Cis-activation of PD-1+ effector T cells with dual-targeting immunocytokines generated using a novel chemical conjugation platform

Jean-Philippe Carralot, Matilde Arévalo Ruiz, Robert Tam, Eric Armentani, Vijaya R. Pattabiraman, and Bertolt Kreft
Bright Peak Therapeutics Inc., Basel. Email: science@brightpeaktx.com

BRIGHT PEAK
THERAPEUTICS

ABSTRACT

Immunocytokines (IC) provide the opportunity for targeted delivery of cytokines as payloads to tissues and cells to improve safety and efficacy of cytokine-based therapies. ICs have gained significant interest in recent years and several antibody-cytokine fusion proteins have entered clinical trials. Compared to recombinant fusion proteins, we have developed an entirely different approach to IC generation based on the site-specific, chemical conjugation of synthetic cytokines to antibodies.

Bright Peak generates enhanced and conjugatable cytokines using a novel protein engineering platform based on solid-phase peptide synthesis and subsequent chemical ligation of protein segments. Our synthetic cytokines can then be readily chemically conjugated to specific lysine residues in the Fc region of an existing IgG1, IgG2 or IgG4 antibody without the need for prior antibody engineering. Chemical conjugation of cytokine payloads is rapid, enabling the flexible generation of ICs based on different antibodies and payloads within weeks. We applied our technology to more than 10 antibodies and found that neither antigen binding nor payload potency and selectivity are affected by chemical conjugation. Importantly, binding of the Fc domain of ICs to Fc gamma receptors or FcRn is not significantly affected.

We are initially focusing on the development of PD-1-targeted ICs to achieve dual-targeting of PD-1+ effector T cells (cis-signaling). Using several anti-PD-1 antibodies and various synthetic IL-2 variants as payloads, we created ICs with different drug-antibody ratios and explored alternative conjugation sites within the Fc region. Resulting PD-1/IL-2 ICs are highly active showing significantly enhanced potency due to avidity resulting from binding of the cytokine to PD-1+ effector T cells in cis. Our PD-1/IL-2 ICs induce strong pharmacodynamic effects *in vivo*, and we are currently optimizing the pharmacological profiles of PD-1-targeted ICs for clinical application. In addition, we are actively exploring cis-signaling ICs targeting different surface receptors and immune cells.

Poster #4223
PO.IM02.17: Immunomodulatory Agents and Interventions 3
Section 38
AACR American Association for Cancer Research

PLATFORM

Unique alliance of complementary technologies

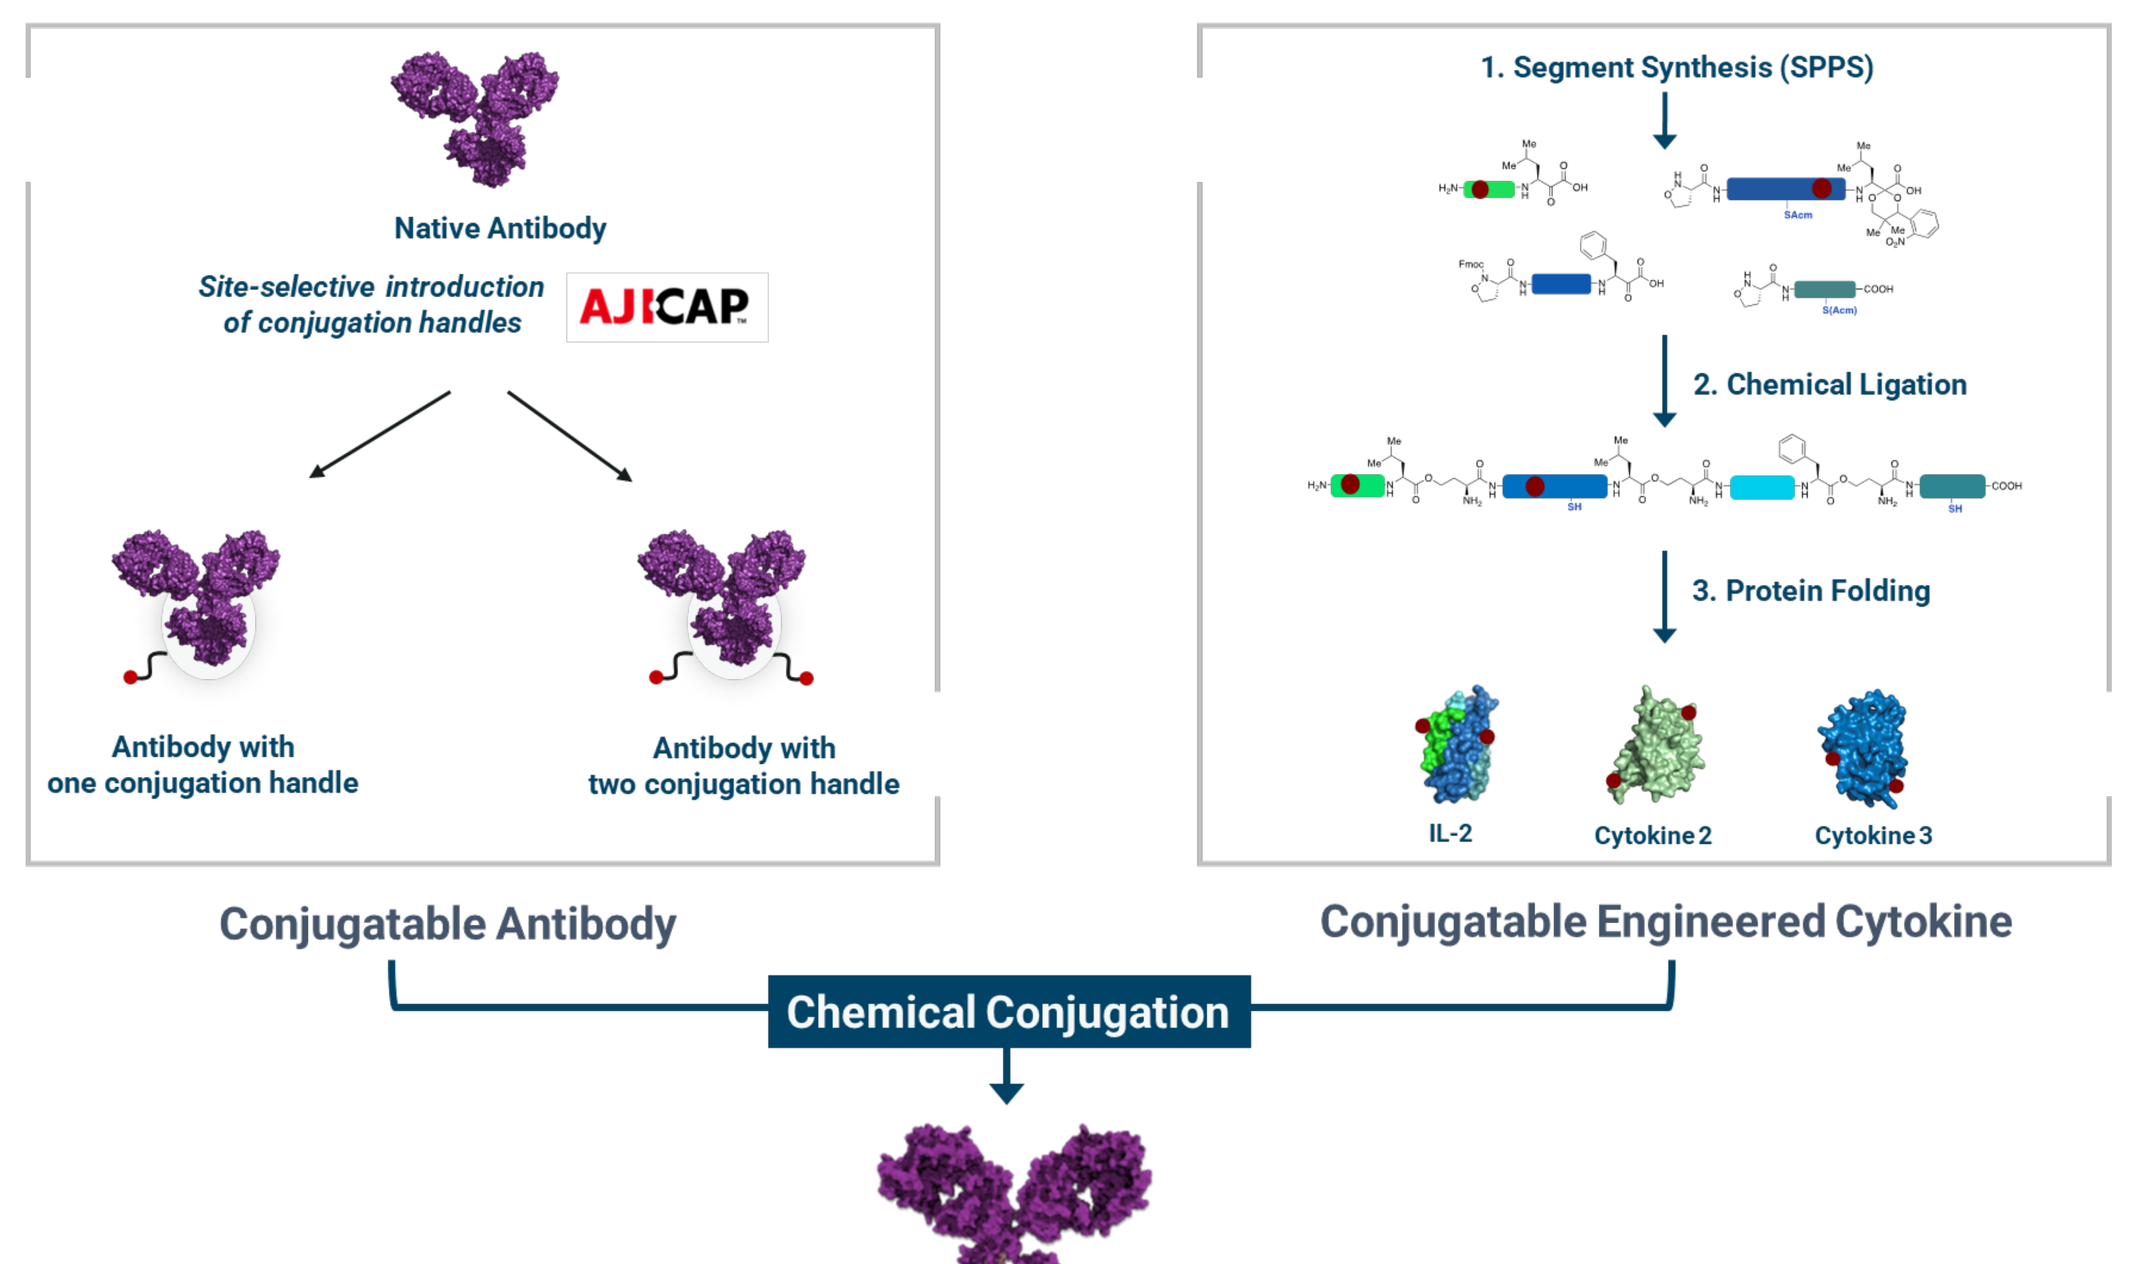
Native Antibody
Site-selective introduction of conjugation handles
AJICAP
Antibody with one conjugation handle
Antibody with two conjugation handle
Conjugatable Antibody
1. Segment Synthesis (SPPS)
2. Chemical Ligation
3. Protein Folding
IL-2
Cytokine 2
Cytokine 3
Conjugatable Engineered Cytokine
Chemical Conjugation

- ➔ First successful use of AJICAP™ ADC technology to conjugate proteins to antibodies
- ➔ Site-specific conjugation to defined lysine residue within Fc domain yielding either DAR1 or DAR2 ICs
- ➔ Cell-free process with high purity & yield

VERSATILITY

Plug-and-play technology enables rapid and flexible generation of Immunocytokines

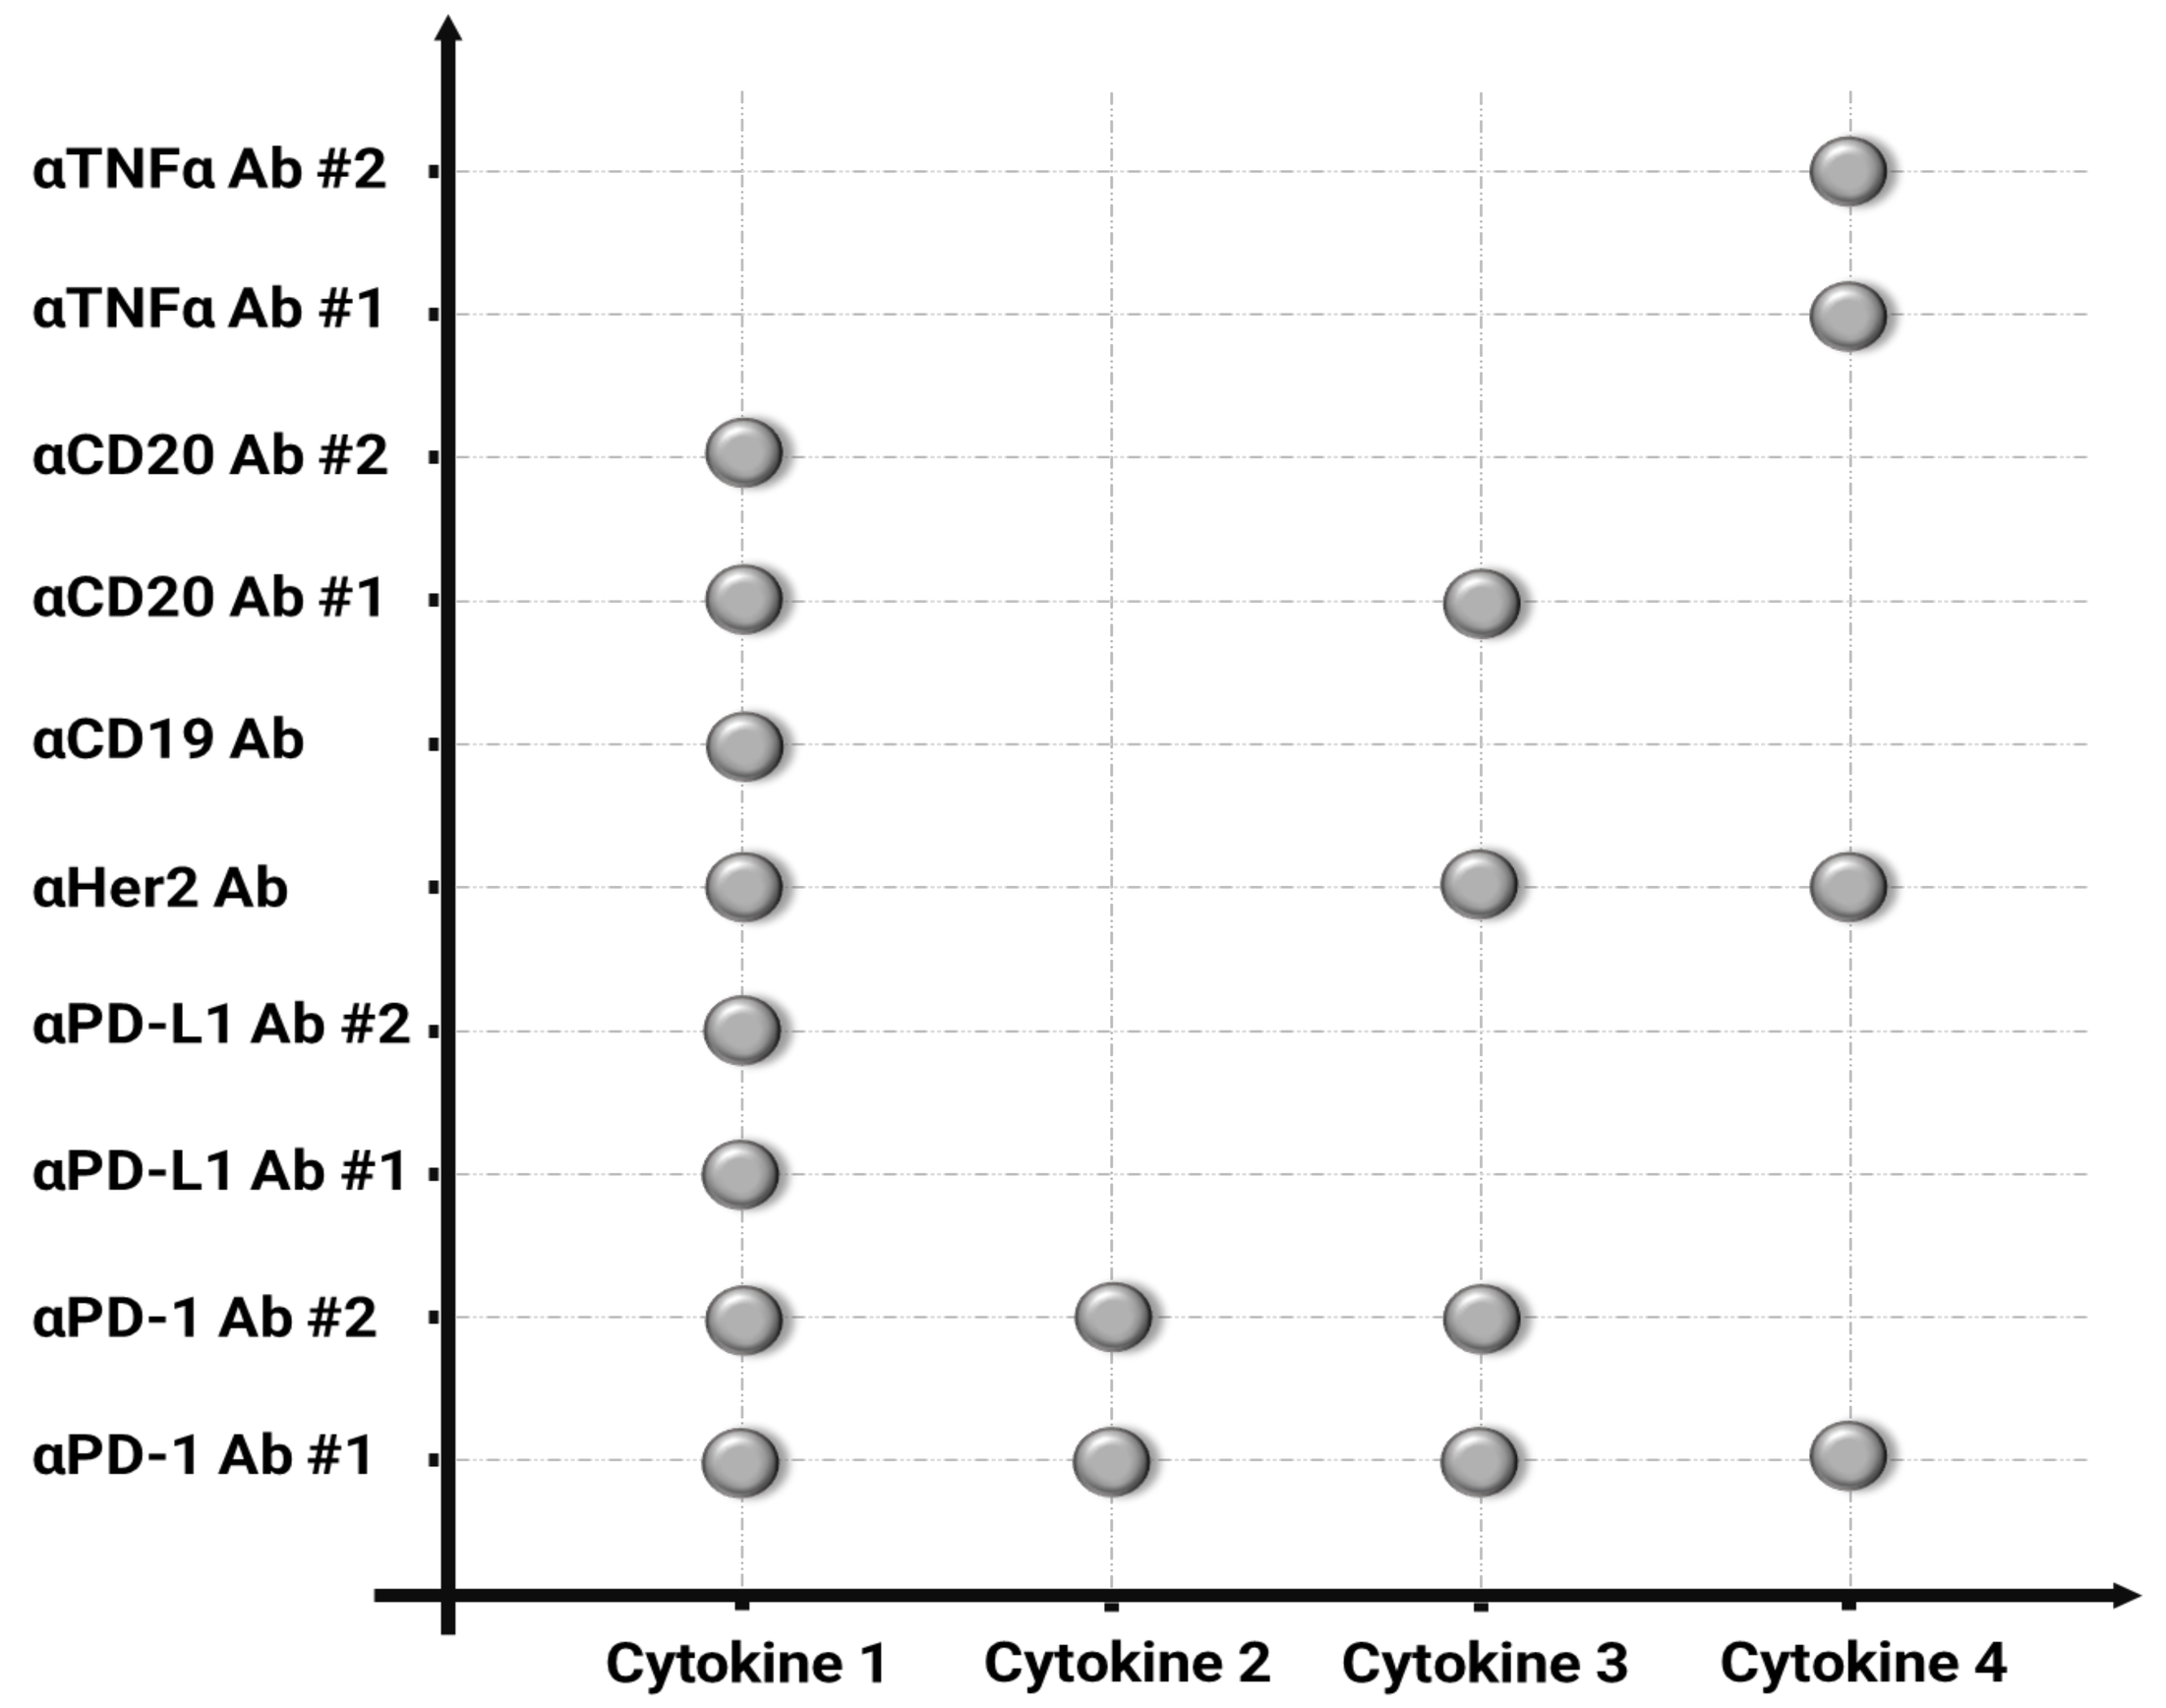
αTNFα Ab #2
αTNFα Ab #1
αCD20 Ab #2
αCD20 Ab #1
αCD19 Ab
αHer2 Ab
αPD-L1 Ab #2
αPD-L1 Ab #1
αPD-1 Ab #2
αPD-1 Ab #1
Cytokine 1
Cytokine 2
Cytokine 3
Cytokine 4

- ➔ Off-the-shelf solution, no antibody engineering required
- ➔ Allows conjugation to human IgG1, IgG2 and IgG4
- ➔ IC generation within just 2-3 weeks

ANTIBODY FUNCTIONS

Antigen binding is not affected by cytokine conjugation to the Fc domain

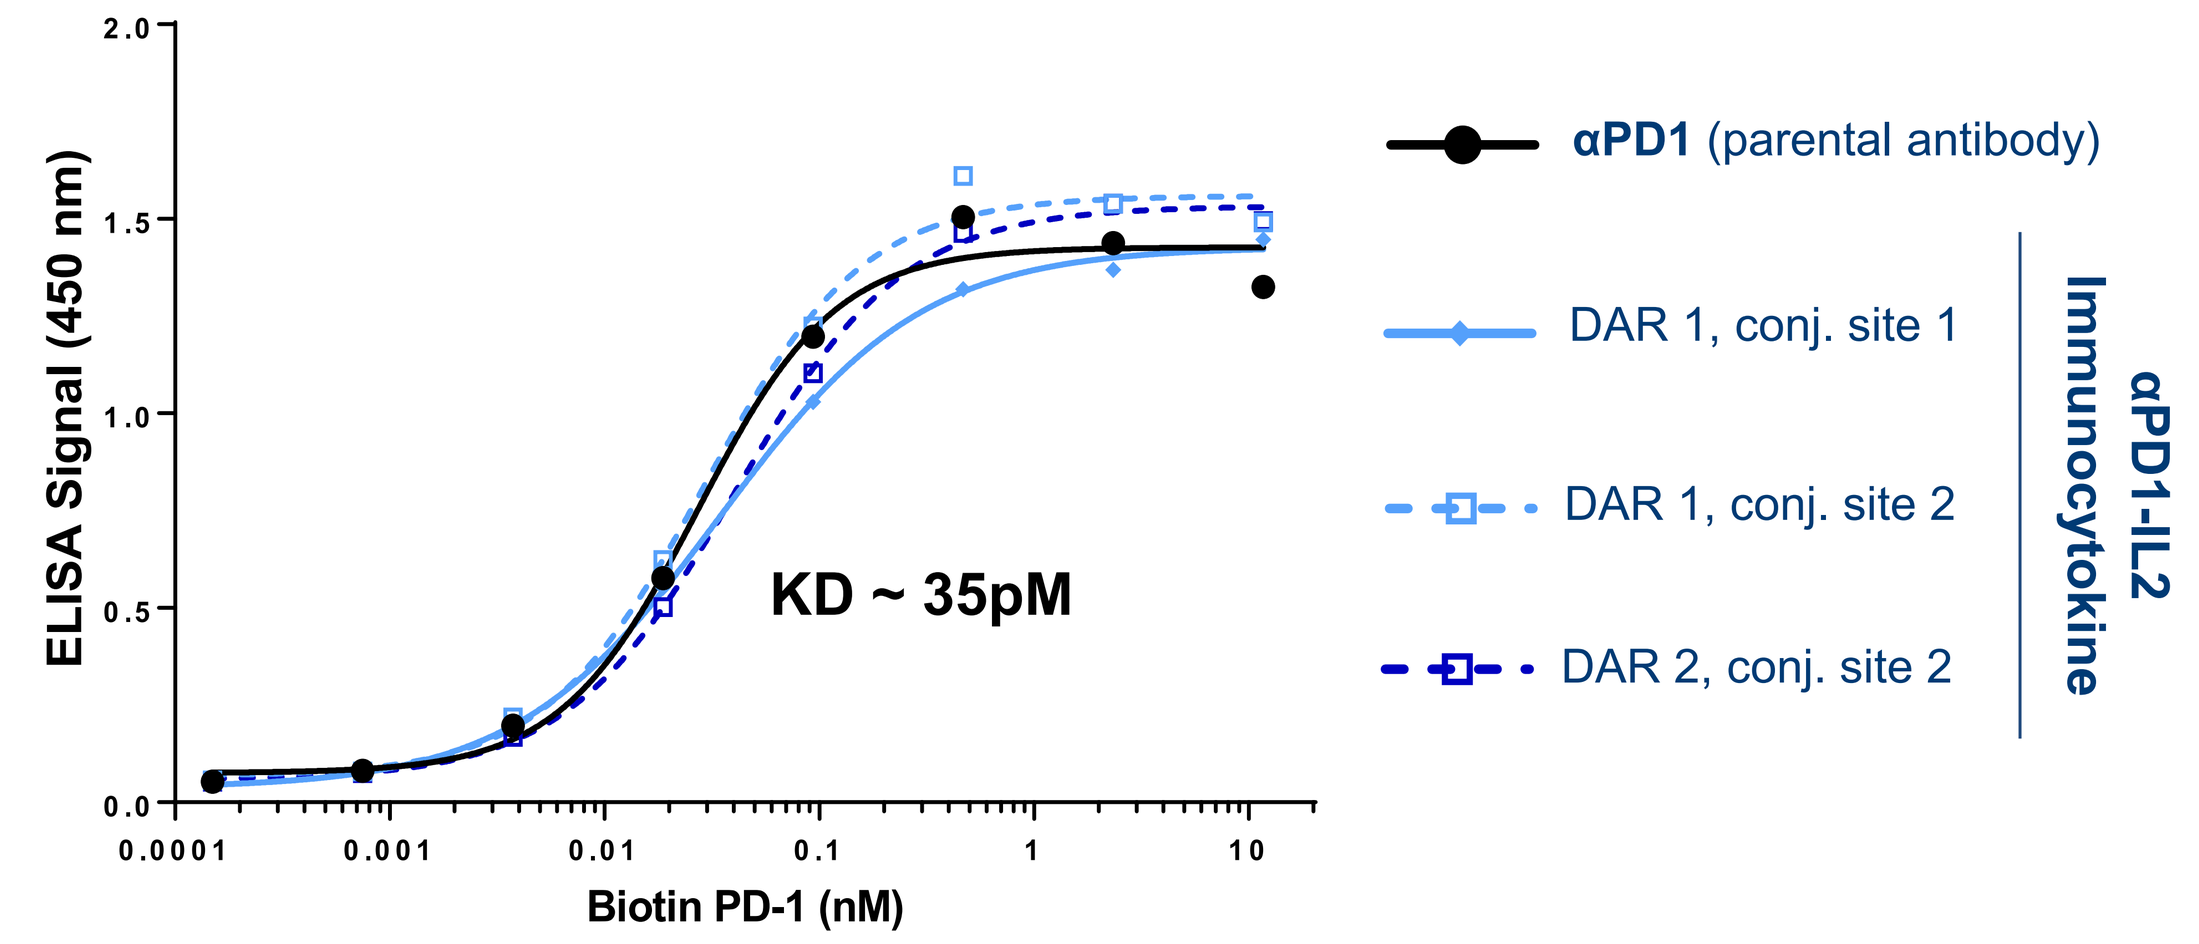
ELISA Signal (450 nm)
Biotin PD-1 (nM)
KD ~ 35pM
αPD1 (parental antibody)
DAR 1, conj. site 1
DAR 1, conj. site 2
DAR 2, conj. site 2
αPD1-IL2 Immunocytokine

Figure 1. ELISA measuring binding of different αPD1-IL2 IC formats to human PD-1.

Binding to Neonatal Fc receptor (FcRn) is marginally reduced after conjugation

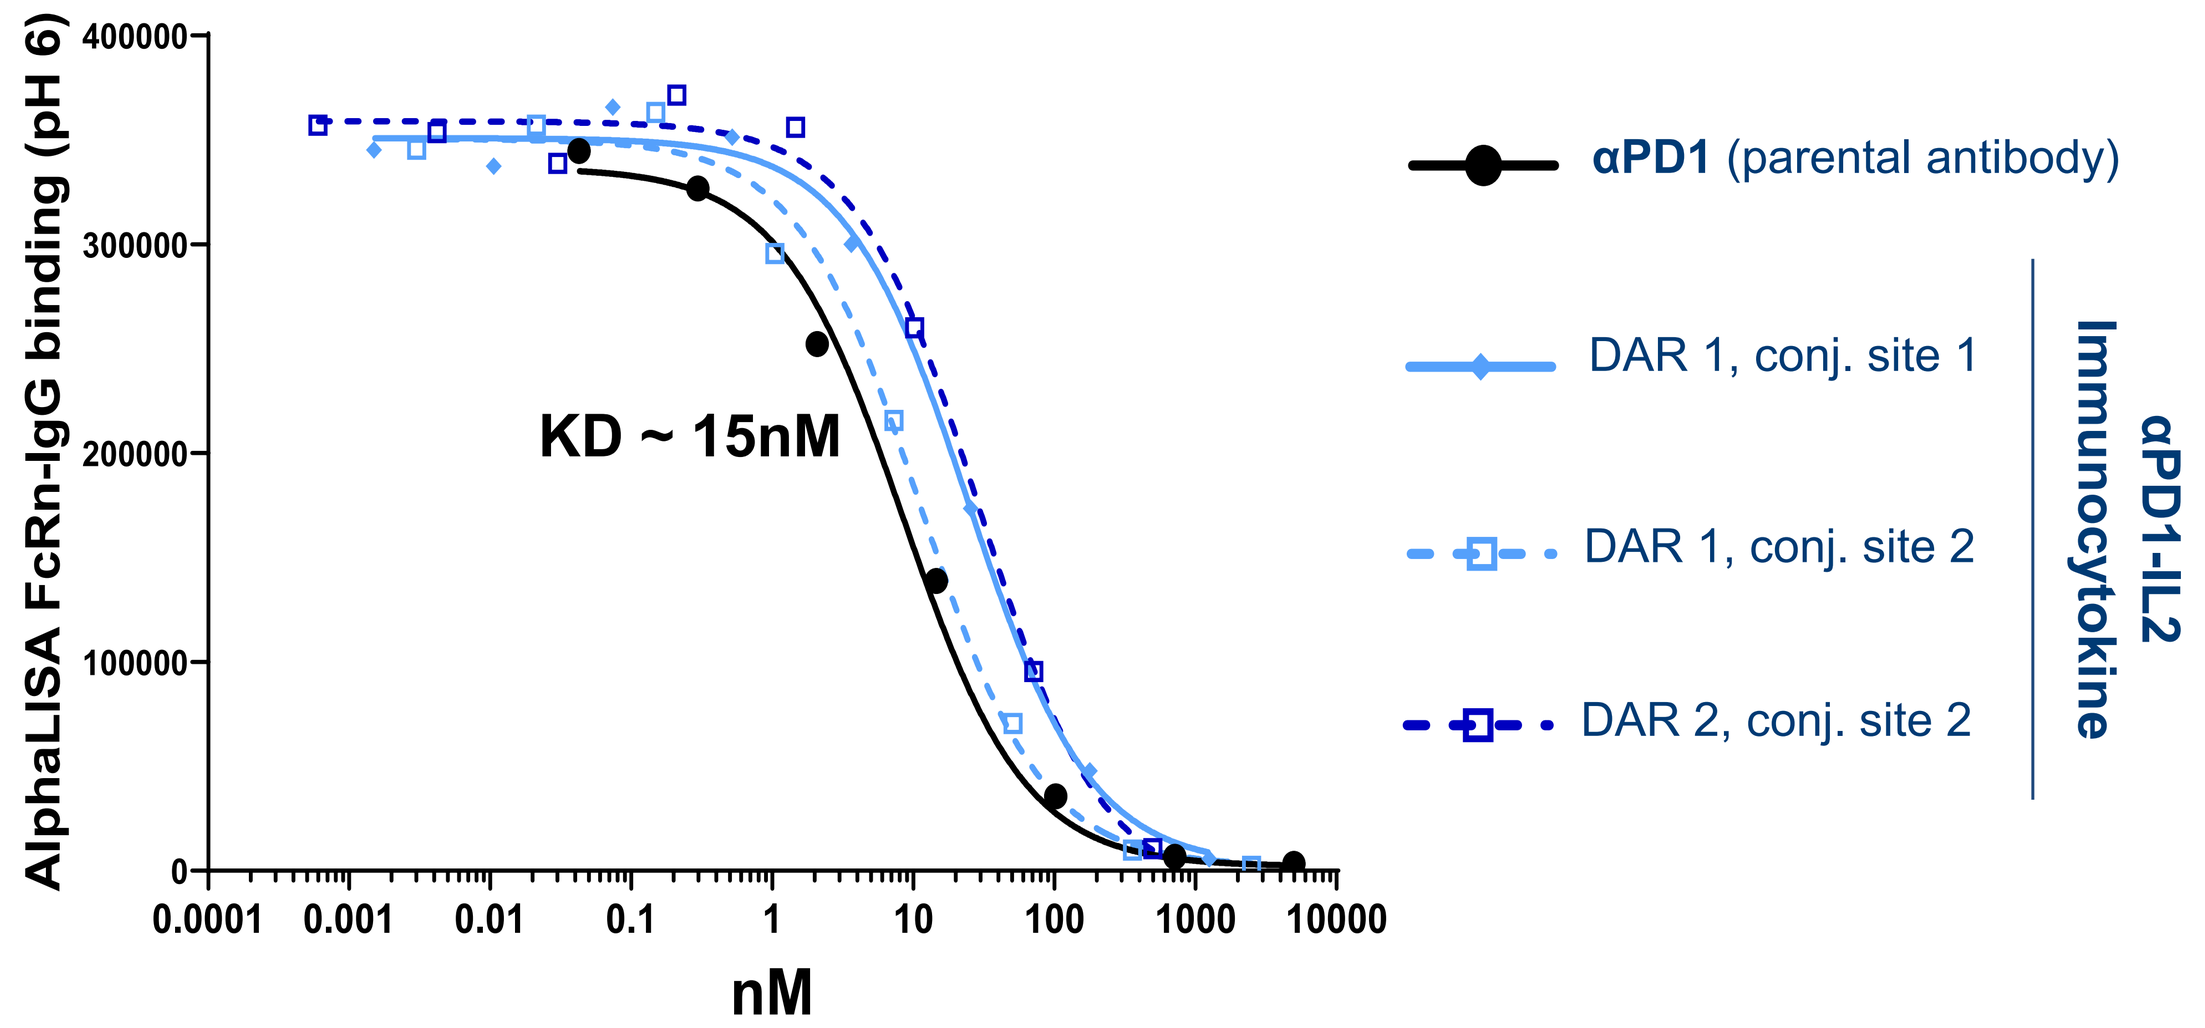
AlphaLISA FcRn-IgG binding (pH 6)
nM
KD ~ 15nM
αPD1 (parental antibody)
DAR 1, conj. site 1
DAR 1, conj. site 2
DAR 2, conj. site 2
αPD1-IL2 Immunocytokine

Figure 2. Binding of different immunocytokine formats to human FcRn receptor at pH 6 (Perkin Elmer alphaLISA Human FcRn Binding, Kit AL3095C).

Binding to FcγRI but not to FcγRIII is impacted by the drug-antibody ratio (DAR)

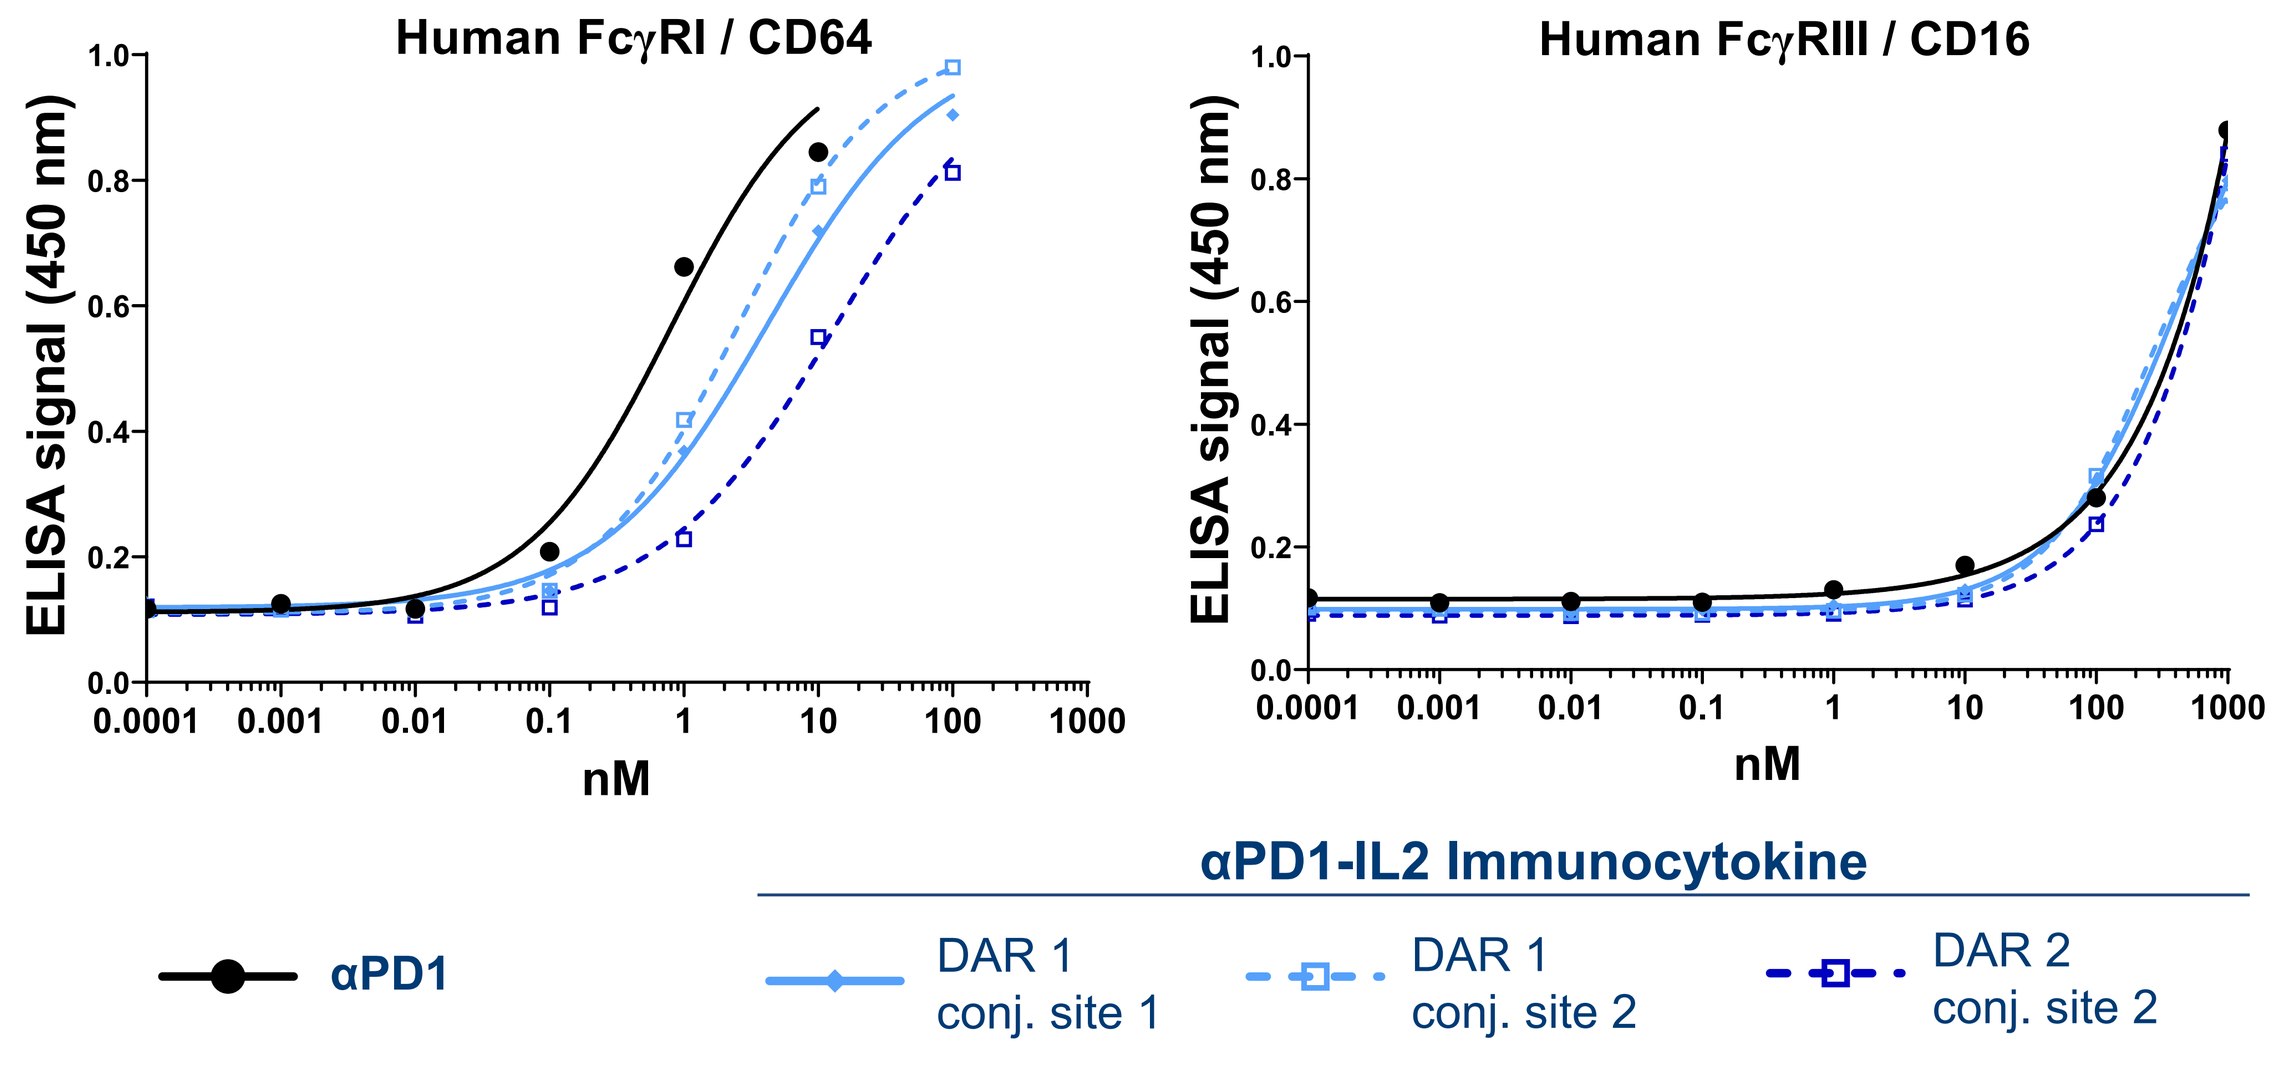
Human FcγRI / CD64
Human FcγRIII / CD16
ELISA signal (450 nm)
nM
αPD1
αPD1-IL2 Immunocytokine
DAR 1 conj. site 1
DAR 1 conj. site 2
DAR 2 conj. site 2

Figure 3. ELISA measuring binding of different αPD1-IL2 IC formats to human Fcγ Receptor I (CD64) and Fcγ Receptor III (CD16)

CYTOKINE/PAYLOAD FUNCTION

Minor reduction of potency but preserved CD8+/Treg selectivity after Ab conjugation

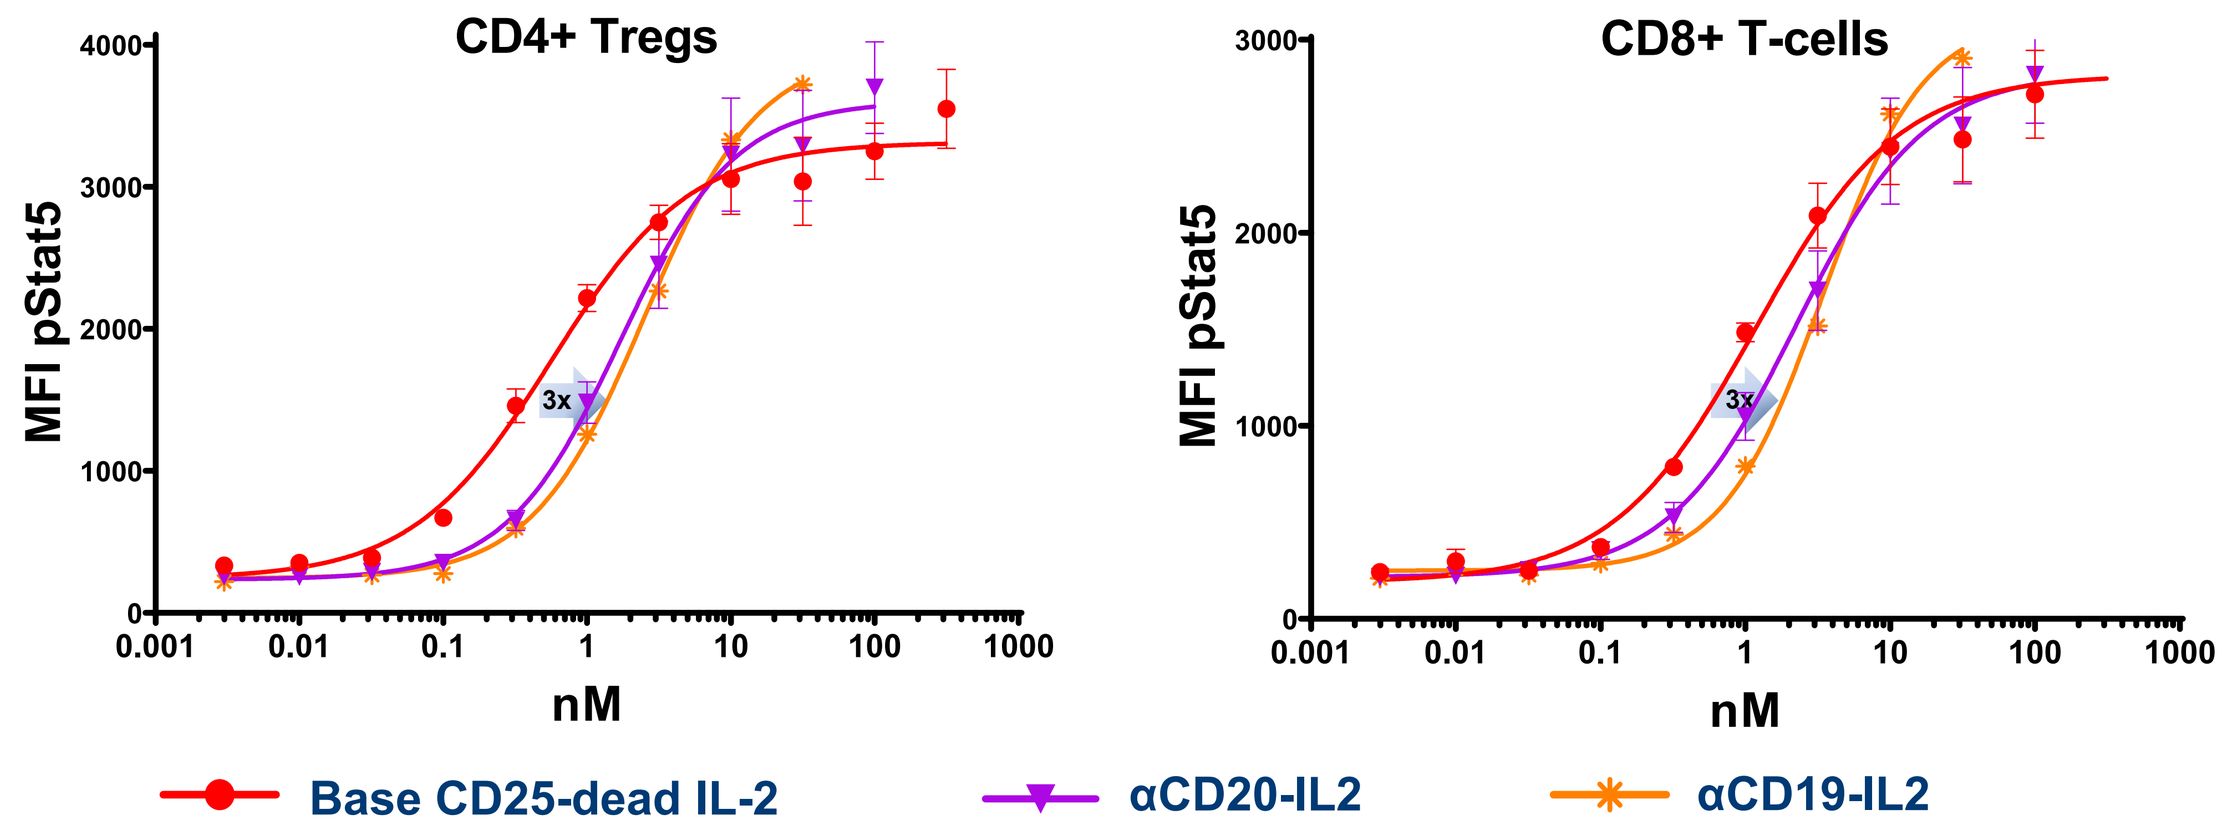
CD4+ Tregs
CD8+ T-cells
MFI pStat5
nM
3x
Base CD25-dead IL-2
αCD20-IL2
αCD19-IL2

Figure 4. STAT5 phosphorylation in primary human CD8+ T cells and Tregs after stimulation with indicated molecules.
(average ± SEM of three donors)

TARGETED DELIVERY

PD1-IL2 shows avidity-mediated enhanced potency on PD-1^{high} CD8+ T cells

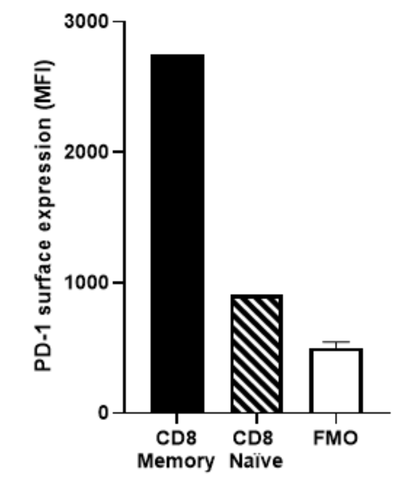
PD-1 surface expression (MFI)
CD8 Memory
CD8 Naive
FMO

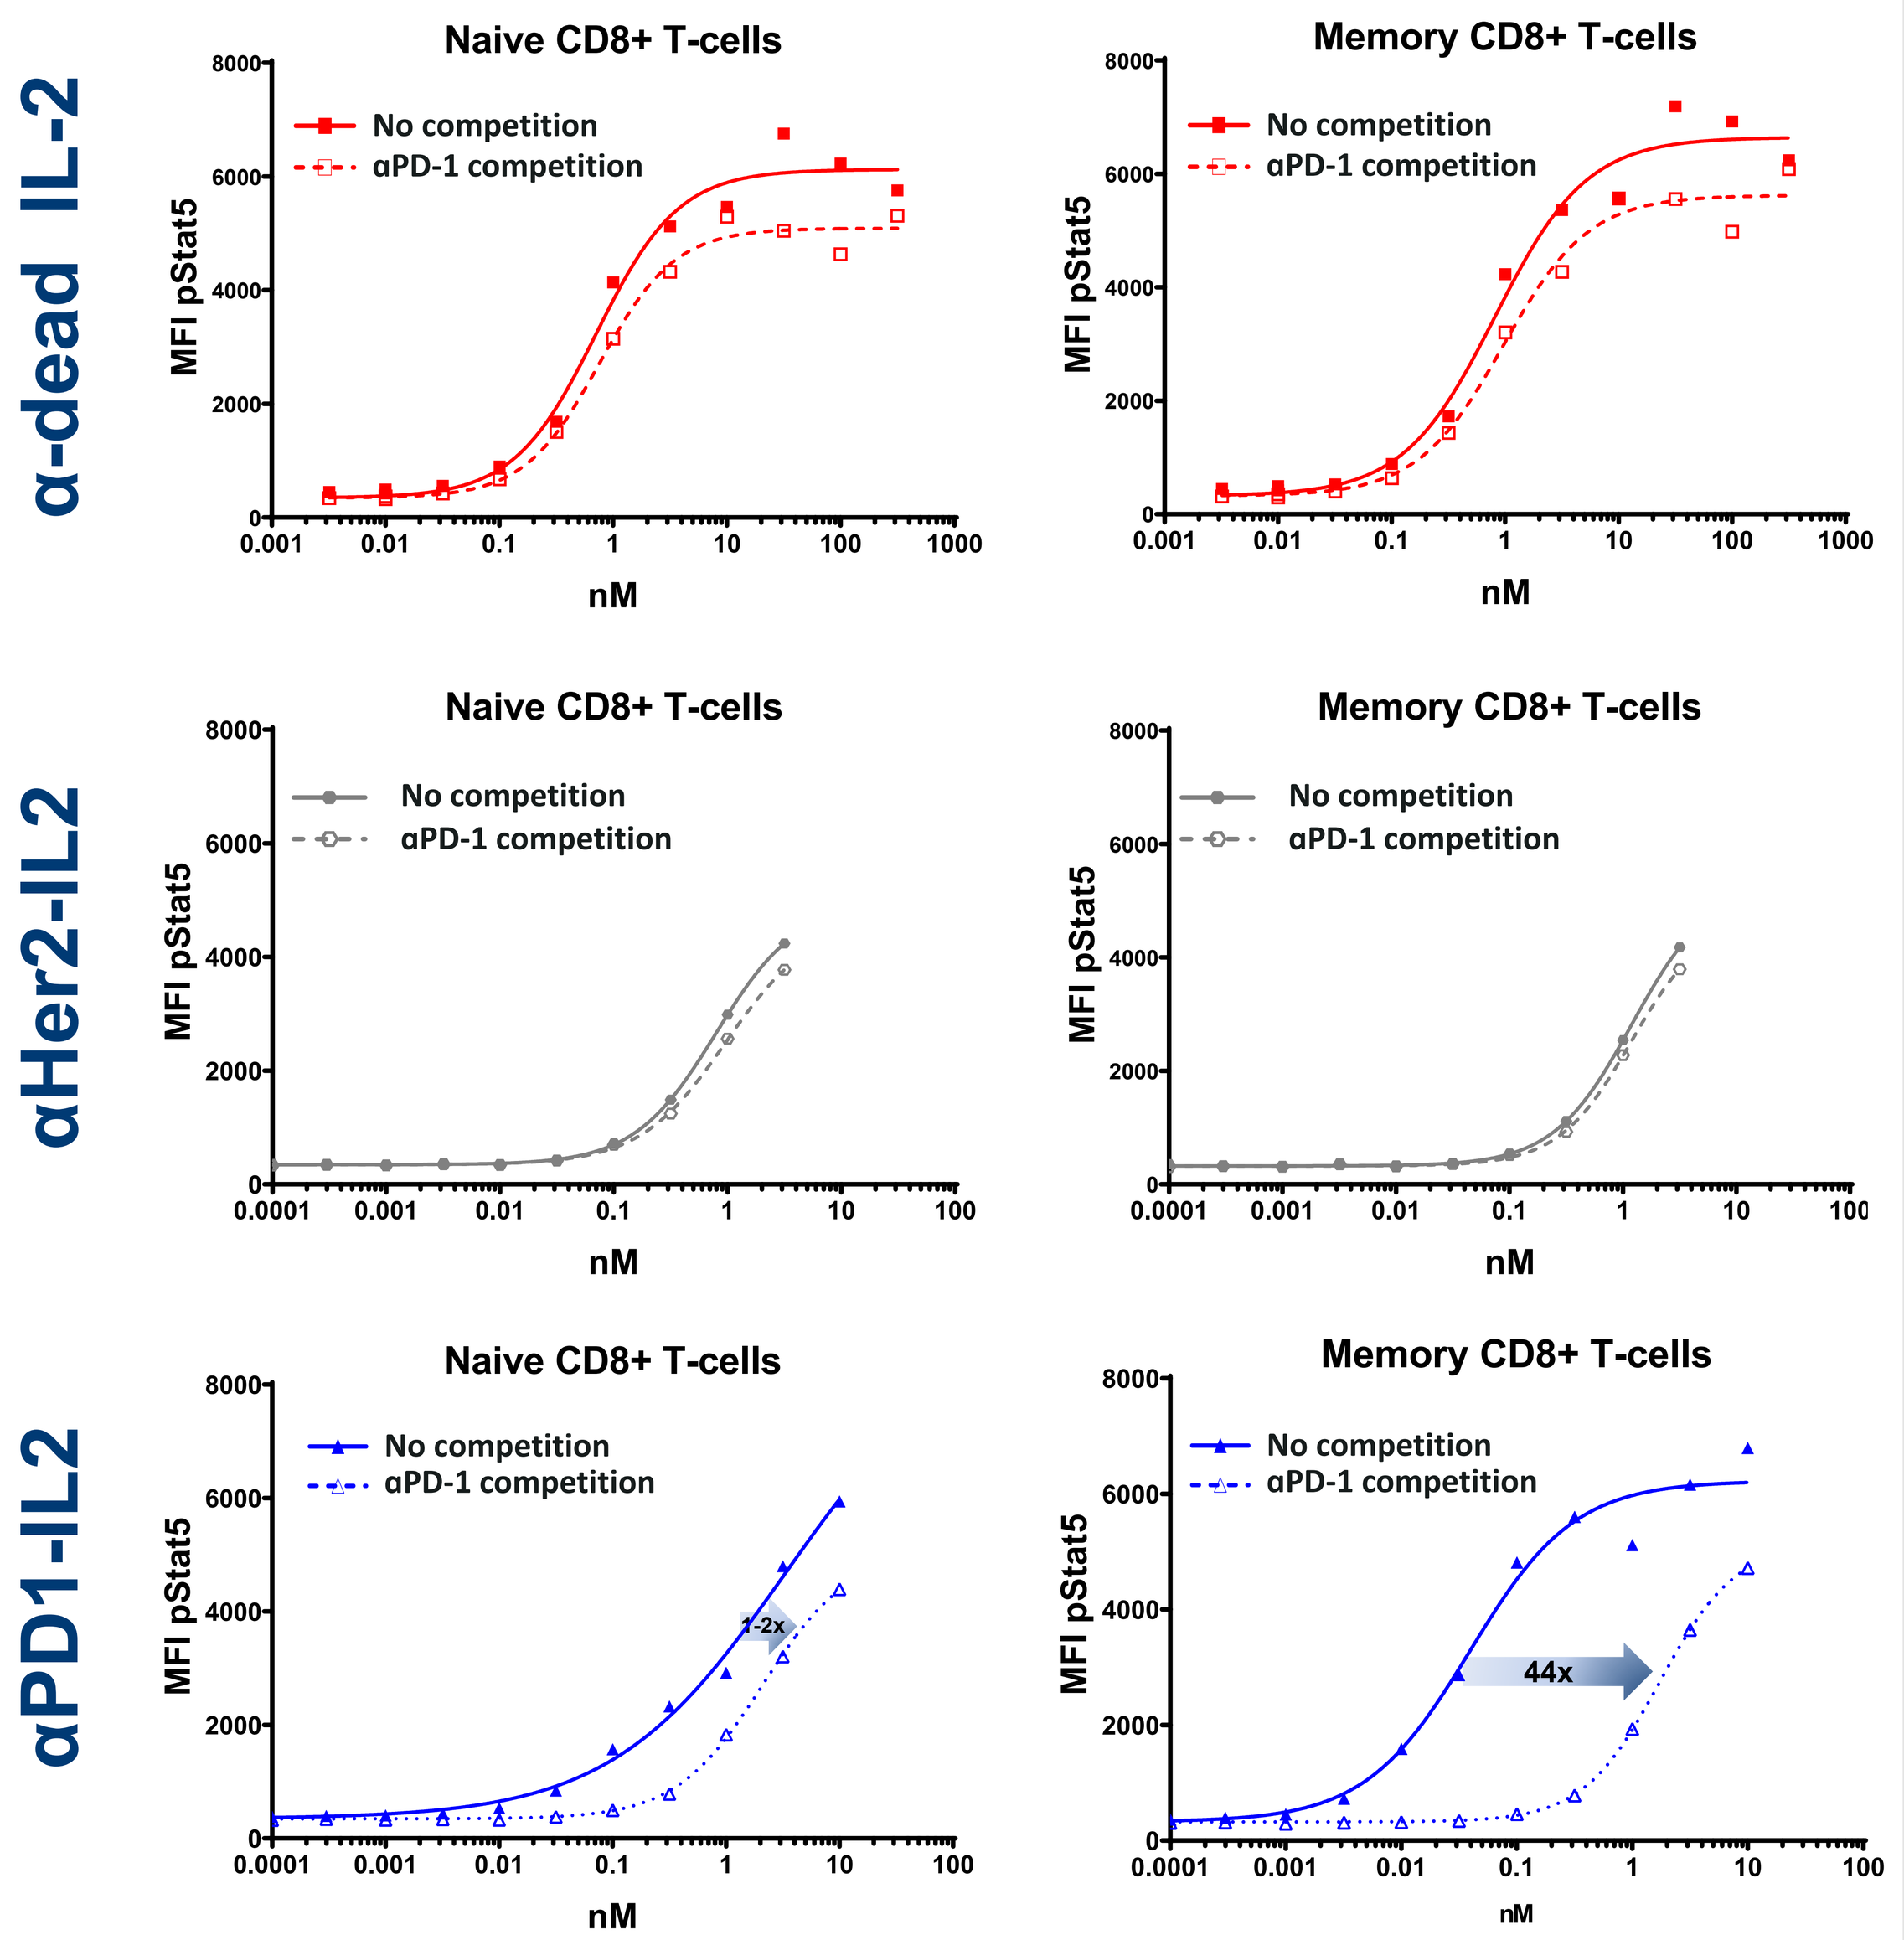
α-dead IL-2
αHer2-IL2
αPD1-IL2
Naive CD8+ T-cells
Memory CD8+ T-cells
MFI pStat5
nM
No competition
αPD-1 competition
1-2x
44x

Figure 5. STAT5 phosphorylation in primary human memory and naïve CD8+ T-cells after stimulation with indicated molecules in the absence (solid lines) or presence (dotted lines) of excess αPD-1 antibody (100nM).

MOUSE PD/EFFICACY (Pilot)

αPD1-IL2 (α-dead) immunocytokine triggers higher CD8+ T-cell expansion and tumor growth inhibition than non-targeted IL-2 (α-dead) IC.

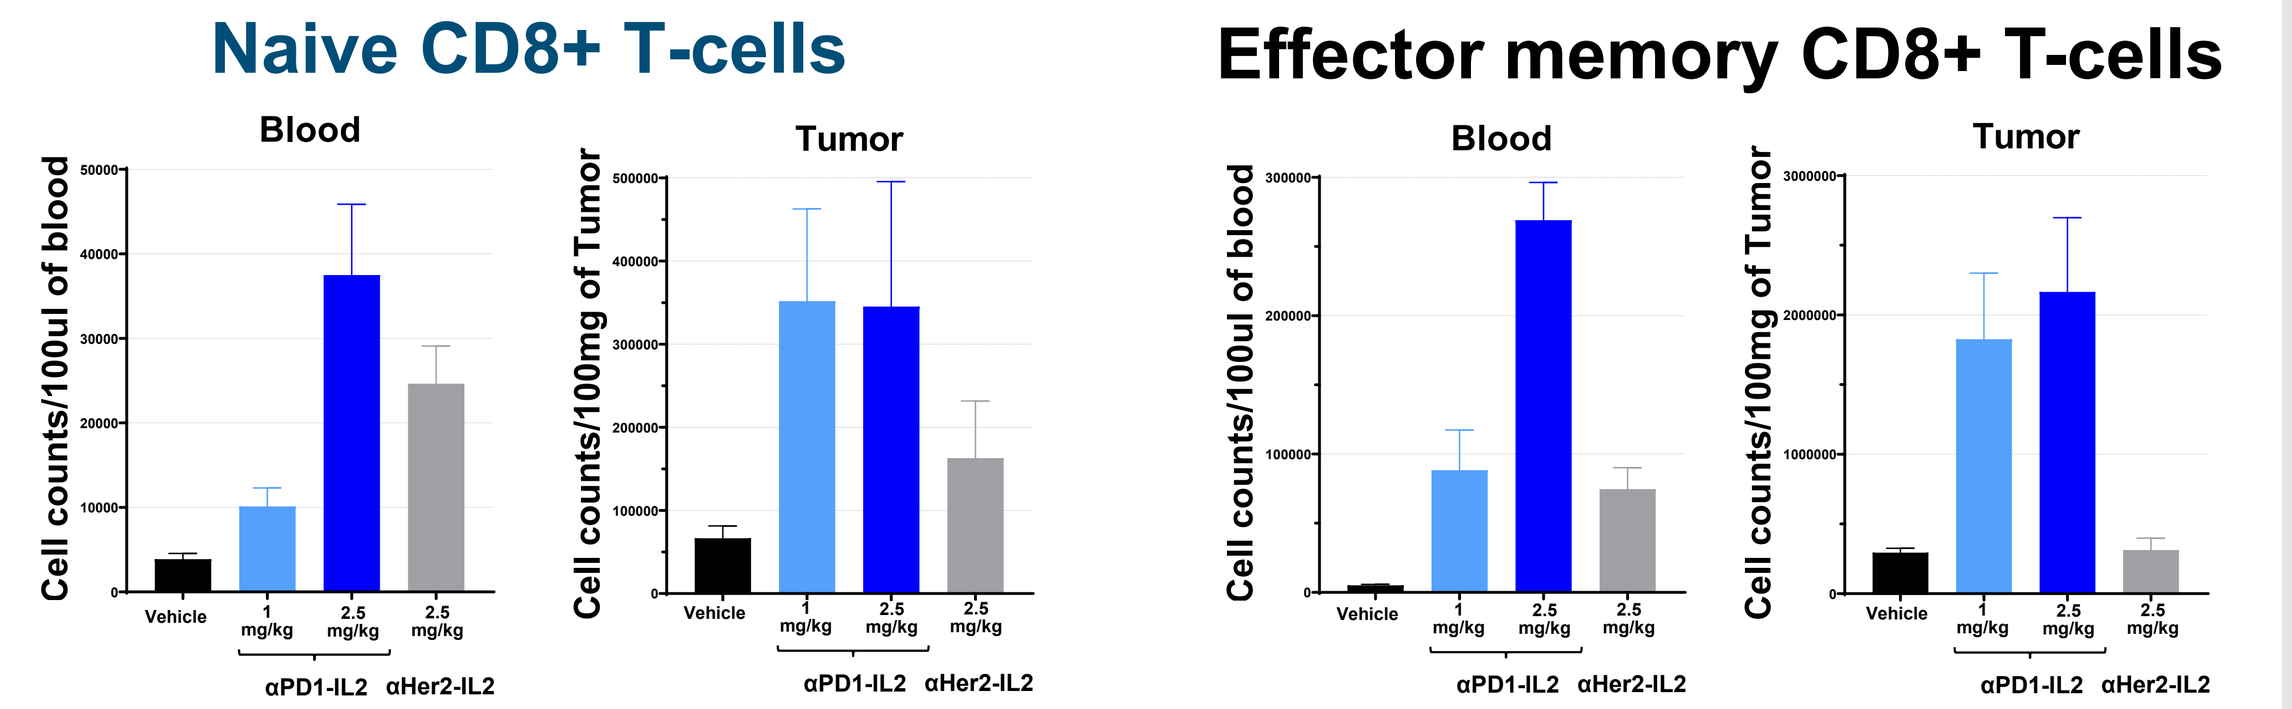
Naive CD8+ T-cells
Effector memory CD8+ T-cells
Blood
Tumor
Cell counts/100ul of blood
Cell counts/100mg of Tumor
Vehicle
1 mg/kg
2.5 mg/kg
αPD1-IL2
αHer2-IL2

Day 7 post injection

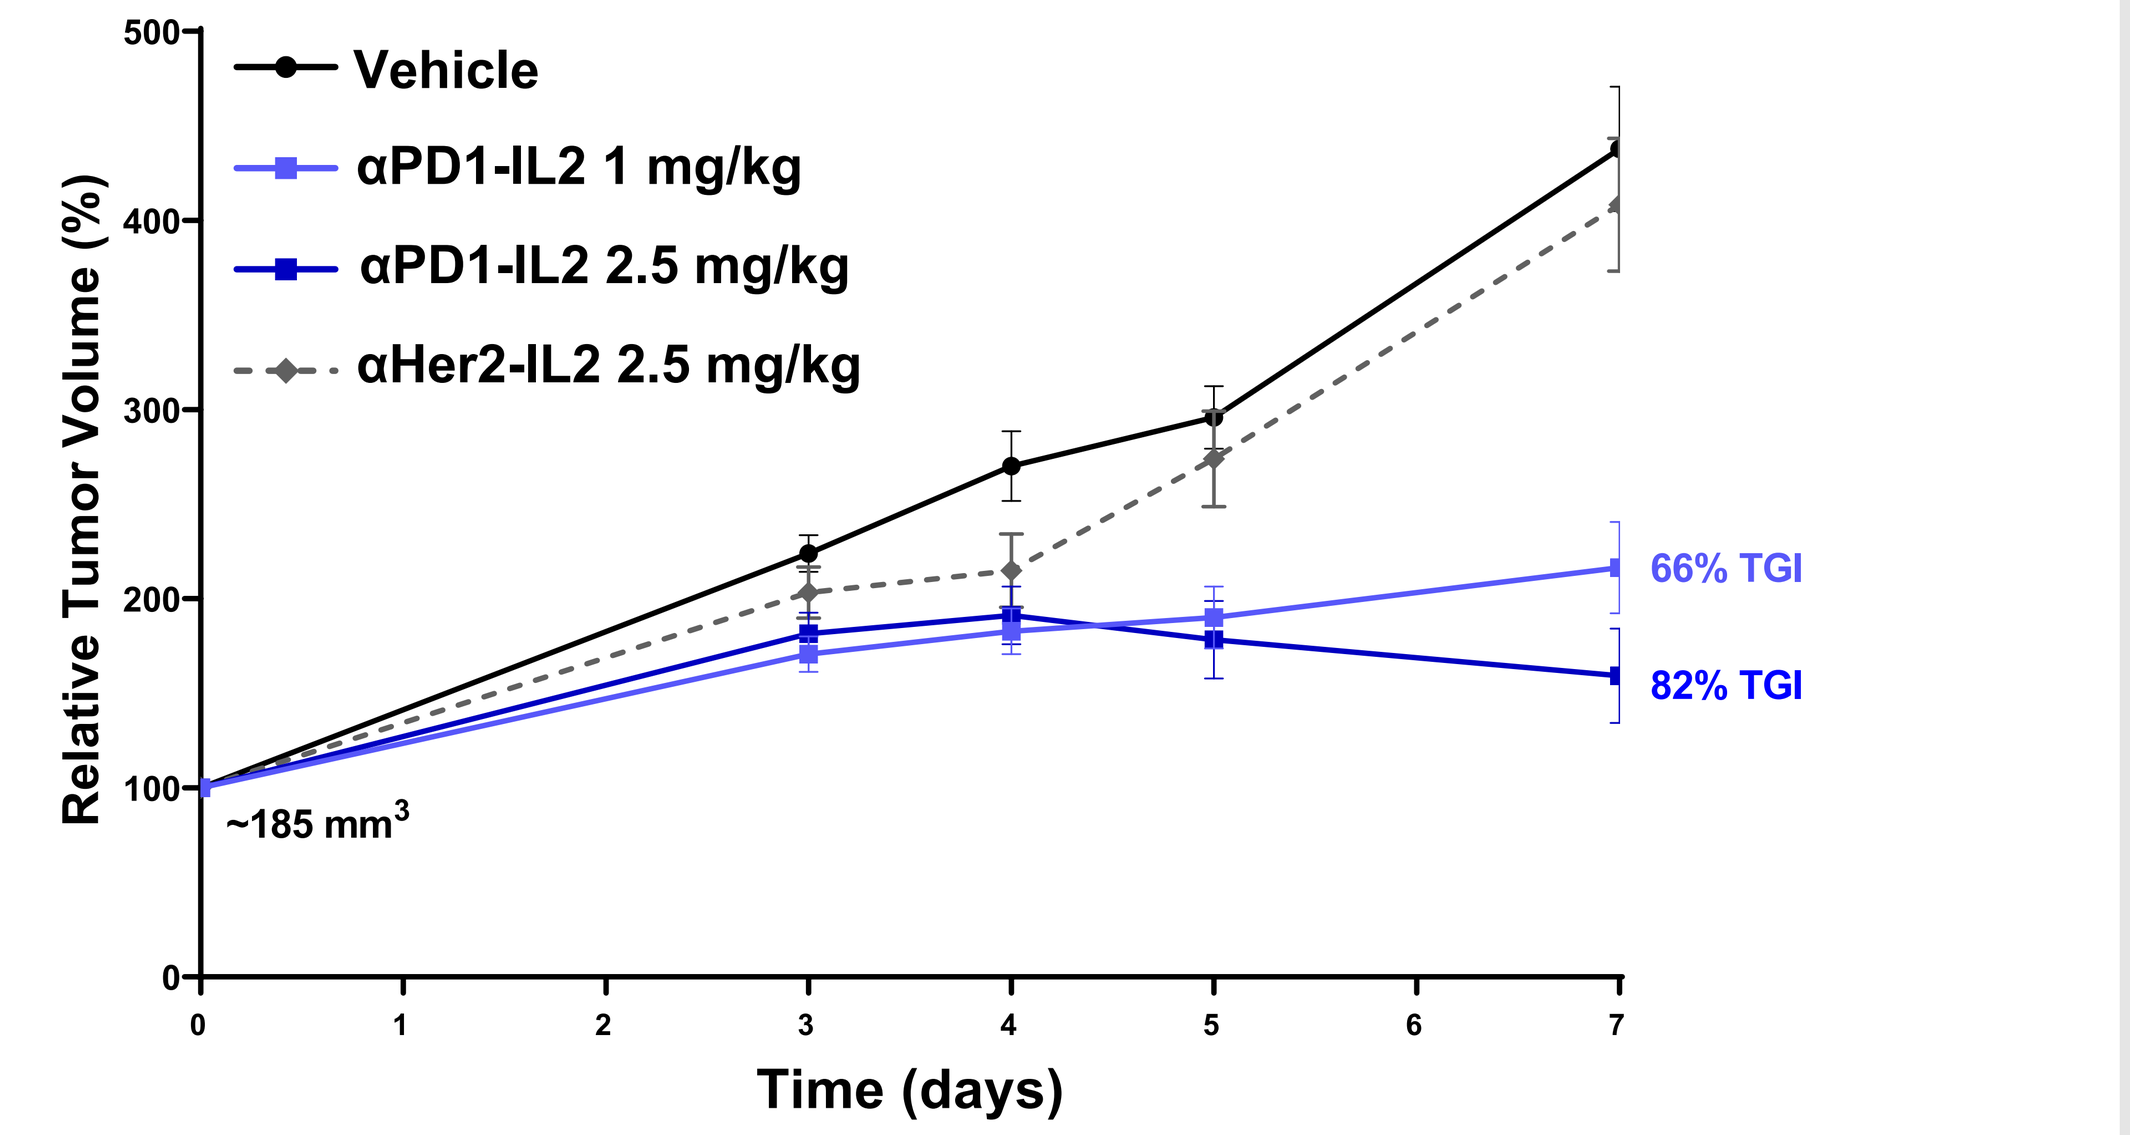
Vehicle
αPD1-IL2 1 mg/kg
αPD1-IL2 2.5 mg/kg
αHer2-IL2 2.5 mg/kg
Relative Tumor Volume (%)
Time (days)
~185 mm3
66% TGI
82% TGI

Figure 6. CT26 tumor-bearing humanized PD-1 BALB/c mice were treated with a single dose of indicated molecules injected i.v.
(average ± SEM of three mice per group)

CONCLUSIONS

- Bright Peak immunocytokines are generated by an approach that is entirely different from conventional fusion protein synthesis - via chemical conjugation of engineered cytokines to the Fc domain of existing human Abs in a rapid (2-3 week), cell-free process.
- Conjugation is site-specific and existing antibodies can be used "off-the-shelf" without antibody engineering.
- ICs have been successfully generated using more than 10 existing antibodies with at least 4 different engineered cytokine payloads.
- Upon conjugation, both Ab and cytokine functions are preserved and only a minor impact on IC binding to FcRn or Fcγ receptors has been observed.
- As proof-of-concept, we generated an αPD1-IL2 (α-dead) IC that demonstrates avidity effects with strongly enhanced potency in PD-1-high CD8+ T cells and significantly greater anti-tumor activity in vivo than a non-targeted α-dead IL-2 IC.