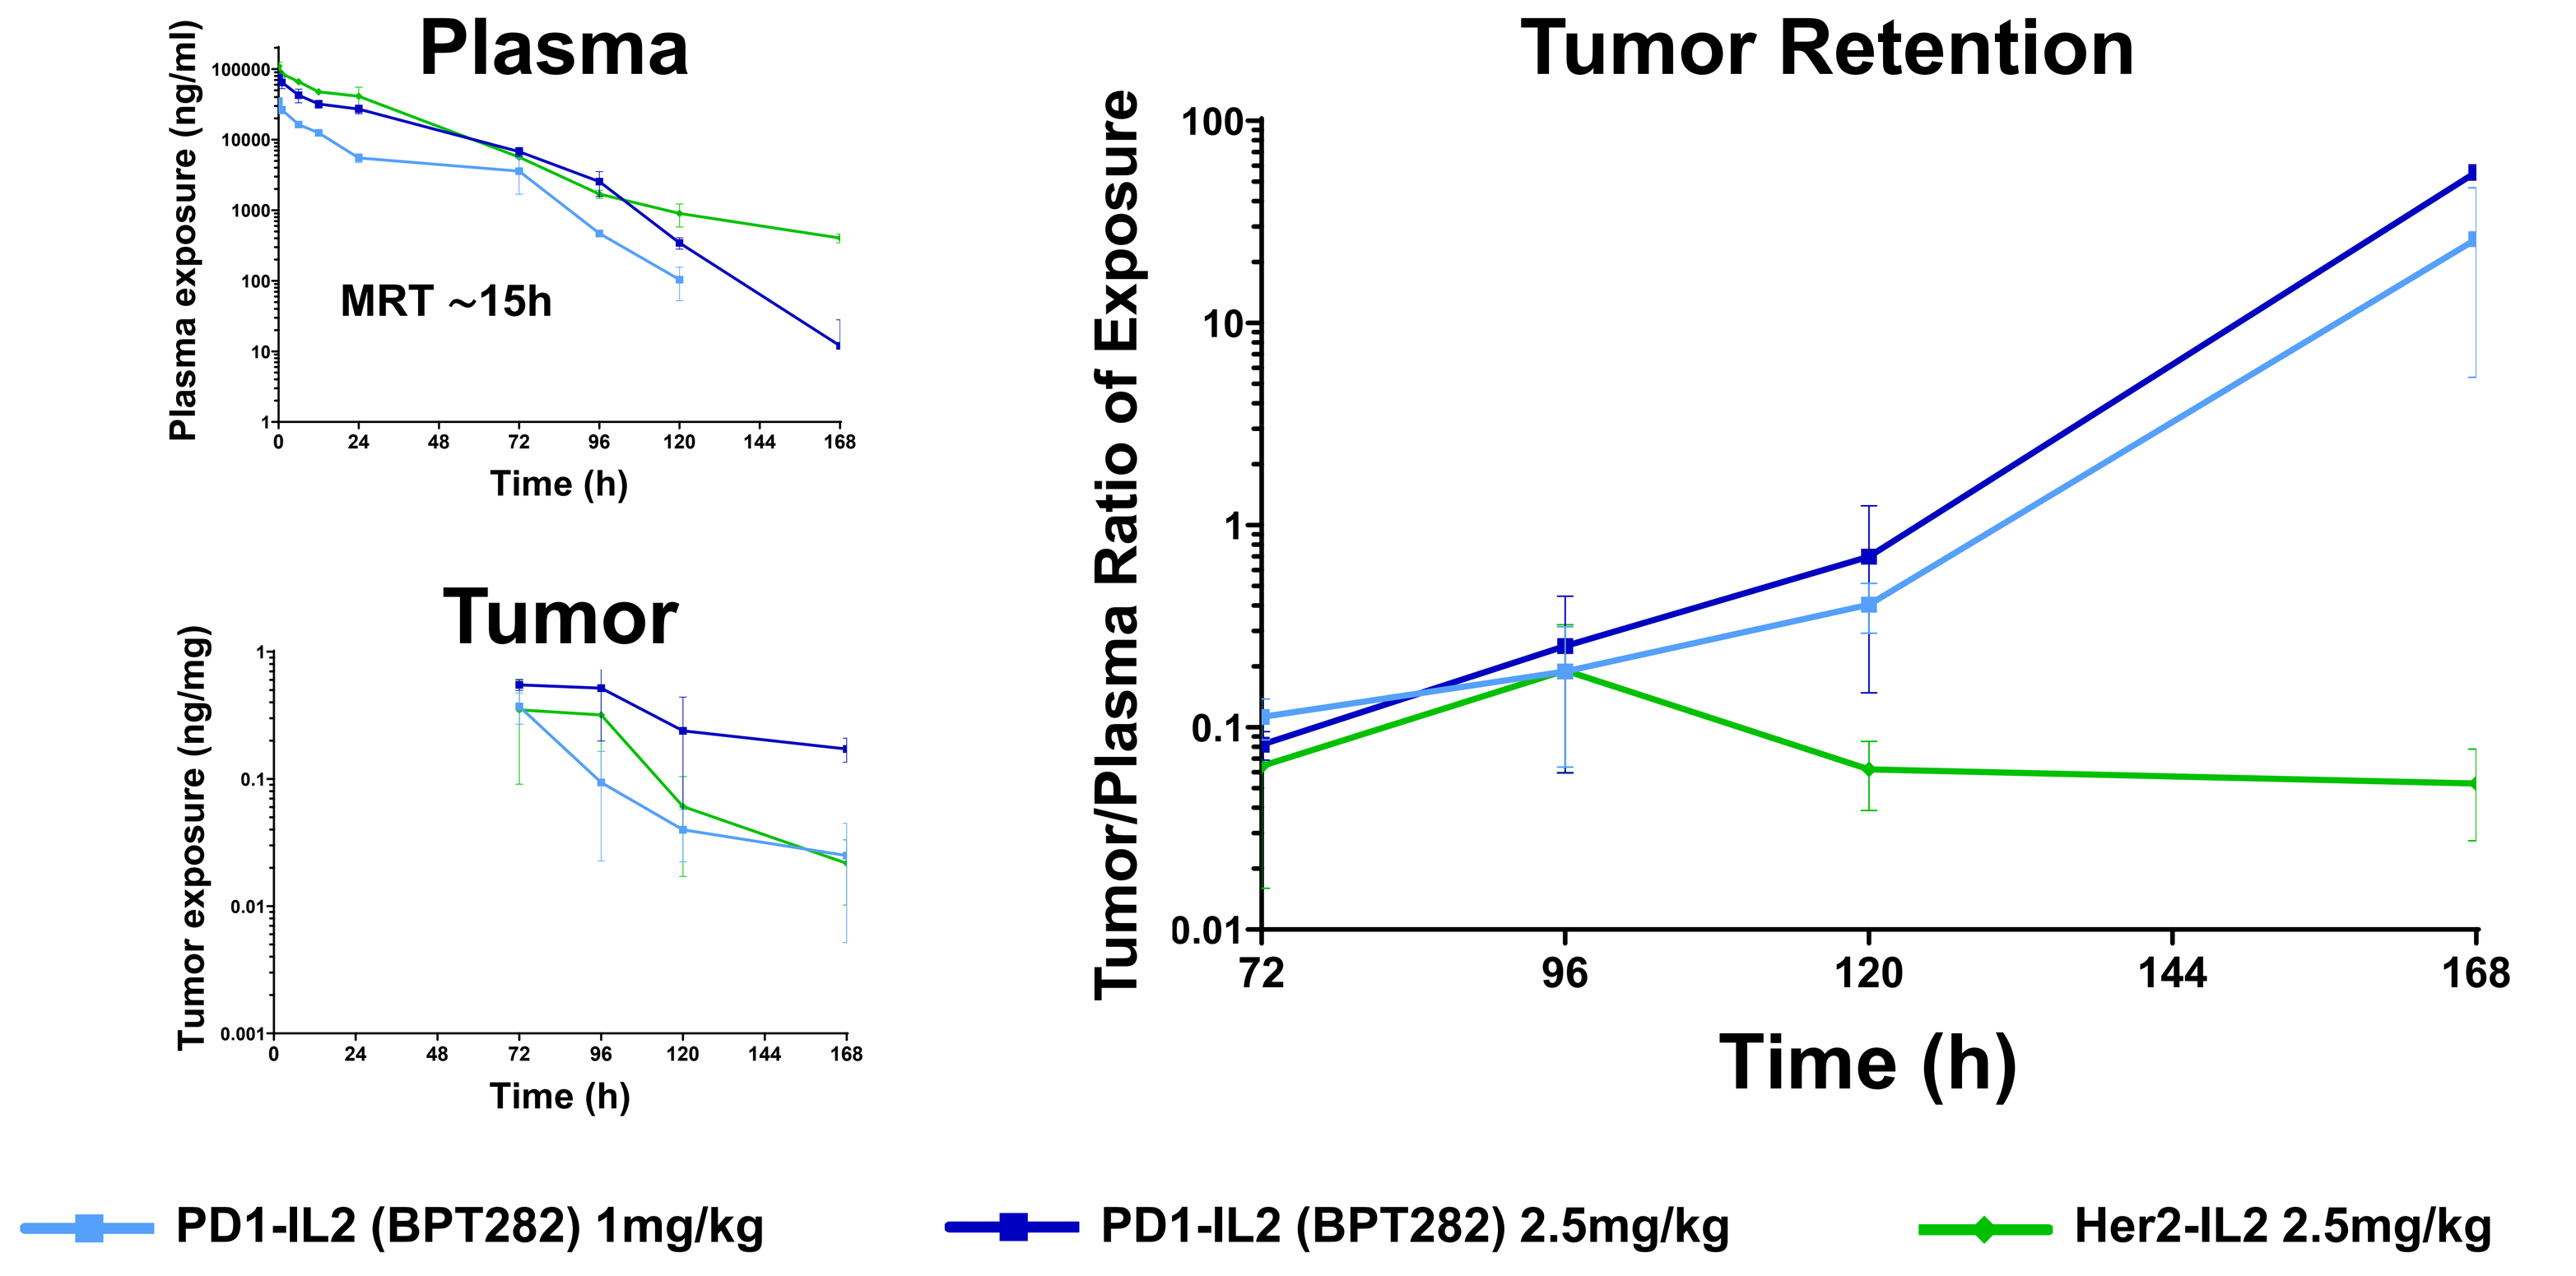
Plasma
Plasma exposure (ng/ml)
MRT ~15h
Time (h)
Tumor
Tumor exposure (ng/mg)
Tumor Retention
Tumor/Plasma Ratio of Exposure
Time (h)
PD1-IL2 (BPT282) 1mg/kg
PD1-IL2 (BPT282) 2.5mg/kg
Her2-IL2 2.5mg/kg

Figure 6. Exposure after a single intravenous (i.v.) injection in CT26 tumor-bearing BALB/c mice. (average ± SD of three mice ; MRT: Mean Residence Time)

- ➔ PD1-IL2 and Her2-IL2 show similar clearance in the blood.
- ➔ Over time, **PD1-IL2 but not Her2-IL2 is retained in the tumor**.

EFFICACY IN PD-1 RESISTANT TUMORS

PD1-IL2 Overcomes Resistance to Anti-PD-1 Therapy

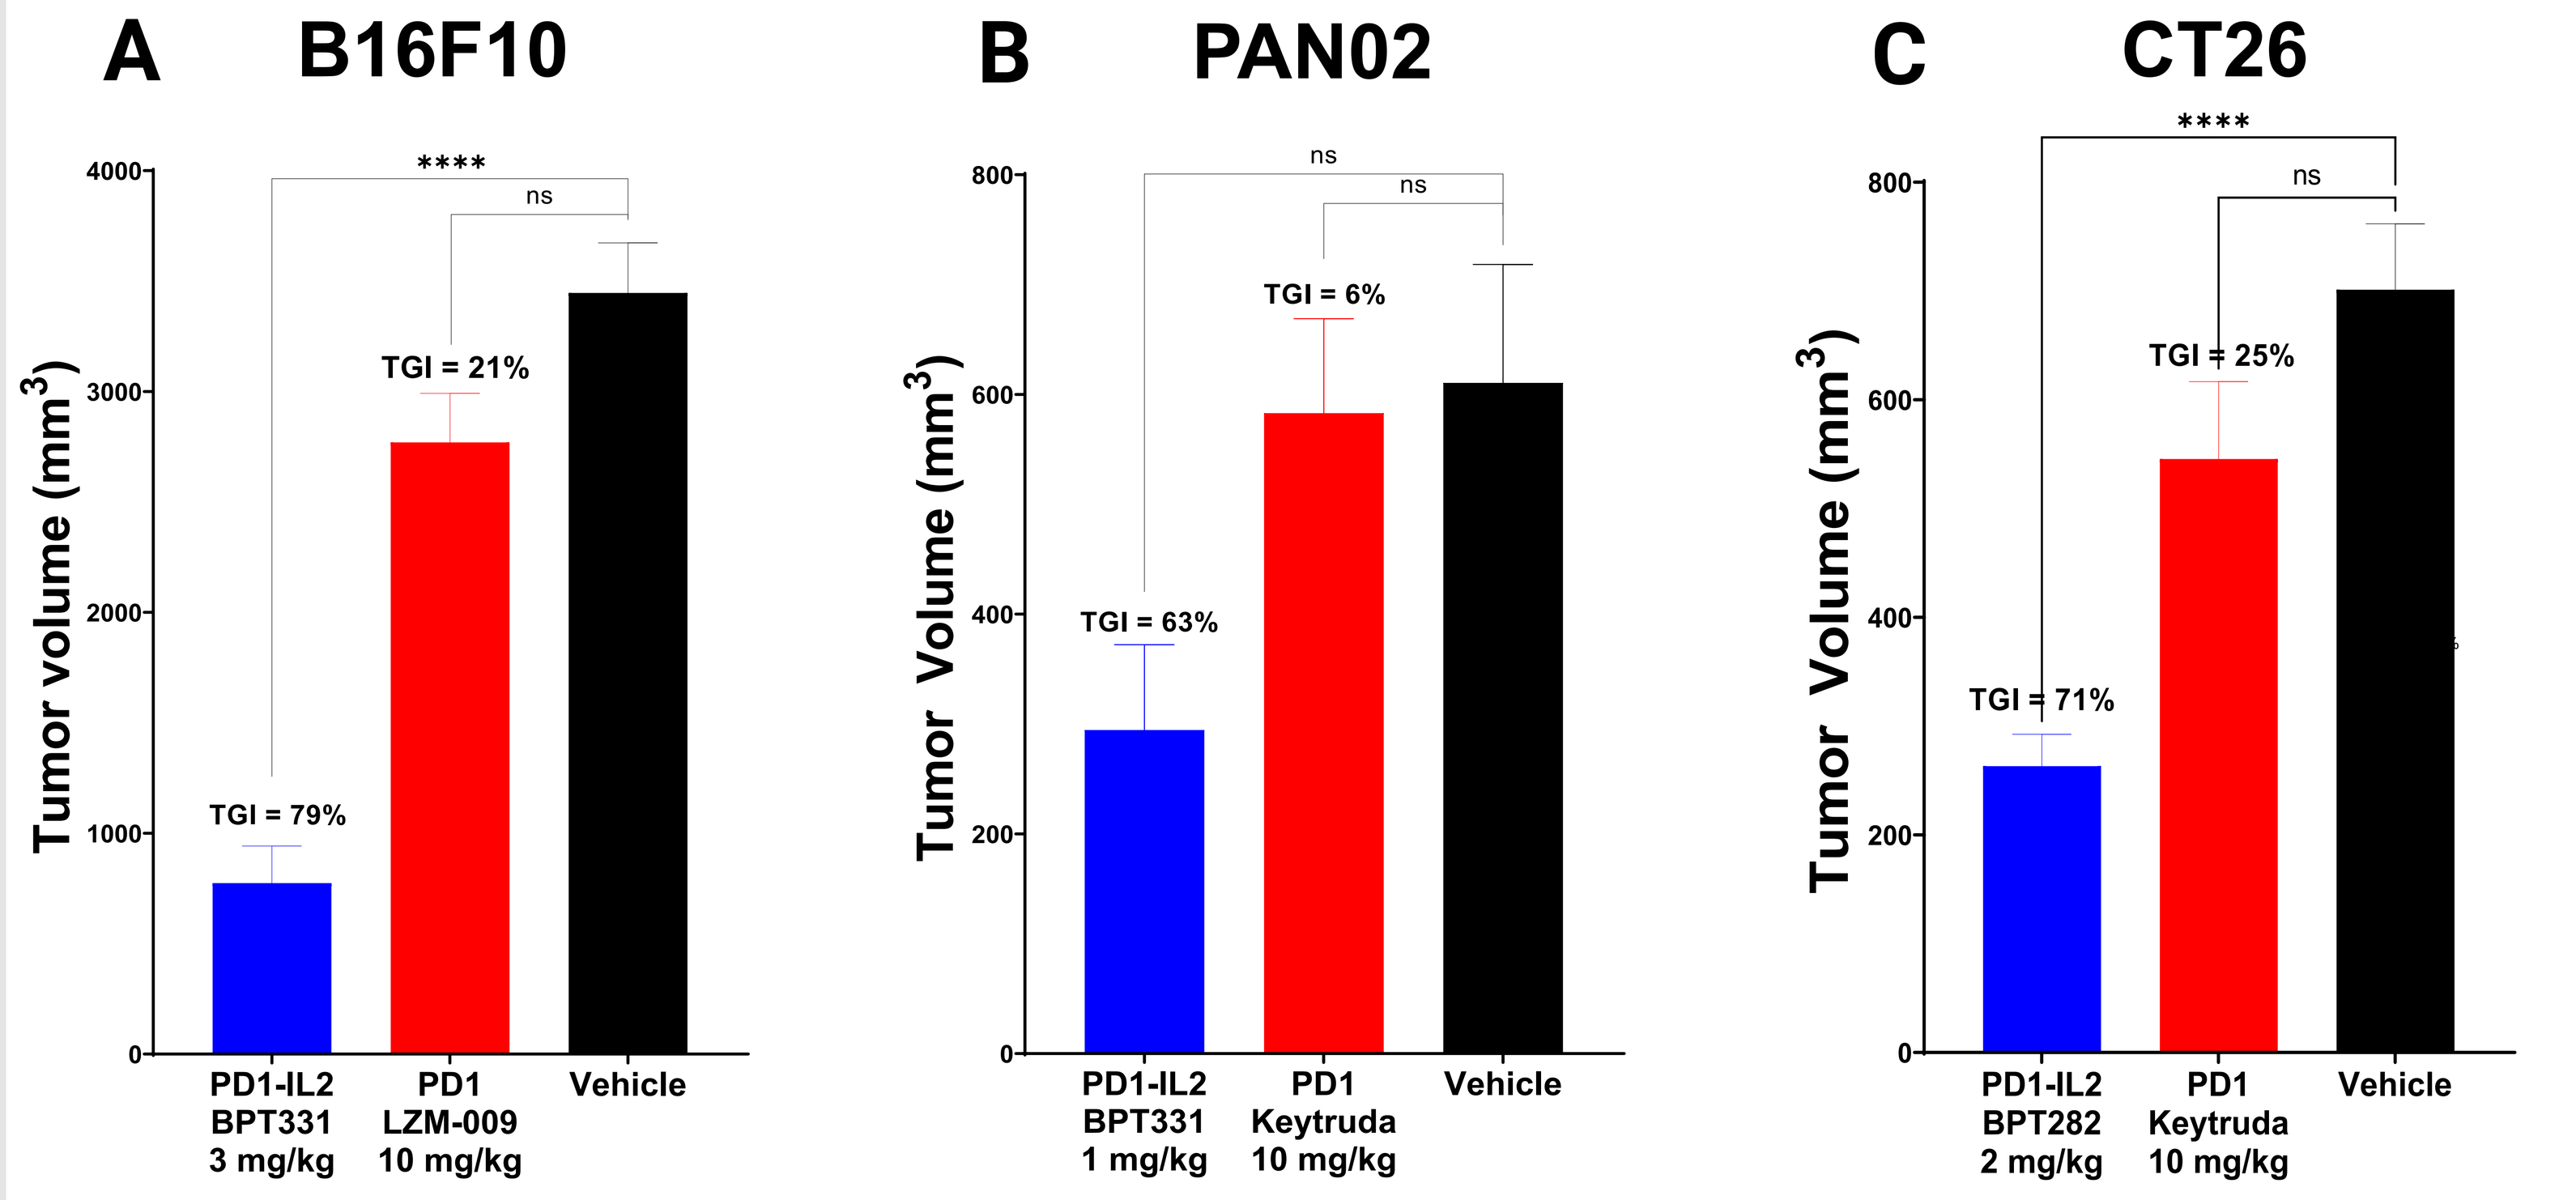
A B16F10
B PAN02
C CT26
Tumor volume (mm3)
TGI = 79%
TGI = 21%
TGI = 63%
TGI = 6%
TGI = 71%
TGI = 25%
PD1-IL2 BPT331 3 mg/kg
PD1 LZM-009 10 mg/kg
Vehicle
PD1-IL2 BPT331 1 mg/kg
PD1 Keytruda 10 mg/kg
PD1-IL2 BPT282 2 mg/kg

ns

Figure 8. Tumor growth inhibition (TGI) in different models grown sub-cutaneously in mice expressing huPD-1. **A** B16F10 melanoma model: tumor size at start of treatment ~75mm^3 ; single dose i.v.; d7 measurement; n=9 mice/group. **B** PAN02 pancreatic model : tumor size at start of treatment ~179mm^3 ; QWx2 i.v.; d14 measurement; n=5 mice/group. **C** CT26 colon carcinoma model: tumor size at start of treatment ~88 mm^3 ; single dose i.v.; d7 measurement; n=9 mice/group. (average ± SEM ; One-way Anova test ; **** p-value<0.0001, ns non-significant)

- ➔ PD1-IL2 shows **significant efficacy in models resistant to anti-PD-1** therapy.
- ➔ Superior efficacy is achieved at **3-10x lower doses compared** to anti-PD-1 Ab.